ARCELLX

ACLX-002*, a Novel CD123-targeted Universal CAR-T Cell Therapy for Relapsed or Refractory Acute Myeloid Leukemia

Abstract 587

Justin P. Edwards, Liubov Zaritskaya, Jeffrey Swers, Sinnie Ng, Jenny Mu, Laurene Cheung, Alexandra Witter, Samantha McCullough, Hui Wang, Ankit Gupta, David LaFleur, David Hilbert, David Tice

Arcellx, Gaithersburg, MD

JEdwards@arcellx.com

Introduction

The ARC-SparX Platform and ACLX-002*

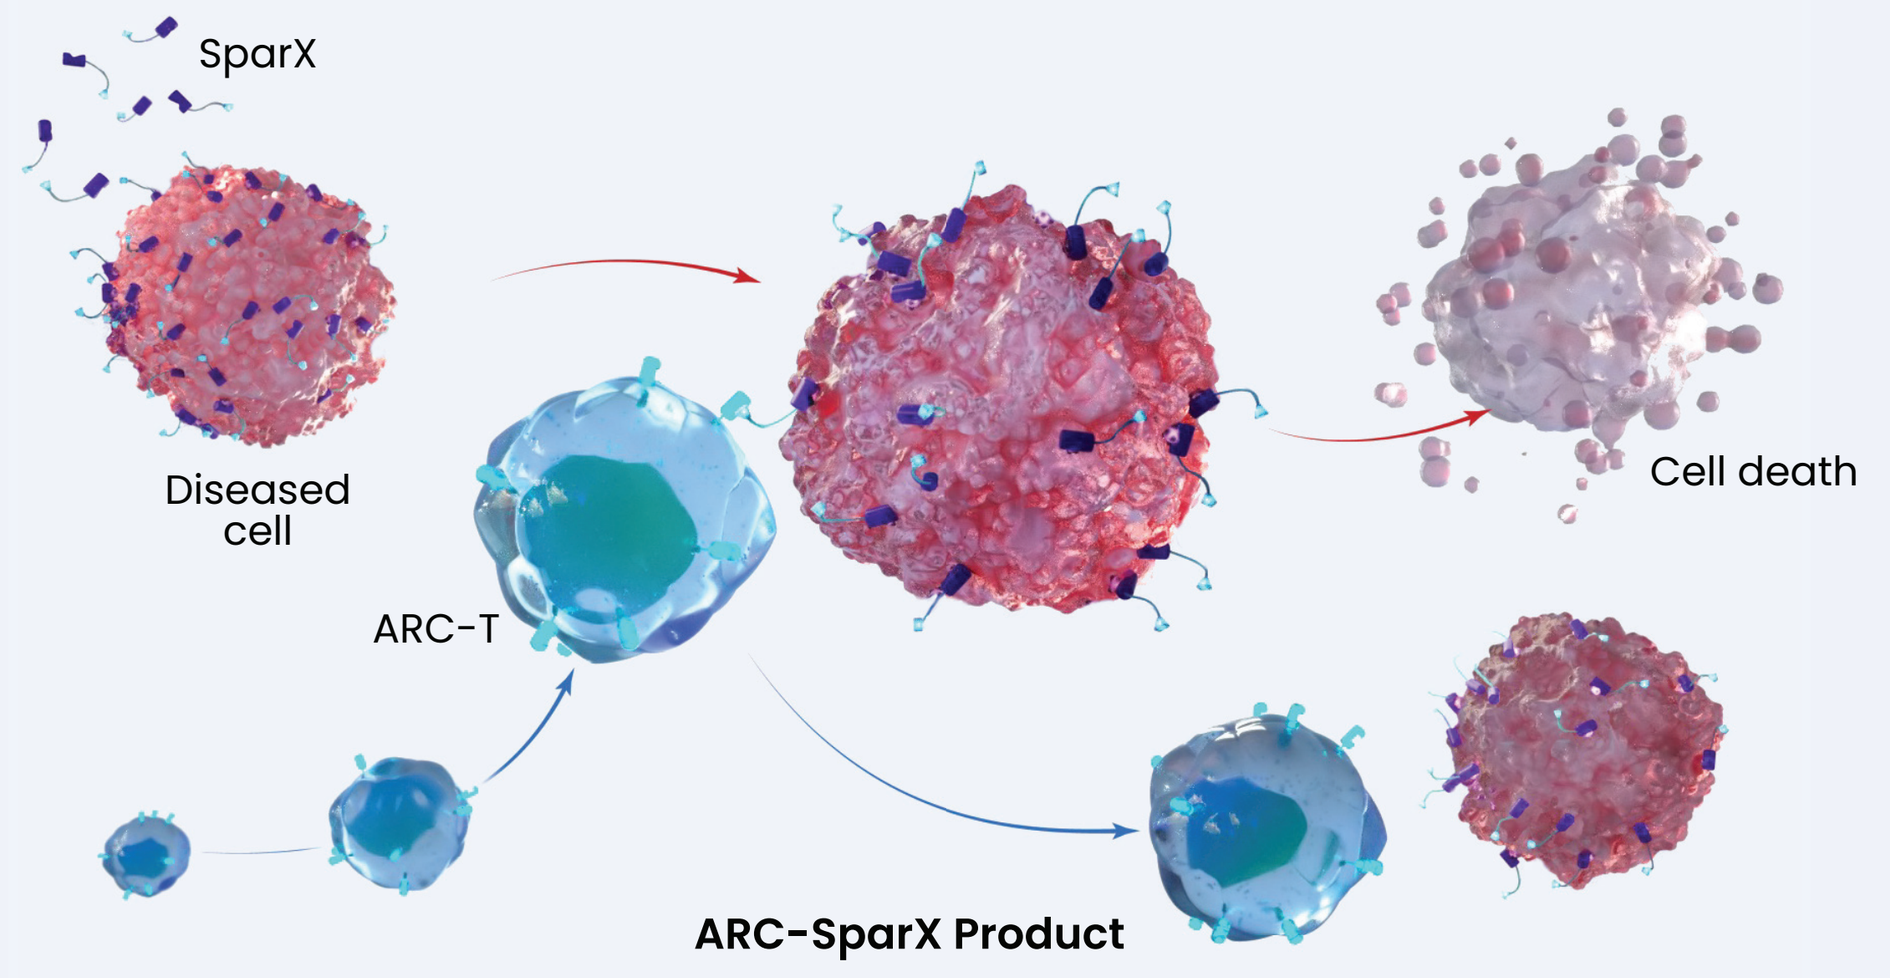

SparX
Diseased cell
Cell death
ARC-T
ARC-SparX Product

The ARC-SparX platform separates the antigen-recognition and killing functions of conventional CAR-T therapy and is comprised of two key components:

1. **SPRX002** (**s**oluble **pr**otein **a**ntigen-**r**eceptor **X**-linker) protein: binds CD123 on diseased cells and flags those cells for destruction.
2. **ARC-T Cells** (**A**ntigen **R**eceptor **C**omplex **T** Cells): bind and kill SPRX002 flagged cells.
3. **ACLX-002** is the combination of the ARC-T cells and SPRX002.

SPRX002 protein

- No inherent therapeutic activity
- Contains one CD123-targeting domain
- Binds CD123+ acute myeloid leukemia cells
- Engineered to minimize immunogenic potential
- "TAG" is a fragment of human alpha fetoprotein (AFP)

ARC-T cell

- CAR is comprised of the extracellular novel binding domain fused to CD8α spacer/transmembrane region fused to 4-1BB/CD3ζ signaling domains
- Only activated upon formation of a tricomplex of ARC-T + SPRX002 + CD123 positive acute myeloid leukemia cells
- Same viral vector regardless of antigen target

Results

SPRX002 binds to CD123+ target cells in a CD123-dependent manner and coincident with ARC-T cells

