ACLX-001*, a novel BCMA-targeted CAR-T cell therapy that can be activated and silenced in vivo with soluble protein adapters in a dose dependent manner

Abstract 1549

Justin P. Edwards, Janine Buonato, David LaFleur, Jeffrey Swers, Jenny Mu, Liubov Zaritskaya, Sinnie Ng, Ankit Gupta, Hui Wang, David Hilbert, and David Tice

Arcellx, Inc., Gaithersburg, MD

JEdwards@arcellx.com

Introduction

The ARC-sparX Platform and ACLX001*

The ARC-sparX platform separates the antigen-recognition and killing functions of conventional CAR-T therapy and is comprised of two key components:

1. SPRX001 (**s**oluble **p**rotein **a**ntigen-**r**eceptor **X**-linker) protein: binds BCMA on diseased cells and flags those cells for destruction.
2. ARC-T Cells (**A**ntigen **R**eceptor **C**omplex **T** Cells): bind and kill SPRX001 flagged cells.
3. ACLX001 is the combination of the ARC-T cells and SPRX001.

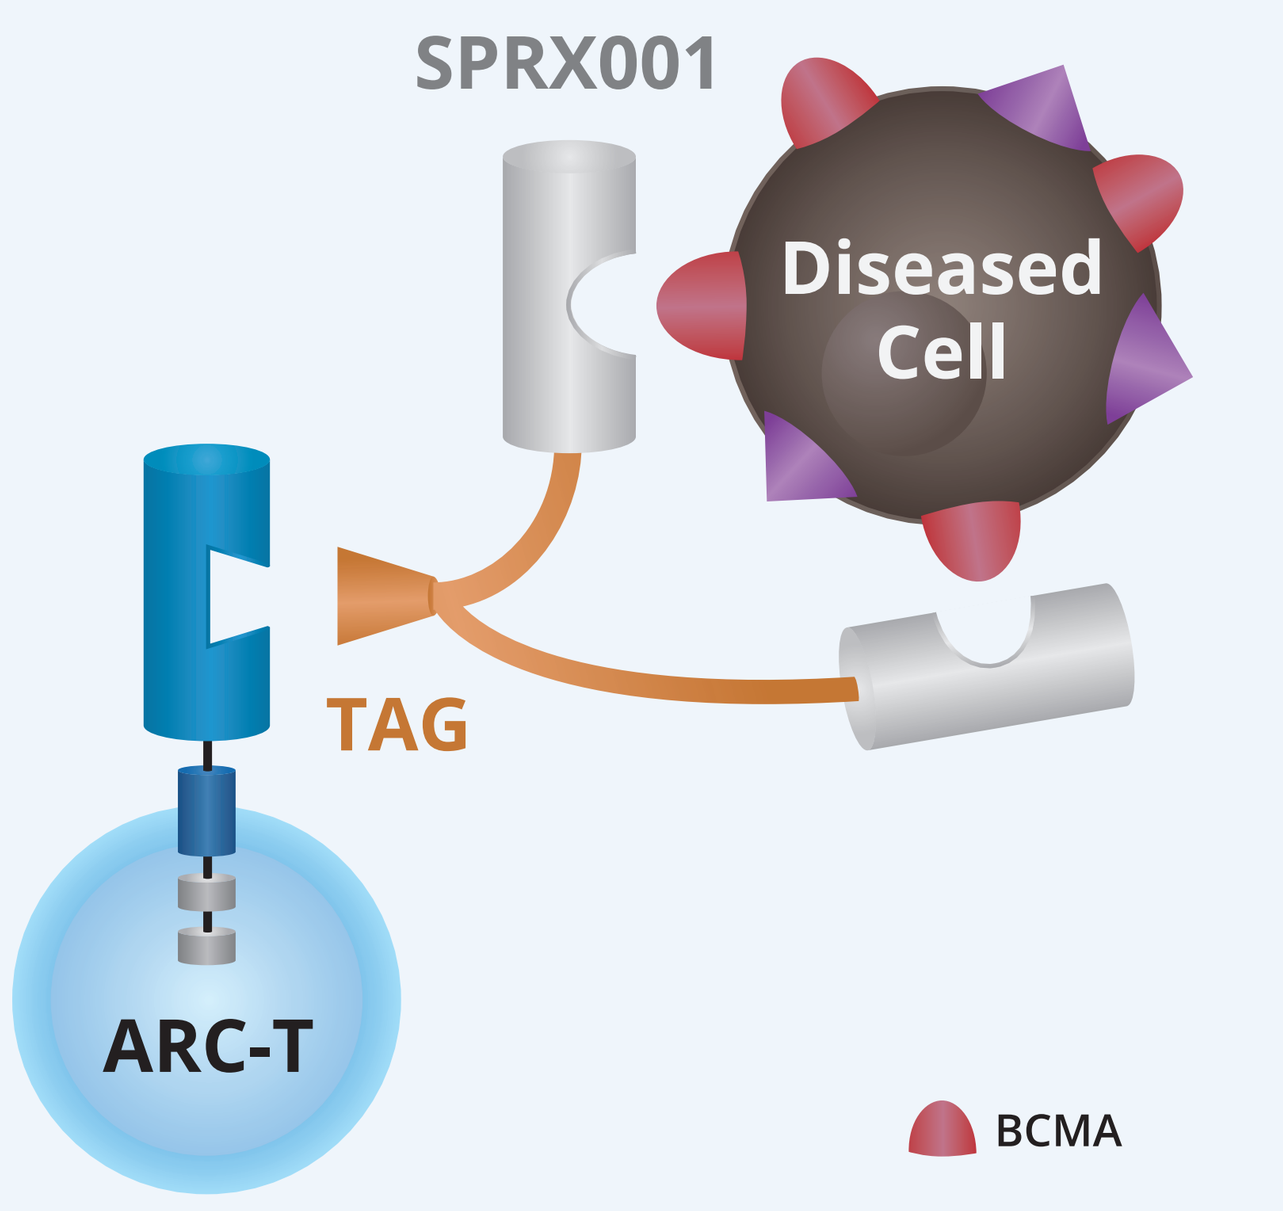

SPRX001
Diseased Cell
TAG
ARC-T
BCMA

SPRX001 protein

- No inherent therapeutic activity
- Contains two BCMA-targeting domains
- Binds BCMA+ multiple myeloma cells
- Engineered to minimize immunogenic potential
- "TAG" is a fragment of human alpha fetoprotein (AFP)

ARC-T cell

- CAR is comprised of the extracellular novel binding domain fused to CD8α spacer/transmembrane region fused to 4-1BB/CD3ζ signaling domains
- Only activated upon formation of a tricomplex of ARC-T + SPRX001 + BCMA positive multiple myeloma cells
- Same viral vector regardless of antigen target

Results

ACLX001 demonstrates dose- and target-dependent cytokine production, degranulation, cytotoxic activity, and proliferation