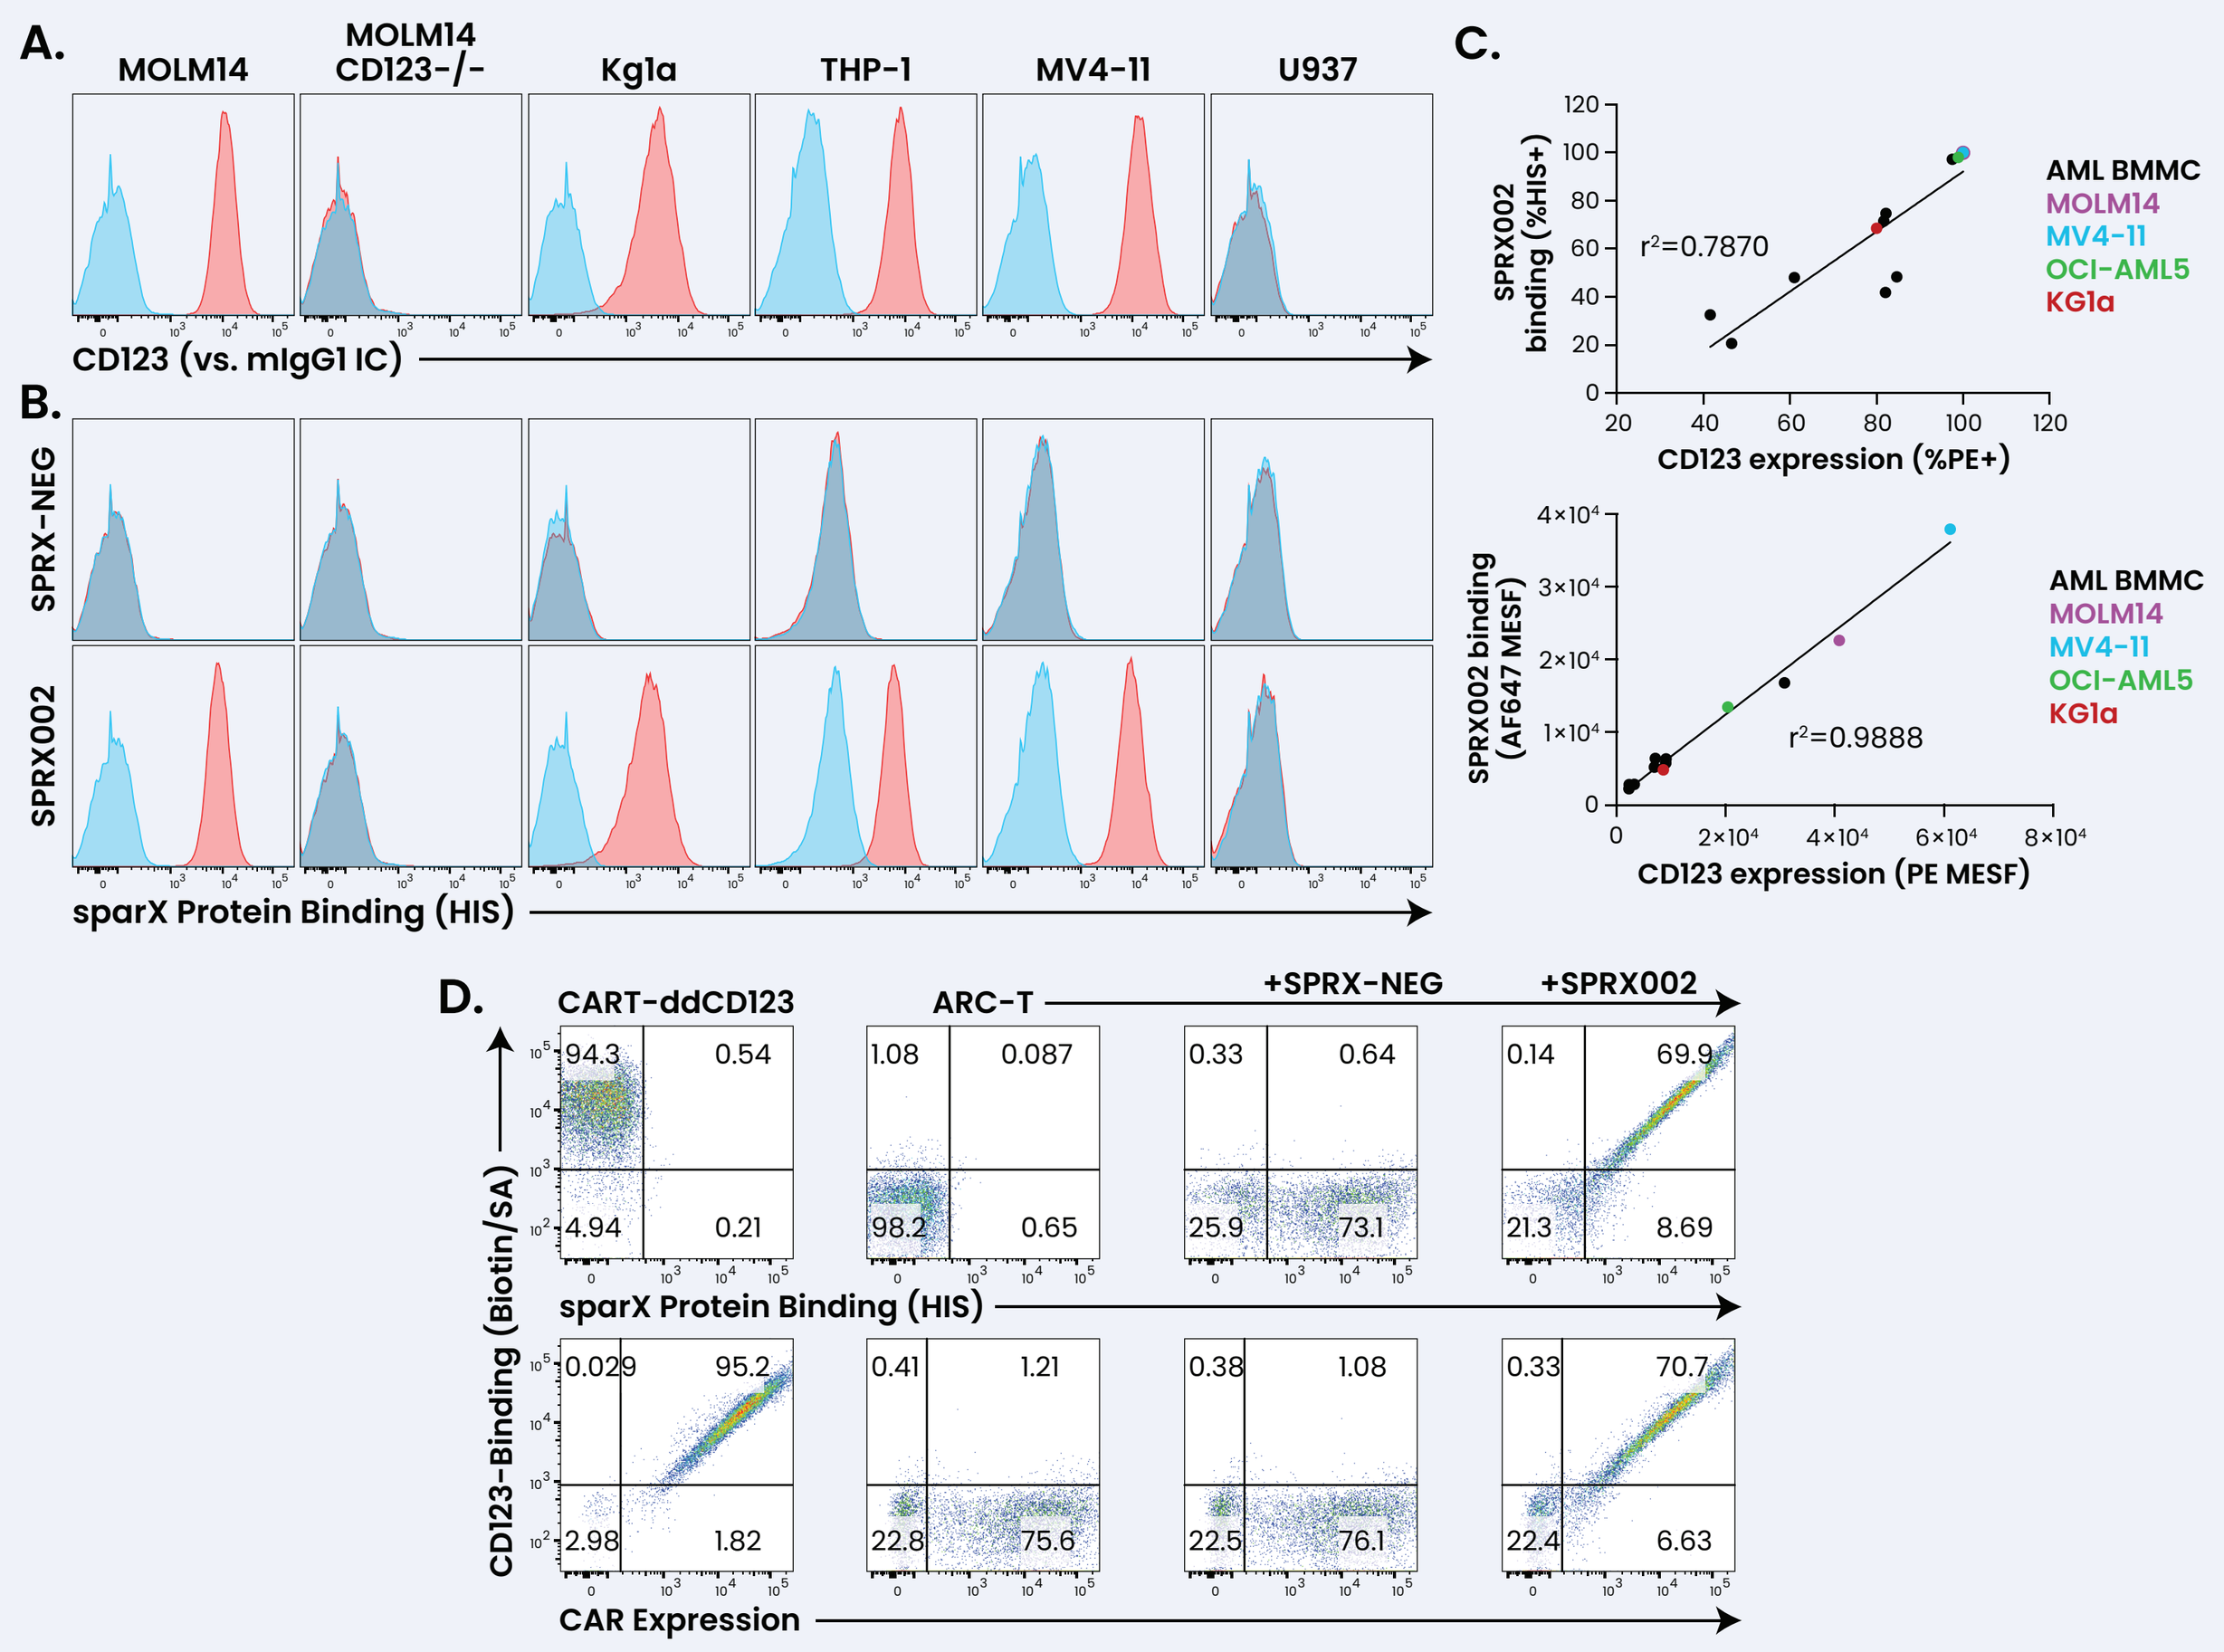

A.
MOLM14
MOLM14 CD123-/-
Kg1a
THP-1
MV4-11
U937
CD123 (vs. mIgG1 IC)
B.
SPRX-NEG
SPRX002
sparX Protein Binding (HIS)
C.
SPRX002 binding (%HIS+)
r²=0.7870
CD123 expression (%PE+)
AML BMMC
MOLM14
MV4-11
OCI-AML5
KG1a
SPRX002 binding (AF647 MESF)
r²=0.9888
CD123 expression (PE MESF)
D.
CART-ddCD123
ARC-T
+SPRX-NEG
+SPRX002
CD123-Binding (Biotin/SA)
CAR Expression

SPRX002 binds to CD123+ targets in a CD123-dependent manner. **(A)** Various AML cell lines were stained for CD123 expression (pink) or with isotype control (blue). **(B)** Cell lines from A were incubated with the CD123-binding SPRX002 protein or with the negative control protein SPRX-NEG, then binding detected with anti-HIS antibody. **(C)** CD123 expression was detected by a PE- conjugated antibody and SPRX002 binding was detected with an anti-HIS A647 antibody on primary AML bone marrow mononuclear cells (BMMCs) and AML cell lines. The correlation of CD123 percent positivity with SPRX002 binding (top) and CD123 molecules of equivalent soluble fluorescence (MESF) versus anti-HIS MESF (bottom) is presented. **(D)** SPRX-binding to ARC-T confers CD123 binding capacity. ARC-T cells were sequentially incubated with SPRX002 or the negative control SPRX-NEG, then biotinylated CD123-FC. Binding of sparX proteins was detected via anti-HIS and CD123-binding was detected with streptavidin-APC. In parallel, a D-domain based CAR-T targeting CD123, CART-ddCD123, was used for binding.

ARC-T cells demonstrate SPRX002 dose-dependent cytokine production, T cell proliferation, and cytotoxic activity of co-cultured CD123-expressing cancer cell lines in vitro

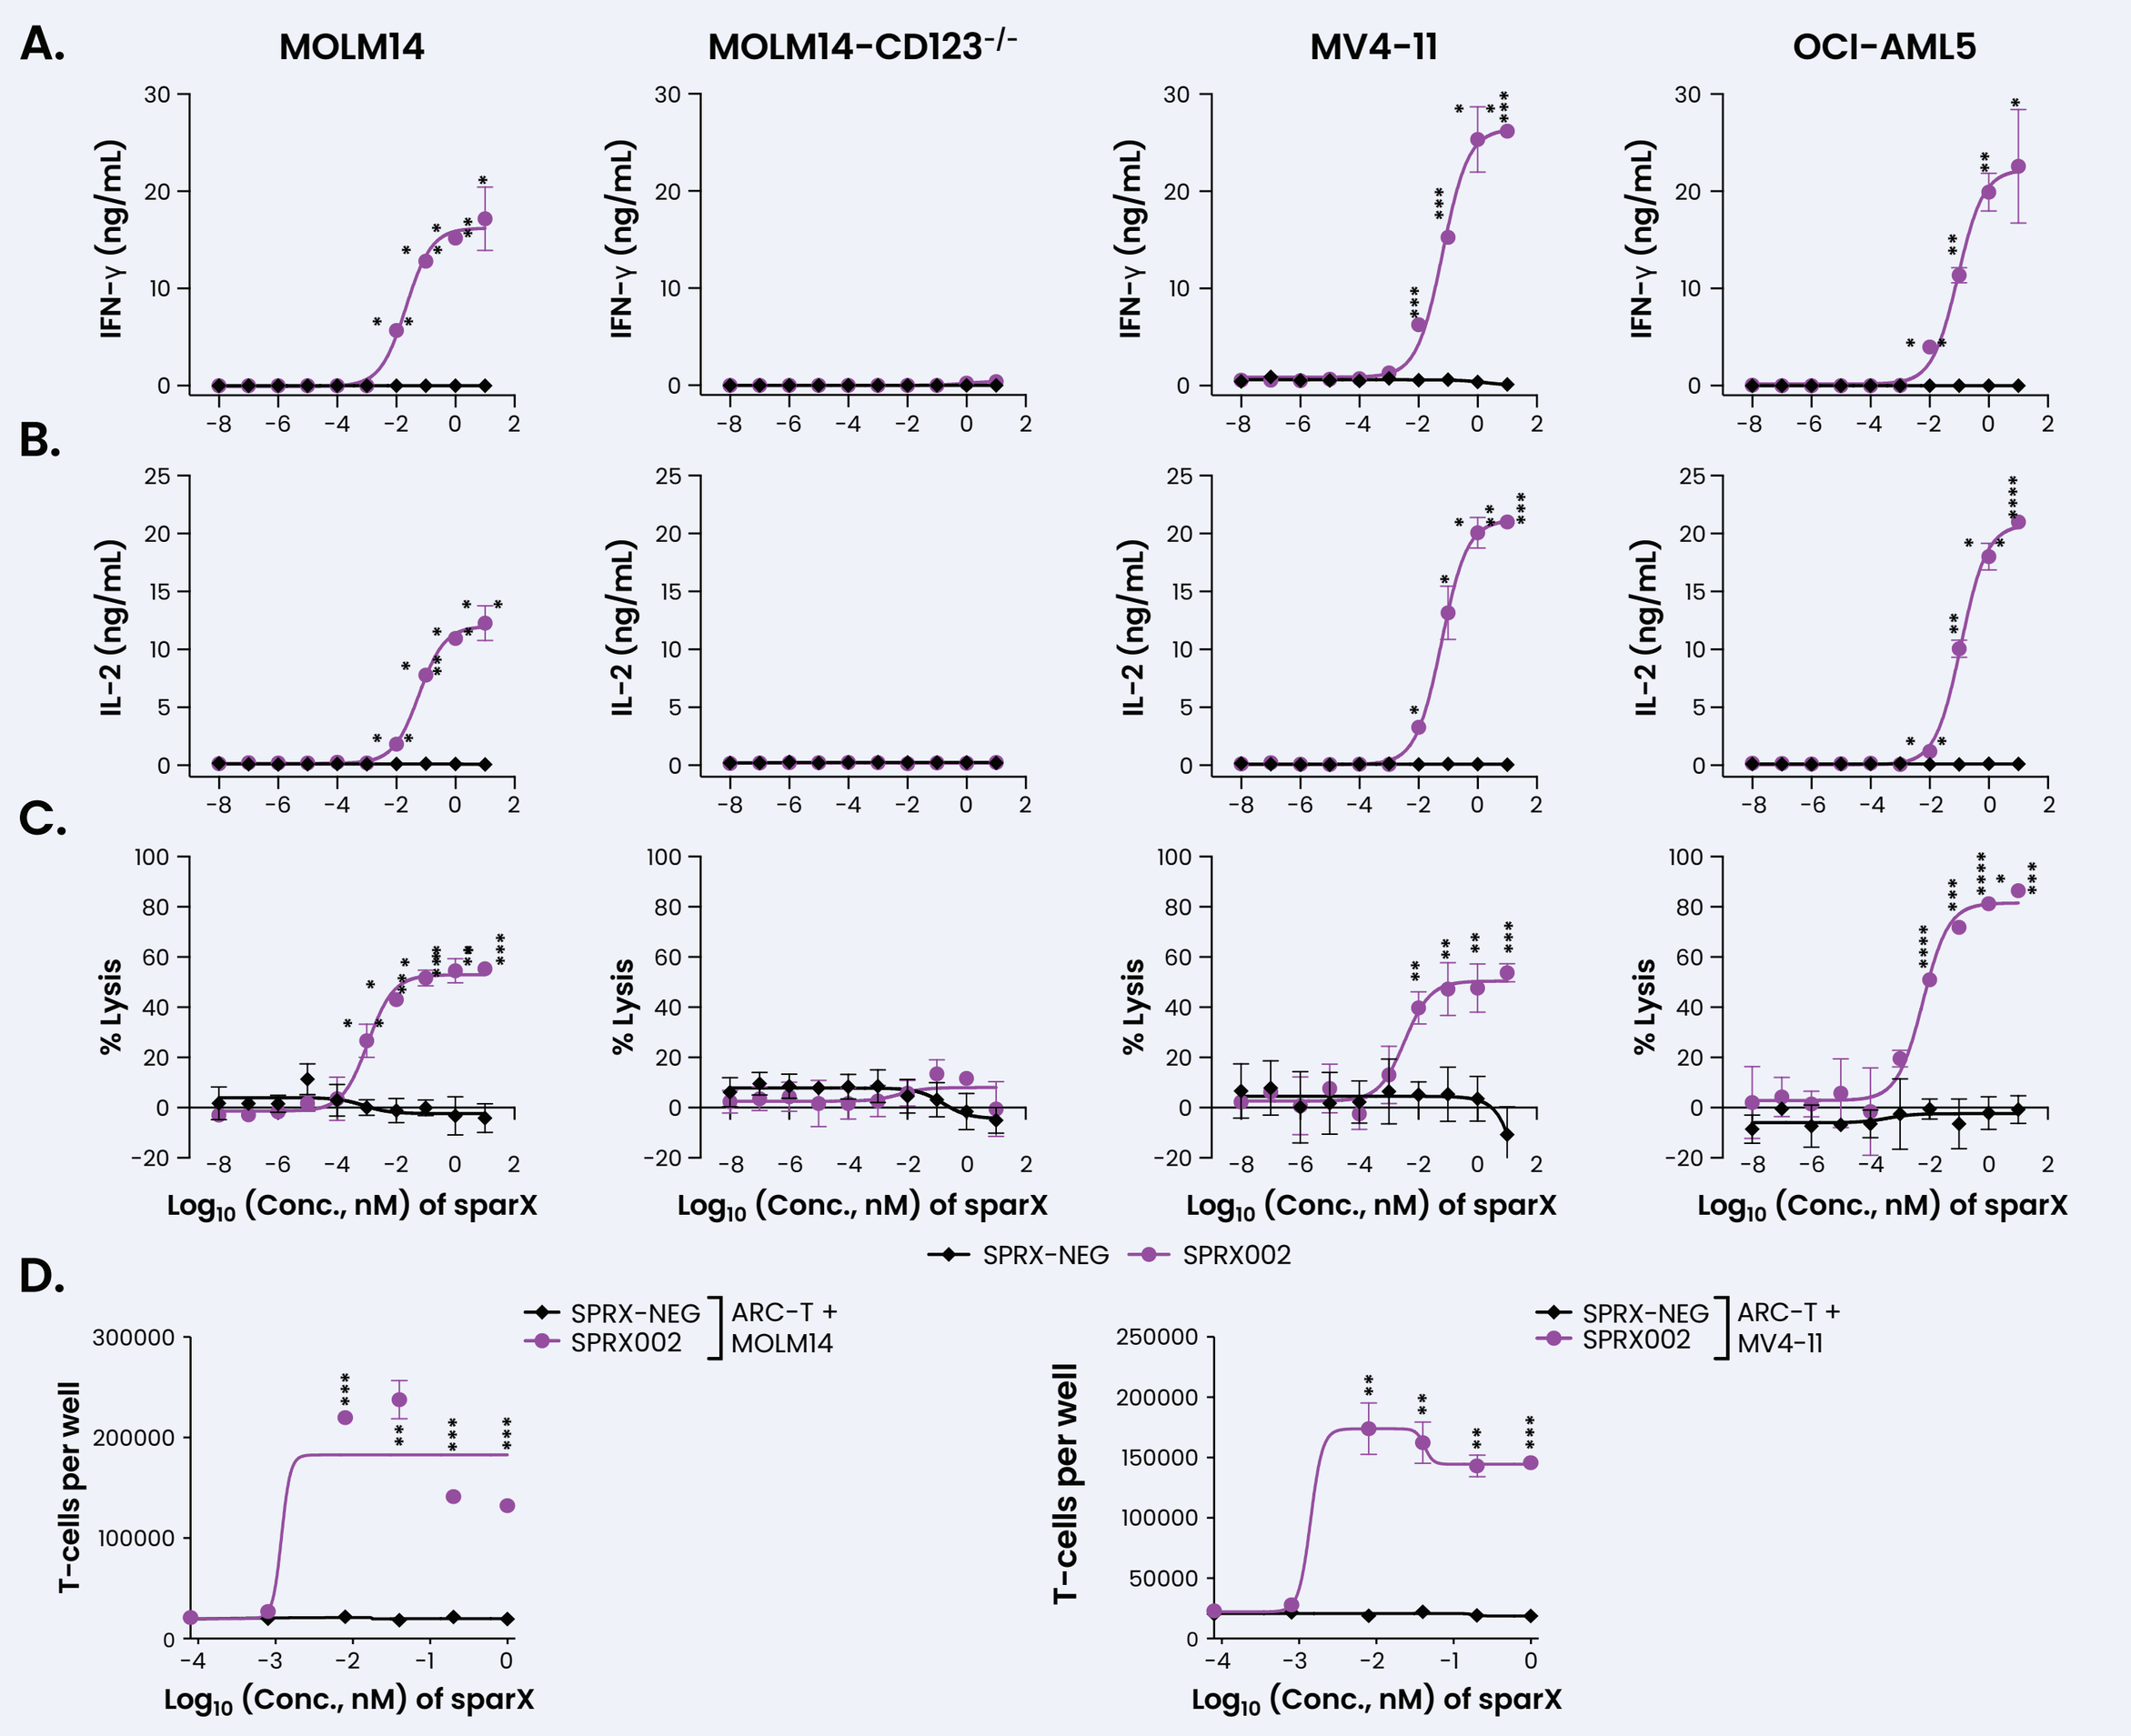

A.
MOLM14
MOLM14-CD123-/-
MV4-11
OCI-AML5
IFN-γ (ng/mL)
B.
IL-2 (ng/mL)
C.
% Lysis
Log10 (Conc., nM) of sparX
SPRX-NEG
SPRX002
D.
T-cells per well
SPRX-NEG
SPRX002
ARC-T + MOLM14
ARC-T + MV4-11

Concentration	MOLM14	MOLM14 KO	MV4-11	OCI-AML5
EC$_{50}$ nM IL-2	0.0580	n.a.	0.0599	0.115
EC$_{50}$ nM IFN-γ	0.0212	n.a.	0.0652	0.090
EC$_{50}$ Cytotoxicity (nM)	0.001	n.a.	0.003	0.006
EC$_{50}$ Proliferation (nM)	0.00141	NT	0.00120	NT

ARC-T cells demonstrate SPRX002 dose-dependent cytokine production and cytotoxic activity of co-cultured CD123-expressing cancer cell lines in vitro. **(A-C)** ARC-T cells were cultured at an E:T ratio of 1:2 with various luciferase enabled CD123+ AML cell lines or the CD123$^{-/-}$ MOLM14-LUC cells and a concentration range of SPRX002 protein or the negative control SPRX-NEG for approximately 16 hours. After overnight culture, the cytokines IFN-γ and IL-2 were measured by ELISA **(A and B, respectively)** and cytotoxicity assessed by remaining luciferase activity **(C)**. **(D)** SPRX002 protein drives proliferation in the presence of CD123+ target cells. ARC-T cells were deprived of IL-2 overnight, then labeled with a proliferation dye. Labeled ARC-T cells were cultured at an E:T ratio of 1:2 with MV4-11 (GFP/LUC) or MOLM14 (GFP/LUC) and a concentration range of SPRX002 protein or the negative control SPRX-NEG for 96 hours. After 96 hours, cell pellets were stained for CD3 and T cells (CD3+/GFP-) were enumerated via high throughput sampler.

ACLX-002 completely regressed disseminated MOLM14 tumors in a schedule and dose-dependent manner without the aid of alloreactivity