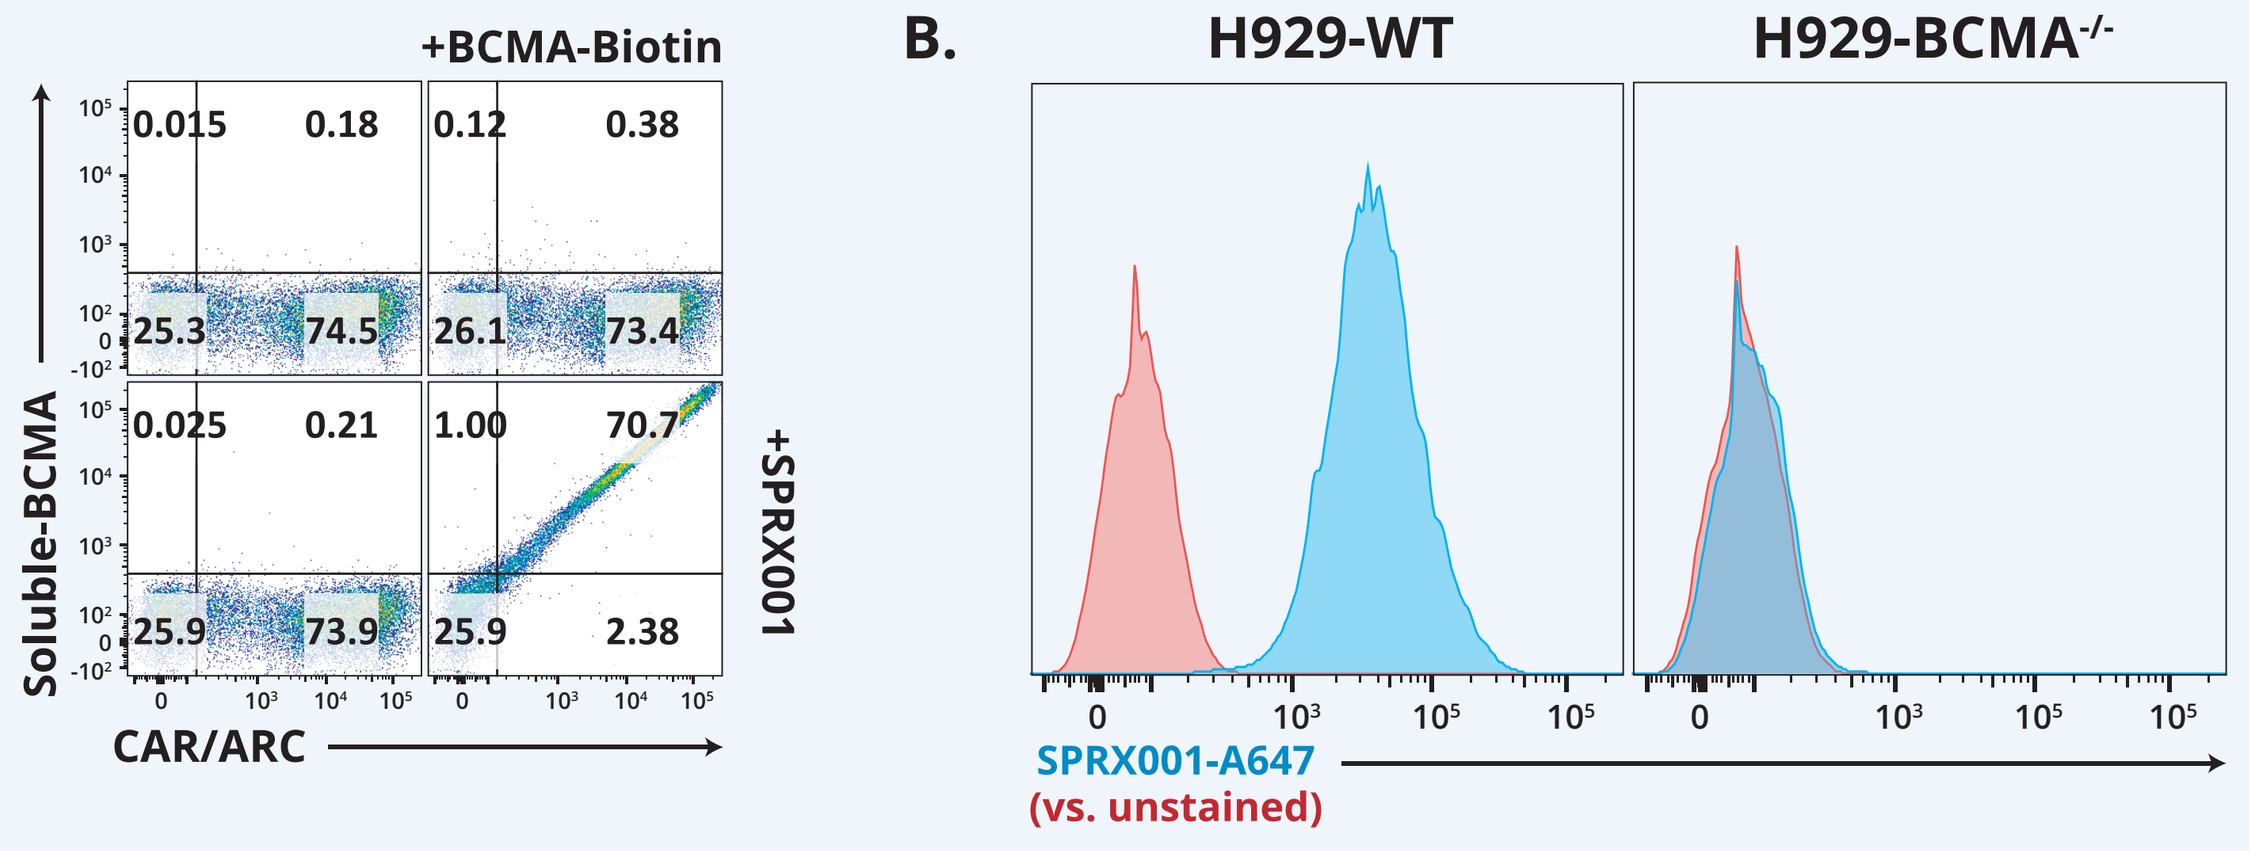

A.
+BCMA-Biotin
+SPRX001
Soluble-BCMA
CAR/ARC
B.
H929-WT
H929-BCMA-/-
SPRX001-A647
(vs. unstained)

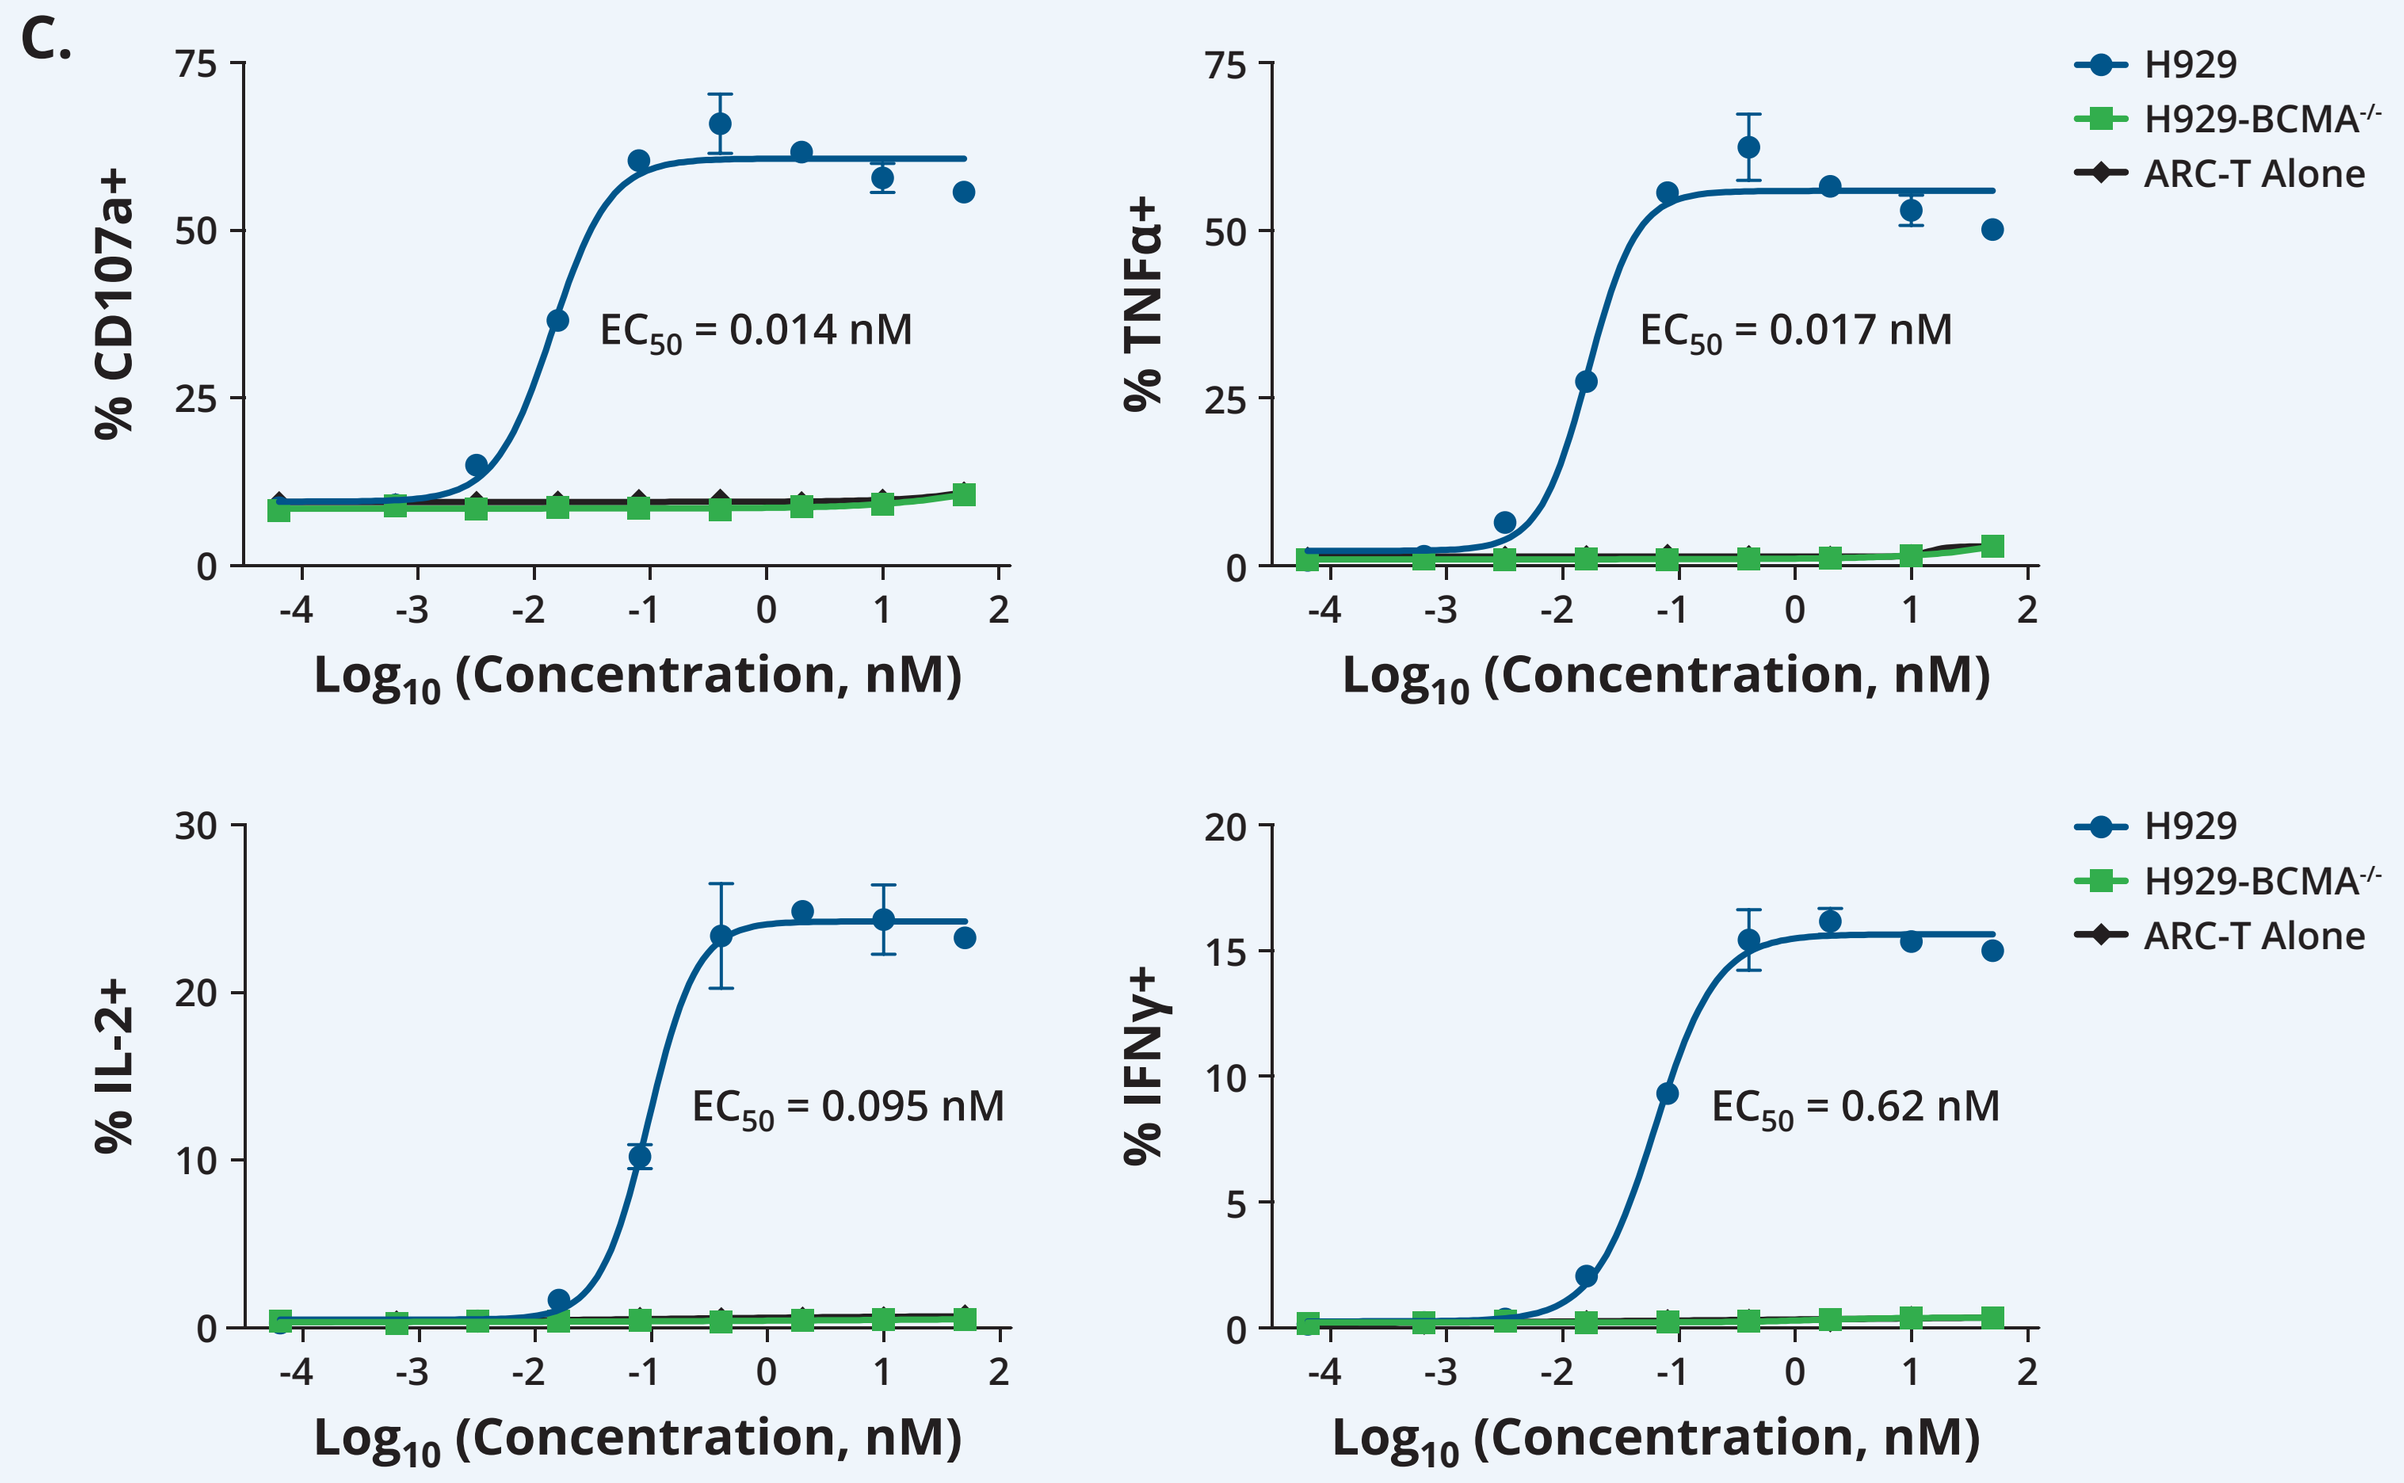

C.
% CD107a+
EC50 = 0.014 nM
% TNFα+
EC50 = 0.017 nM
% IL-2+
EC50 = 0.095 nM
% IFNγ+
EC50 = 0.62 nM
H929
H929-BCMA-/-
ARC-T Alone
Log10 (Concentration, nM)

A. SPRX001 binding to the ARC-T domain confers the ability to engage with BCMA. ARC-T cells were sequentially incubated with SPRX001, BCMA-His-AviTag, then streptavidin-PE to detect binding of BCMA. An anti-FLAG antibody was used in parallel to detect ARC expression.

B. SPRX001 specifically binds BCMA-expressing cells. NCI-H929 cells (wild-type or BCMA-CRISPR-KO) were directly stained with AlexaFluor647 labeled SPRX001.

C. The trimolecular complex of BCMA-SPRX001-ARC induces a cytokine and degranulation response from ARC-T cells. ARC-T cells were cultured with increasing concentrations of SPRX001, with anti-CD107A antibody and monensin for 5 hours in the presence or absence of H929-WT or H929-BCMA$^{-/-}$. Cells were then subject to fixation/intracellular staining for IL-2, IFN-γ, and TNF-α.