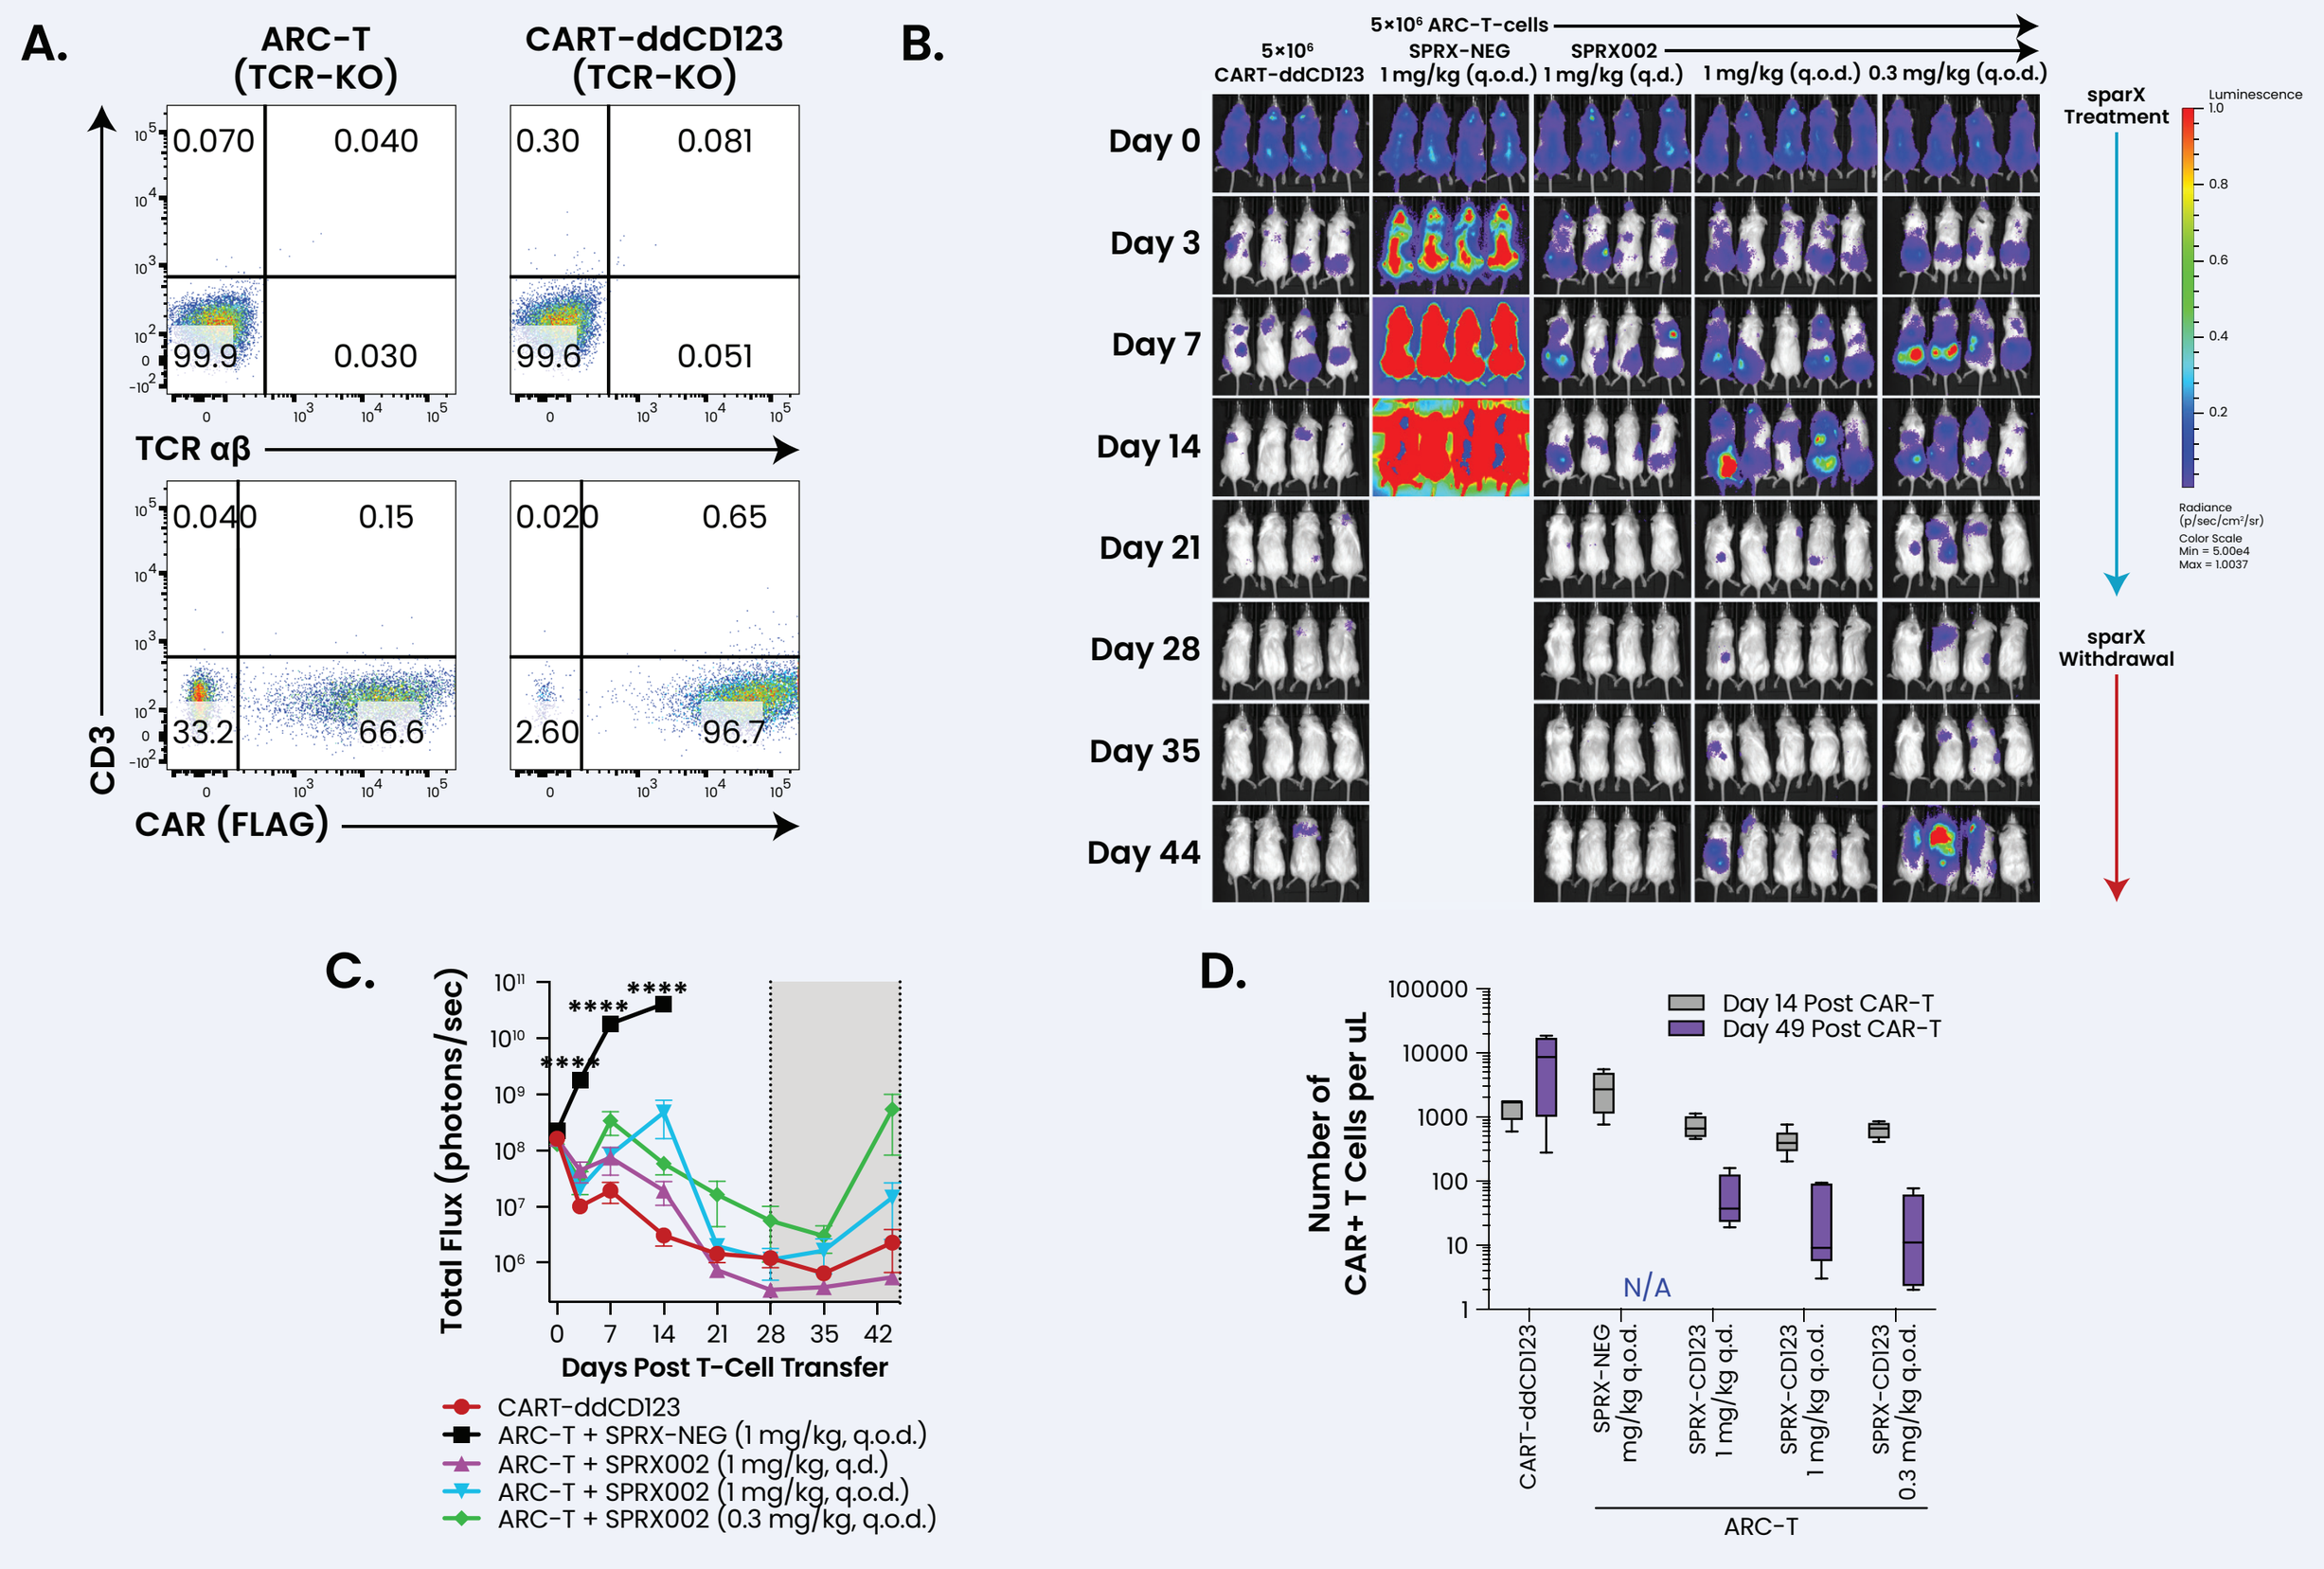

A.
ARC-T (TCR-KO)
CART-ddCD123 (TCR-KO)
TCR αβ
CD3
CAR (FLAG)
B.
Day 0
Day 3
Day 7
Day 14
Day 21
Day 28
Day 35
Day 44
sparX Treatment
sparX Withdrawal
C.
Total Flux (photons/sec)
Days Post T-Cell Transfer
CART-ddCD123
ARC-T + SPRX-NEG (1 mg/kg, q.o.d.)
ARC-T + SPRX002 (1 mg/kg, q.d.)
ARC-T + SPRX002 (1 mg/kg, q.o.d.)
ARC-T + SPRX002 (0.3 mg/kg, q.o.d.)
D.
Number of CAR+ T Cells per uL
Day 14 Post CAR-T
Day 49 Post CAR-T
N/A

ACLX-002 completely regresses disseminated MOLM14 tumors in a schedule and dose-dependent manner without the aid of alloreactivity. **(A)** Generation of native TCR-deficient CAR-T cells: T cells were transduced with lentiviral vectors encoding the ARC-T receptor and a LentiCRISPRv2 construct encoding a gRNA targeting the constant region of the TCR β-chain. After 7 days of expansion remaining CD3+/TCRαβ+ T cells were depleted and CD3- cells were expanded. Prior to cell transfer, T cells were stained for CD3, TCR, and CAR (FLAG). **(B-D)** ACLX-002 treatment in the disseminated AML model MOLM14: NSG mice were engrafted with MOLM14 cells (1×10^6) for 6 days prior to CAR-T transfer. Mice then received 5×10^6 CD3-deficient CD123-targeting CAR-T (CART-ddCD123) or ARC-T cells. Groups receiving ARC-T cells were then treated with SPRX-NEG or SPRX002 daily (q.d.) or every other day (q.o.d.) for 28 days. Animals were monitored for tumor burden via IVIS during treatment or after SPRX002 withdrawal **(B)**. **(C)** Imaging quantitation from data presented in **(B)**. Shaded area indicates SPRX002 withdrawal. **(D)** CAR-T cells were quantitated from 200 µL of whole blood at days 14 and 49 post-T cell transfer.

ACLX-002 completely regressed disseminated MV4-11 tumors in a dose-dependent manner and performs comparably to a CD123-targeting CAR

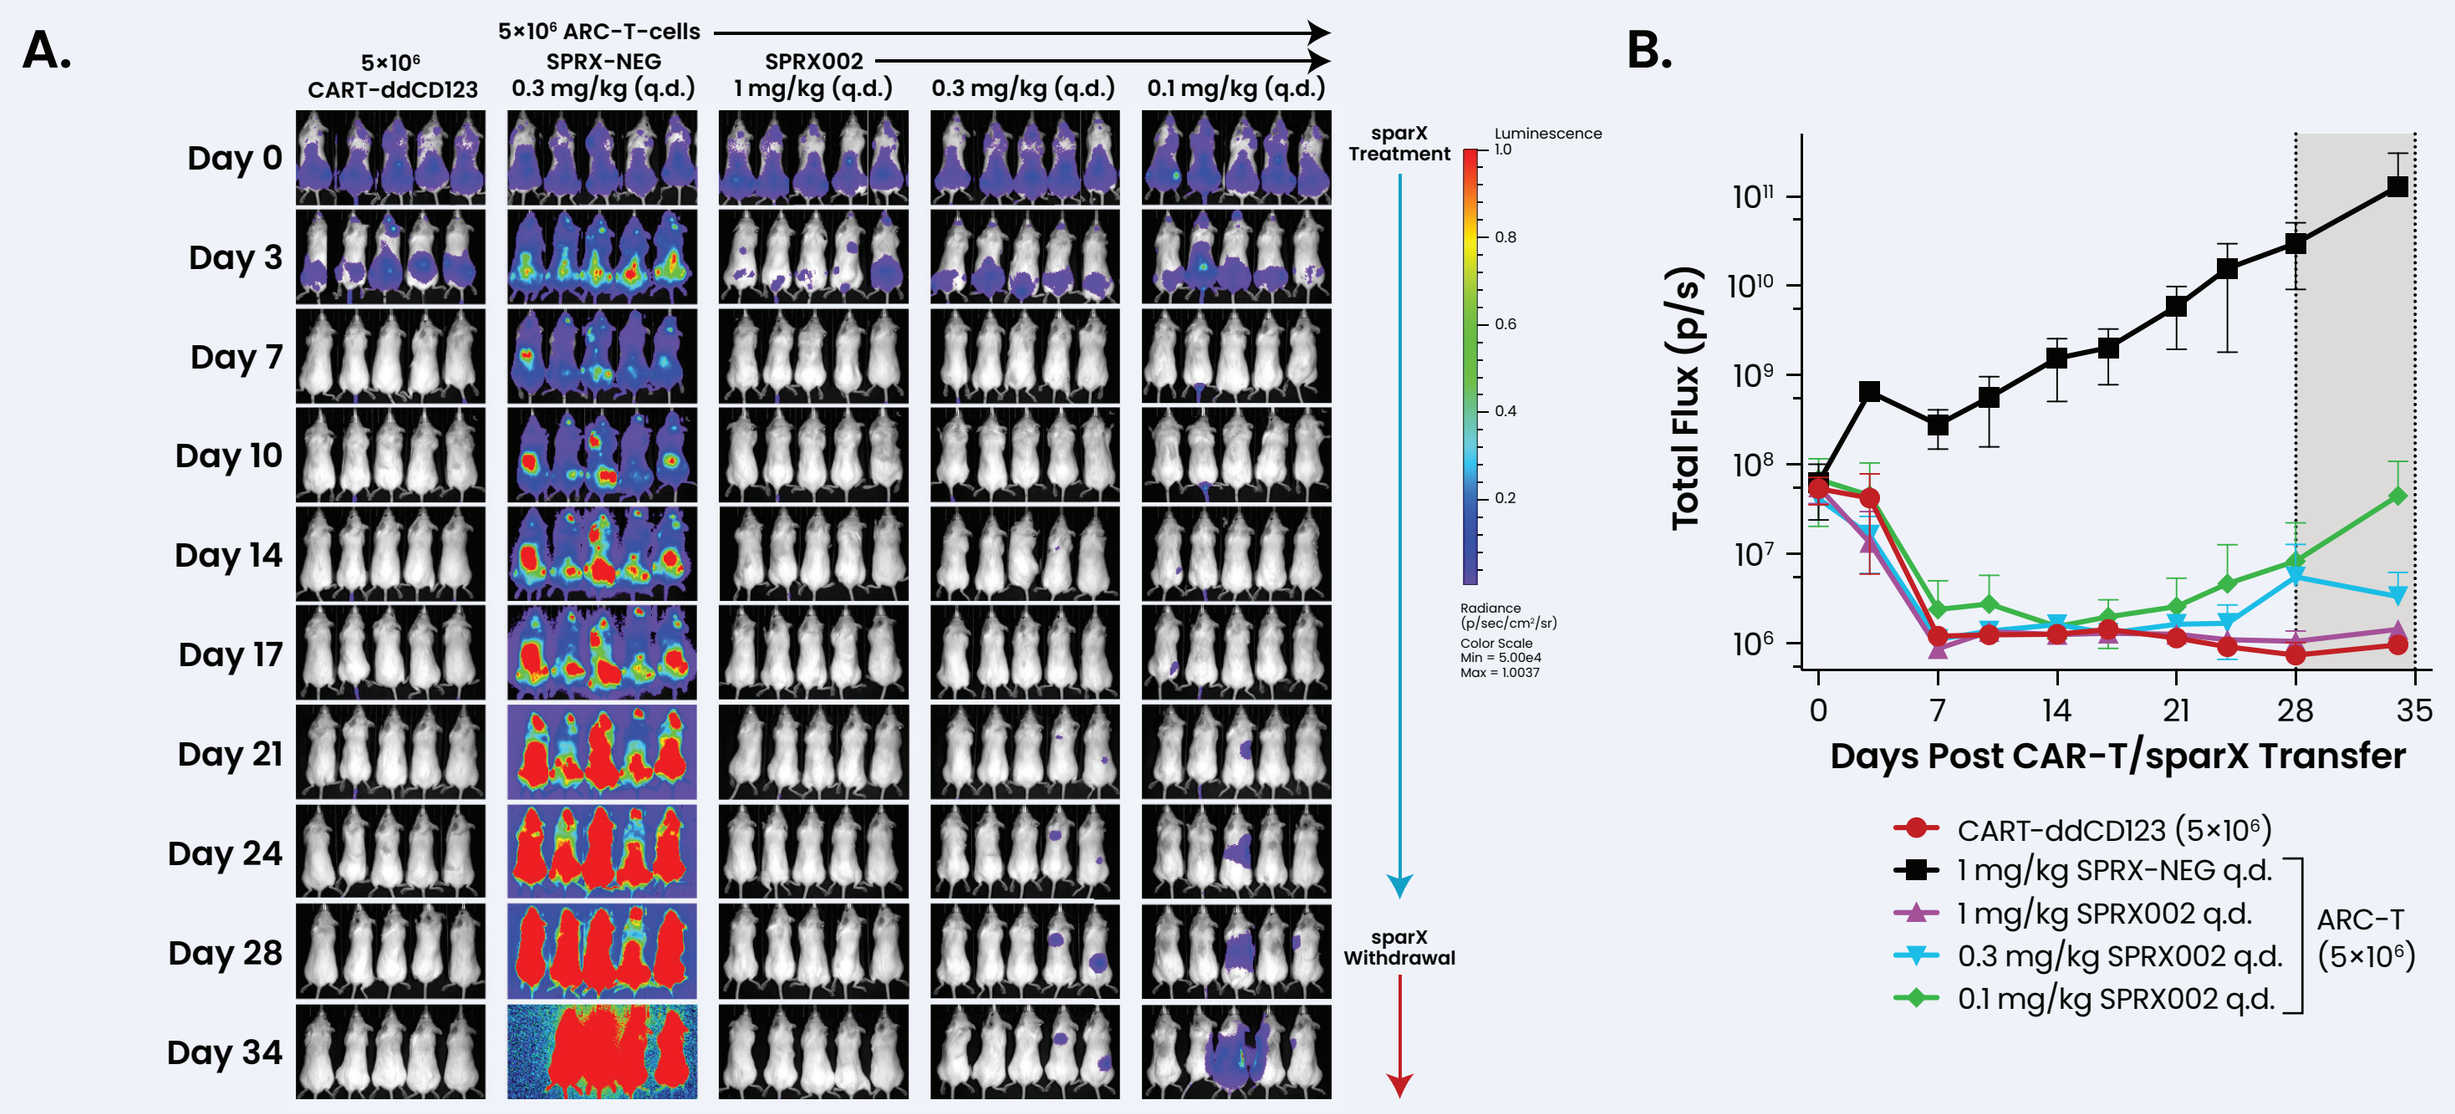

A.
5×10⁶ CART-ddCD123
5×10⁶ ARC-T-cells
SPRX-NEG 0.3 mg/kg (q.d.)
SPRX002 1 mg/kg (q.d.)
0.3 mg/kg (q.d.)
0.1 mg/kg (q.d.)
Day 0
Day 3
Day 7
Day 10
Day 14
Day 17
Day 21
Day 24
Day 28
Day 34
sparX Treatment
sparX Withdrawal
B.
Total Flux (p/s)
Days Post CAR-T/sparX Transfer
CART-ddCD123 (5×10⁶)
1 mg/kg SPRX-NEG q.d.
1 mg/kg SPRX002 q.d.
0.3 mg/kg SPRX002 q.d.
0.1 mg/kg SPRX002 q.d.
ARC-T (5×10⁶)

ACLX-002 completely regressed disseminated MV4-11 tumors in a dose-dependent manner and performs comparably to a CD123-targeting CAR. **(A-B)** ACLX-002 treatment in the disseminated AML model MV4-11: NSG mice were engrafted with MV4-11 cells (5×10^6) for 15 days prior to CAR-T transfer. Mice then received 5×10^6 CD123-targeting CAR-T (CART-ddCD123) or ARC-T cells. Groups receiving ARC-T cells were then treated with SPRX-NEG or SPRX002 daily (q.d.) for 28 days. Animals were monitored for tumor burden via IVIS during treatment or after SPRX002 withdrawal **(A)**. **(B)** Imaging quantitation from data presented in **(A)**. Shaded area indicates SPRX002 withdrawal.

The expression of CD123 on activated T cells does not impact the activity of ACLX-002 in vivo

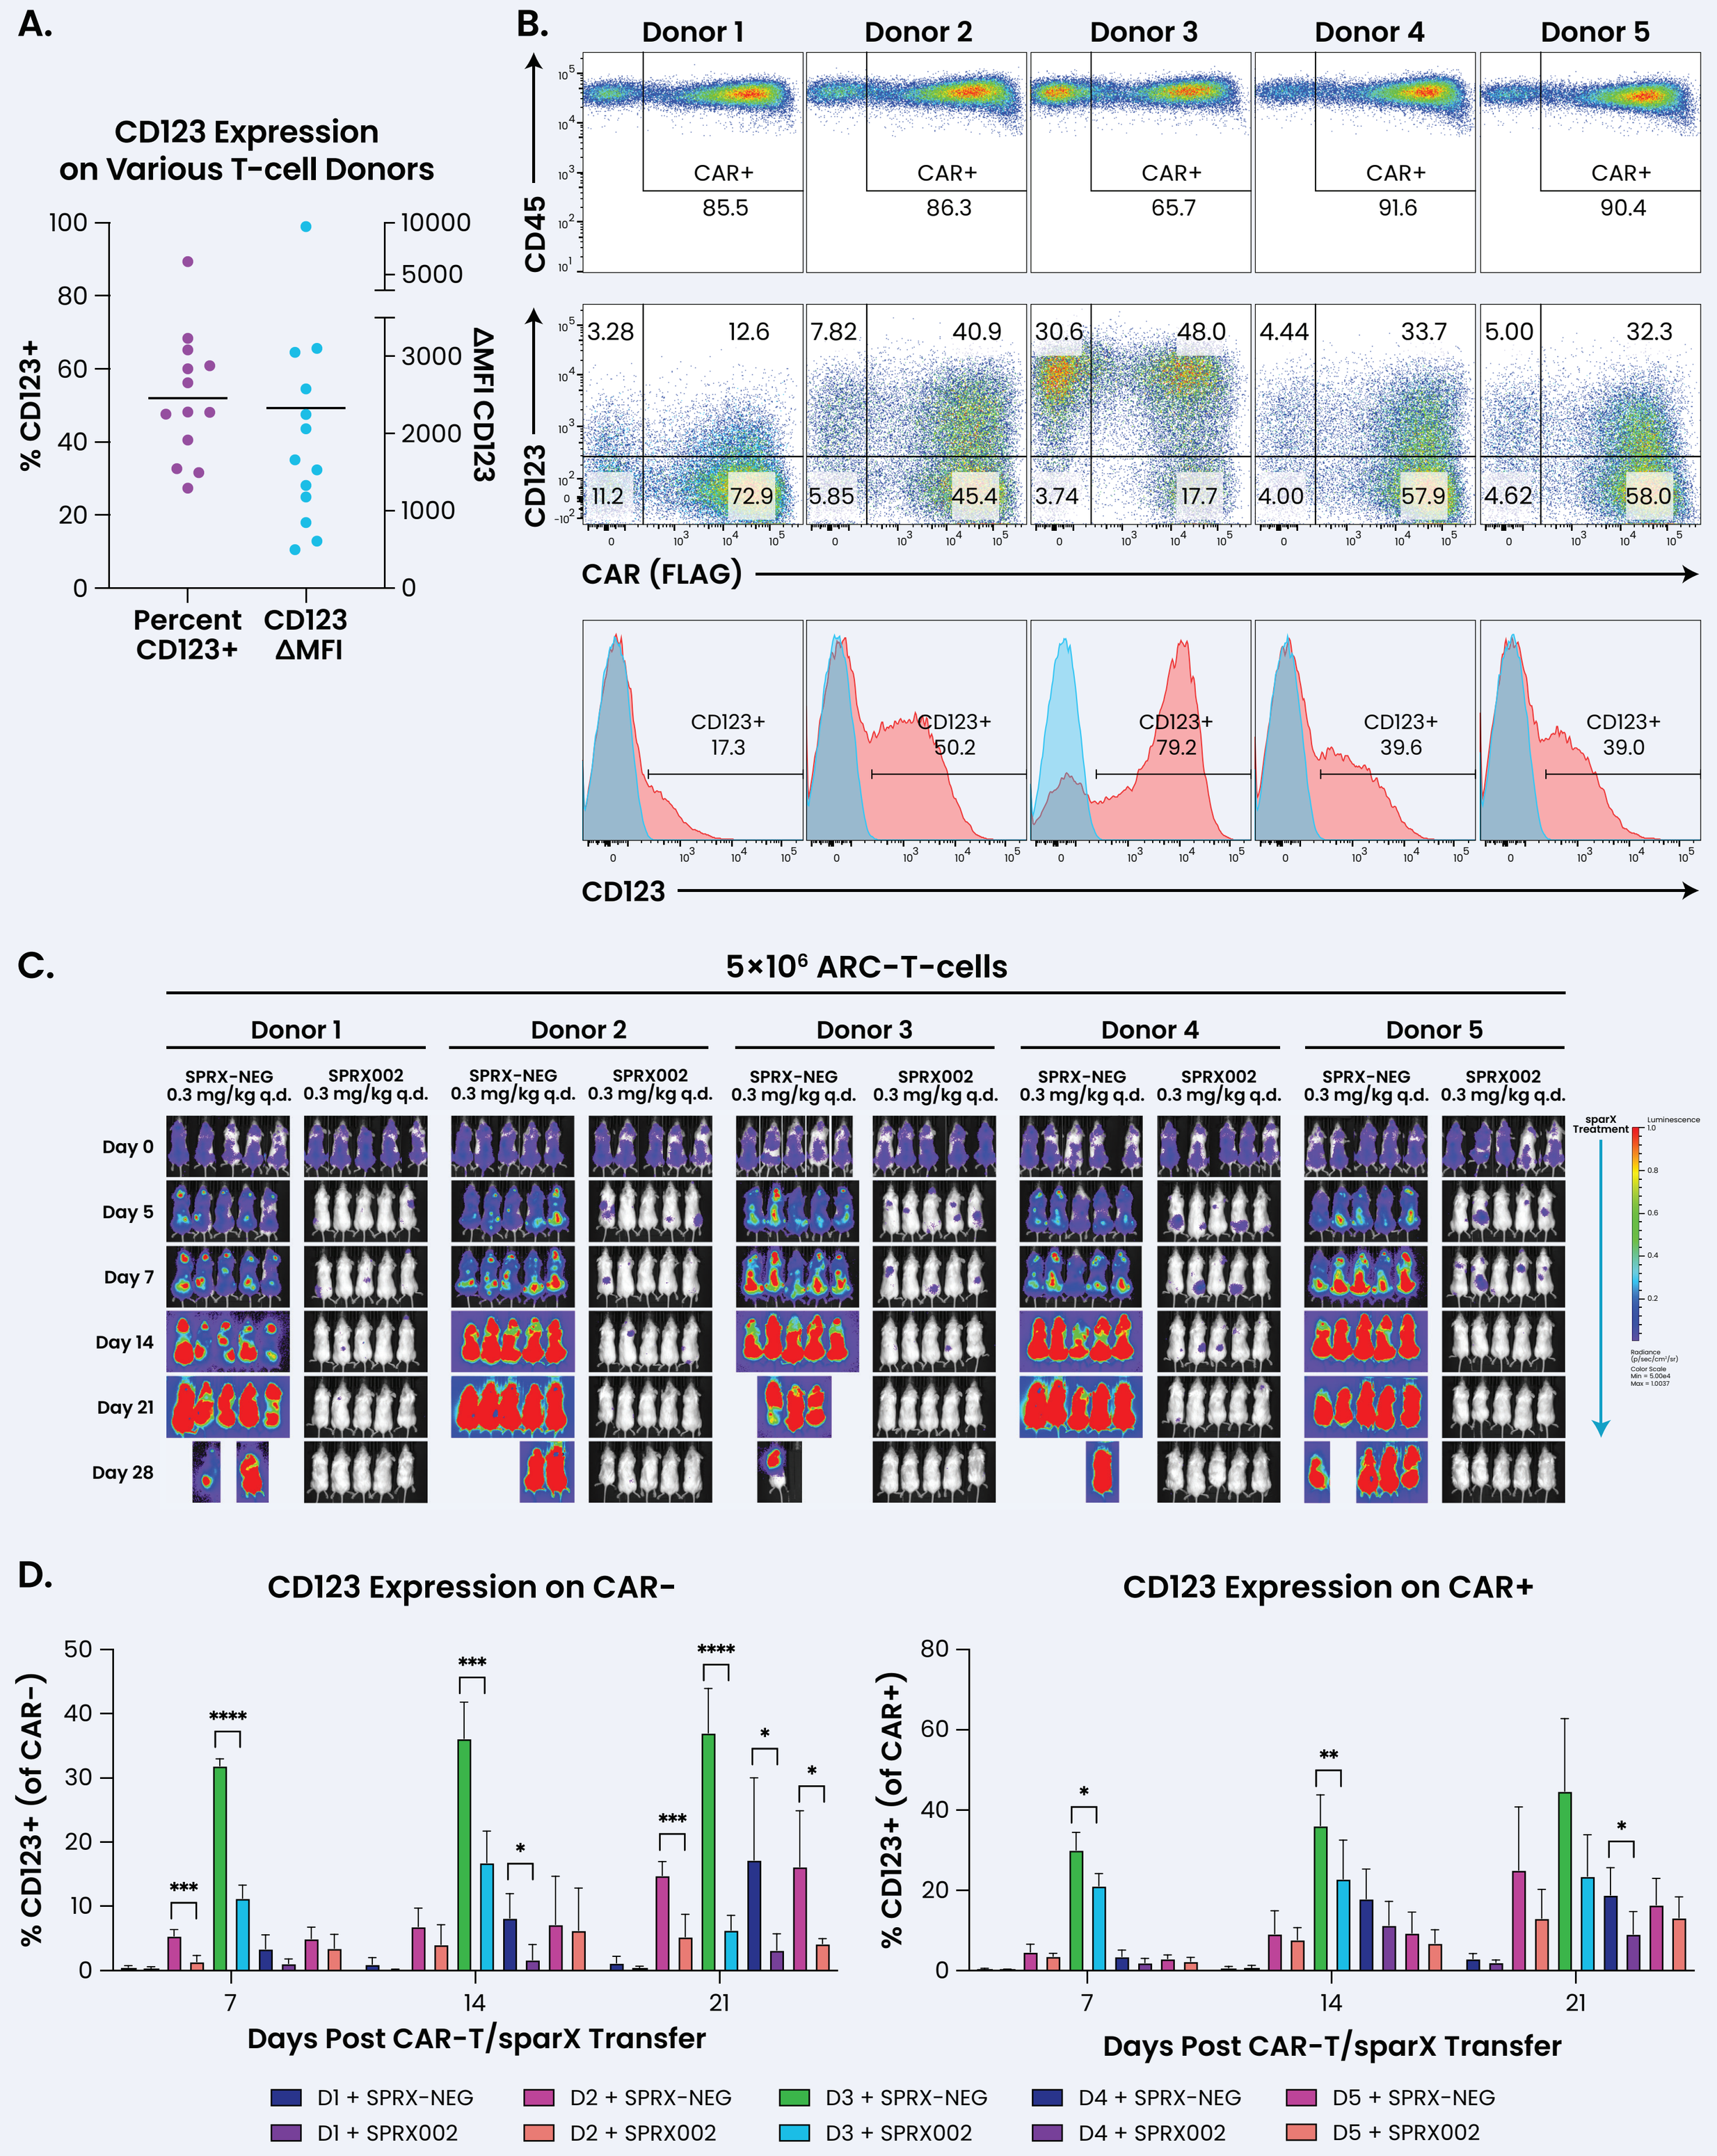

A.
CD123 Expression on Various T-cell Donors
% CD123+
ΔMFI CD123
Percent CD123+
CD123 ΔMFI
B.
Donor 1
Donor 2
Donor 3
Donor 4
Donor 5
CD45
CAR+
CD123
CAR (FLAG)
C.
5×10⁶ ARC-T-cells
SPRX-NEG 0.3 mg/kg q.d.
SPRX002 0.3 mg/kg q.d.
D.
CD123 Expression on CAR-
CD123 Expression on CAR+
% CD123+ (of CAR-)
% CD123+ (of CAR+)
Days Post CAR-T/sparX Transfer
D1 + SPRX-NEG
D1 + SPRX002
D2 + SPRX-NEG
D2 + SPRX002
D3 + SPRX-NEG
D3 + SPRX002
D4 + SPRX-NEG
D4 + SPRX002
D5 + SPRX-NEG
D5 + SPRX002

The expression of CD123 on activated T cells does not impact the activity of ACLX-002 in vivo. **(A)** CD123 is expressed on activated and expanded human T cells. ARC-T cells were generated from 13 normal healthy donors and CD123 percent expression **(left)** and intensity **(right, MFI)** was determined on day 7 post-T cell activation. **(B)** 5 normal healthy donors were chosen based on their CD123 staining intensity to be used to generate ARC-T cells. ARC-T cell expression of the CAR (FLAG) and CD123 were assessed prior to their use in vivo **(C-D)**. ACLX-002 treatment in the disseminated AML model MOLM14: NSG mice were engrafted with MOLM14 cells (1×10^6) for 6 days prior to CAR-T transfer. Mice then received 5×10^6 ARC-T cells derived from the donors in **(B)**. Animals were then treated with SPRX-NEG or SPRX002 daily 28 days at 0.3 mg/kg. Animals were monitored for tumor burden via IVIS **(C)**. **(D)** SPRX002 consistently reduces the proportion of CD123-expressing CAR-T cells, while having a marginal effect on CAR+ T cells. CD123-expression was monitored on circulating CAR- **(left)** and CAR+ **(right)** T cells on days 7, 14, and 21 post-transfer and sparX protein administration. Where indicated, SPRX002 significantly reduced the proportion of CD123-expressing CAR- or CAR+ cells.

ACLX-002 clears or controls multiple patient-derived AML xenografts

Model	Recipient Strain	Patient Characteristics					Disease Characteristics		
		Age	Gender	Ethnicity	Diagnosis	Treatment History	FAB Classification	Cytogenetics	Mutational Status
CTG-2239	NOG	79	Male	Black or African American	De novo	Naive	NOS	Normal	IDH1/2 WT FLT3 N/A NPM mutant
CTG-2229	NCG	53	Male	Caucasian	Refractory	Pretreated	M1 (without maturation)	46XY, del(2)(p13p23), t(4;13)(q31;q34), add(4)(q25), del(6)(q13q25), t(9;22)(q34;q11.2), del(10)(q24), add(16)(q24) [20]	IDH1 mutant (R132C) FLT3 WT NPM WT
CTG-3438	NOG-EXL	62	Female	Caucasian	Relapsed	Pretreated	M4 (myelomonocytic)	Normal	IDH1/2 WT FLT3 WT NPM WT
CTG-2227	NOG	59	Female	Caucasian	Relapsed	Pretreated	M4 (myelomoncytic)	Not available	IDH1 mutant FLT3 ITD mutant NPM mutant
CTG-2240	NOG	75	Male	Not available	De novo	Naïve	AML (11q23 abnormalities)	46, XY, t(9;11)(p22;q23), add(10)(q24) [5]/46, XY [5]	IDH1/2 WT FLT3 WT NPM WT
CTG-2456	NOG	NAV	Female	Caucasian	De novo	Pretreated	NOS	46, XX, del(7)(q22q36) [15]	IDH1/2 WT FLT3 WT NPM WT

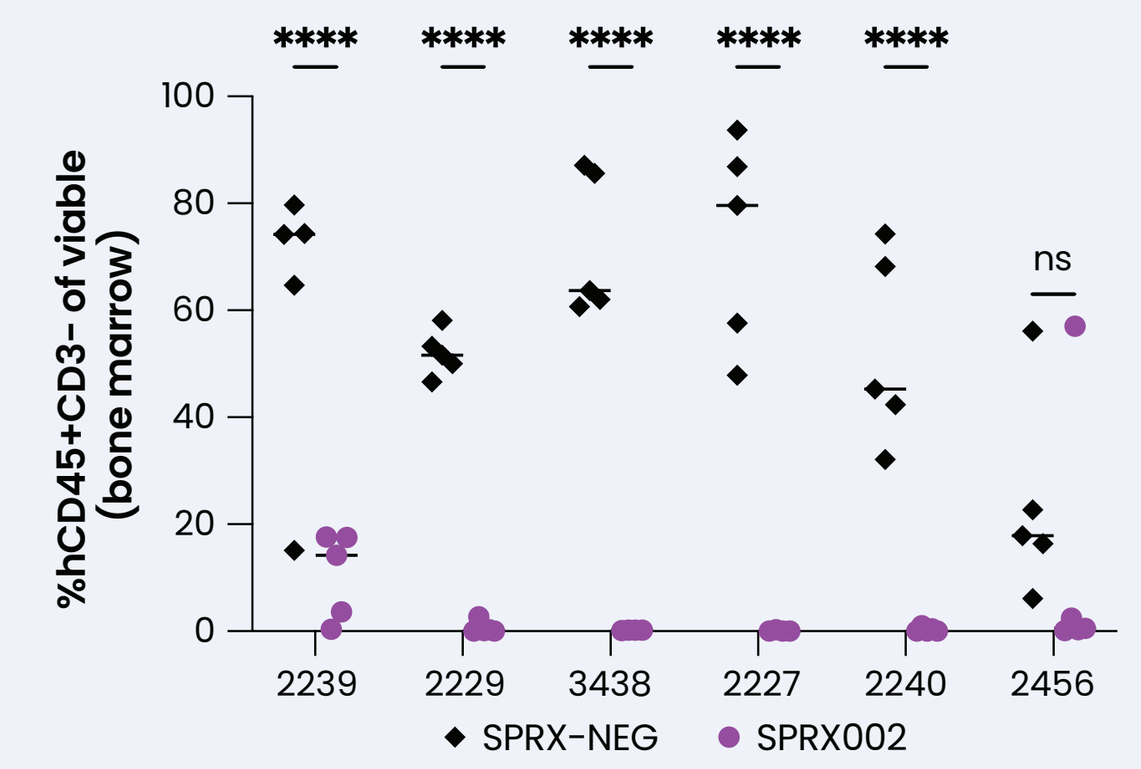

%hCD45+CD3- of viable (bone marrow)

ns
2239
2229
3438
2227
2240
2456
SPRX-NEG
SPRX002

ACLX-002 clears or controls multiple patient-derived AML xenografts (PDX). Generation of AML-PDX models: Prior to inoculation with AML cells, animals were sub-lethally irradiated with 150 cGy whole body irradiation, followed by the transfer of 2×10^6 human T-cell depleted AML cells derived from cryopreserved specimens of patient leukapheresis. The recipient strain (NOG, NOG-EXL, and NCG) was previously established and is indicated in the table above, along with patient and disease characteristics. Model engraftment kinetics were previously established to define a time window in which animals exhibited human tumor cell engraftment in the bone marrow of 20% or greater. For each model, a surrogate cohort of animals (n=3-5) were sacrificed at intermittent time points prior to the estimated engraftment window to collect bone marrow for AML burden analysis. Once surrogate animals had achieved engraftment criterion in bone marrow, defined as %hCD45+ of viable cells ≥ 20% on average of animals sampled, the remainder of pre-study animals were randomized based on body weight for treatment initiation. All on-study animals received a single dose of ARC+ T cells generated using GMP-quality vector and healthy-donor T cells, followed by once daily IP injections of either SPRX-NEG or SPRX002 at 3 mg/kg for 14 days. On day 14 after treatment initiation, animals from all groups were euthanized to collect bone marrow for flow cytometry analysis. The proportion of the live cells within the bone marrow which represented engrafted AML cells (hCD45+CD3-) was determined for all on-study animals.

Conclusions

- ACLX-002 is a novel 2-part therapy for the treatment of acute myeloid leukemia, consisting of a monovalent CD123-targeting SPRX002 protein and ARC-T cells
- SPRX002 specifically binds to CD123+ cells including CD123+ primary AML samples
- ARC-T cells bind SPRX002 through its anti-tag domain, while SPRX002 maintains CD123 binding capacity
- ARC-T cells demonstrate SPRX002 dose-dependent cytokine production, T cell proliferation, and cytotoxic activity against co-cultured CD123-expressing cancer cell lines
- ACLX-002 completely regressed disseminated MOLM14 and MV4-11 tumors in a schedule and dose-dependent manner
 - Without the aid of alloreactivity
 - Performs similarly to a traditional CD123-targeting D-domain based CAR
- Activated T cells and CAR-T cells express CD123 with a wide range of donor-to-donor variability
 - CD123 expression on T cells did not impact the activity of ACLX-002 in vivo
 - SPRX002 dosing reduced the proportion of CD123-expressing T cells, primarily in the CAR-negative fraction in donors with high CD123 expression
- ACLX-002 clears or controls multiple patient-derived AML xenografts

*The data in this poster were generated with related but distinct reagents from the ACLX-002 clinical reagents and are meant to support the clinical development of this product.

Presented at the American Association for Cancer Research 2022 Annual Meeting, April 8–13, 2022

©2022 Arcellx, Inc. All rights reserved.