ACLX001 demonstrates dose- and target-dependent cytokine production, degranulation, cytotoxic activity, and proliferation

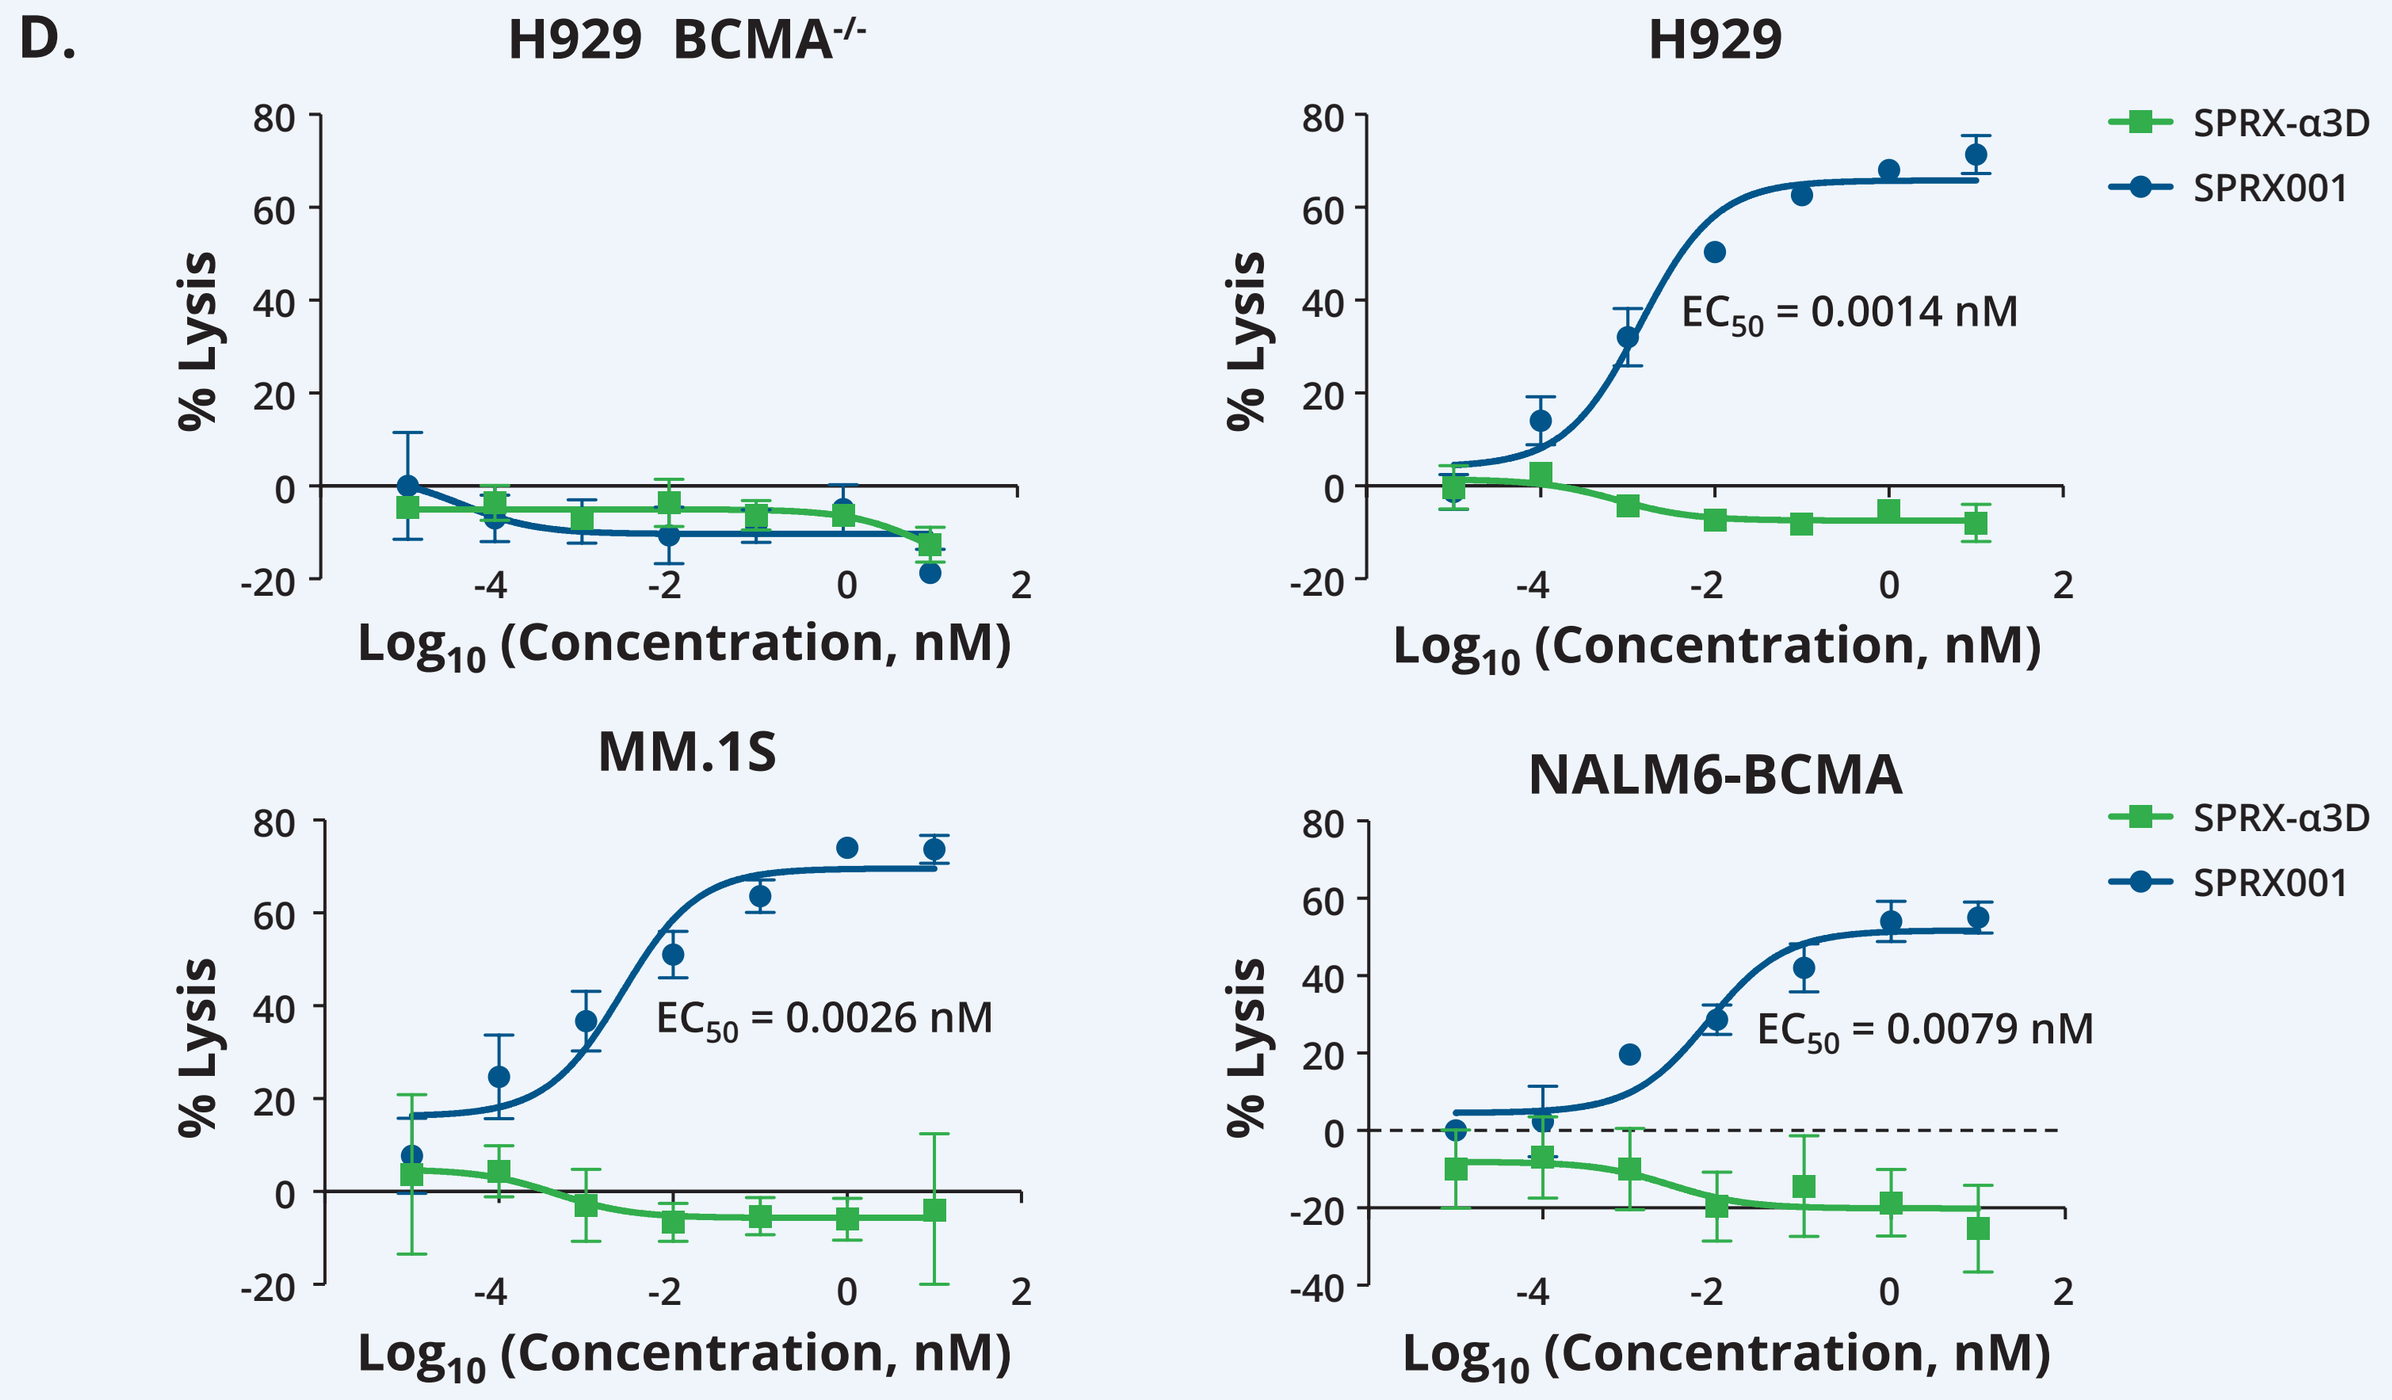

D.
H929 BCMA-/-
H929
EC50 = 0.0014 nM
MM.1S
EC50 = 0.0026 nM
NALM6-BCMA
EC50 = 0.0079 nM
SPRX-α3D
SPRX001
% Lysis
Log10 (Concentration, nM)

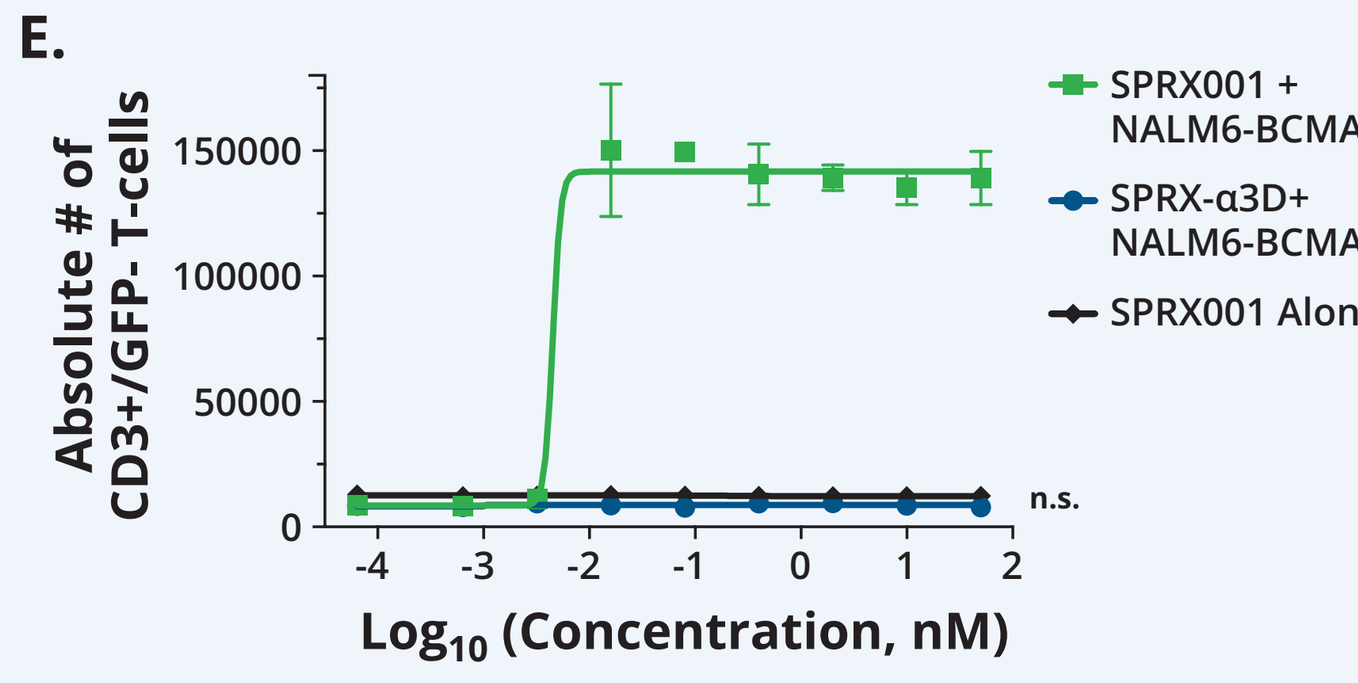

E.
Absolute # of CD3+/GFP- T-cells
SPRX001 + NALM6-BCMA
SPRX-α3D+ NALM6-BCMA
SPRX001 Alone
n.s.
Log10 (Concentration, nM)

D. The trimolecular complex of BCMA-SPRX001-ARC induces a cytotoxic response from ARC-T cells. ARC-T cells were cultured overnight with a dose titration of SPRX001 (or the negative control SPRX-α3D) and H929-WT, H929-BCMA$^{-/-}$, or NALM6-BCMA (all GFP and luciferase expressing). After culture, cytotoxicity was assessed based on remaining luciferase activity in cell pellet.

E. The trimolecular complex of BCMA-SPRX001-ARC drives ARC-T cells proliferation in an SPRX001 dose dependent manner. ARC-T cells were rested overnight in IL-2 free media, then stained with violet proliferation dye to assess cell division. Dye-stained ARC-T cells were cultured for 96-hours with a dose titration of SPRX001 (or the negative control SPRX-α3D) and NALM6-BCMA or SPRX001 and T cells alone. After 96-hours, cells were stained for CD3 and total T cells (CD3+GFP-) were enumerated from individual wells of a 96-well plate using a high throughput sampler.

ACLX001 eliminates MM.1S tumor cells in vivo comparable to a BCMA-targeted CAR-T

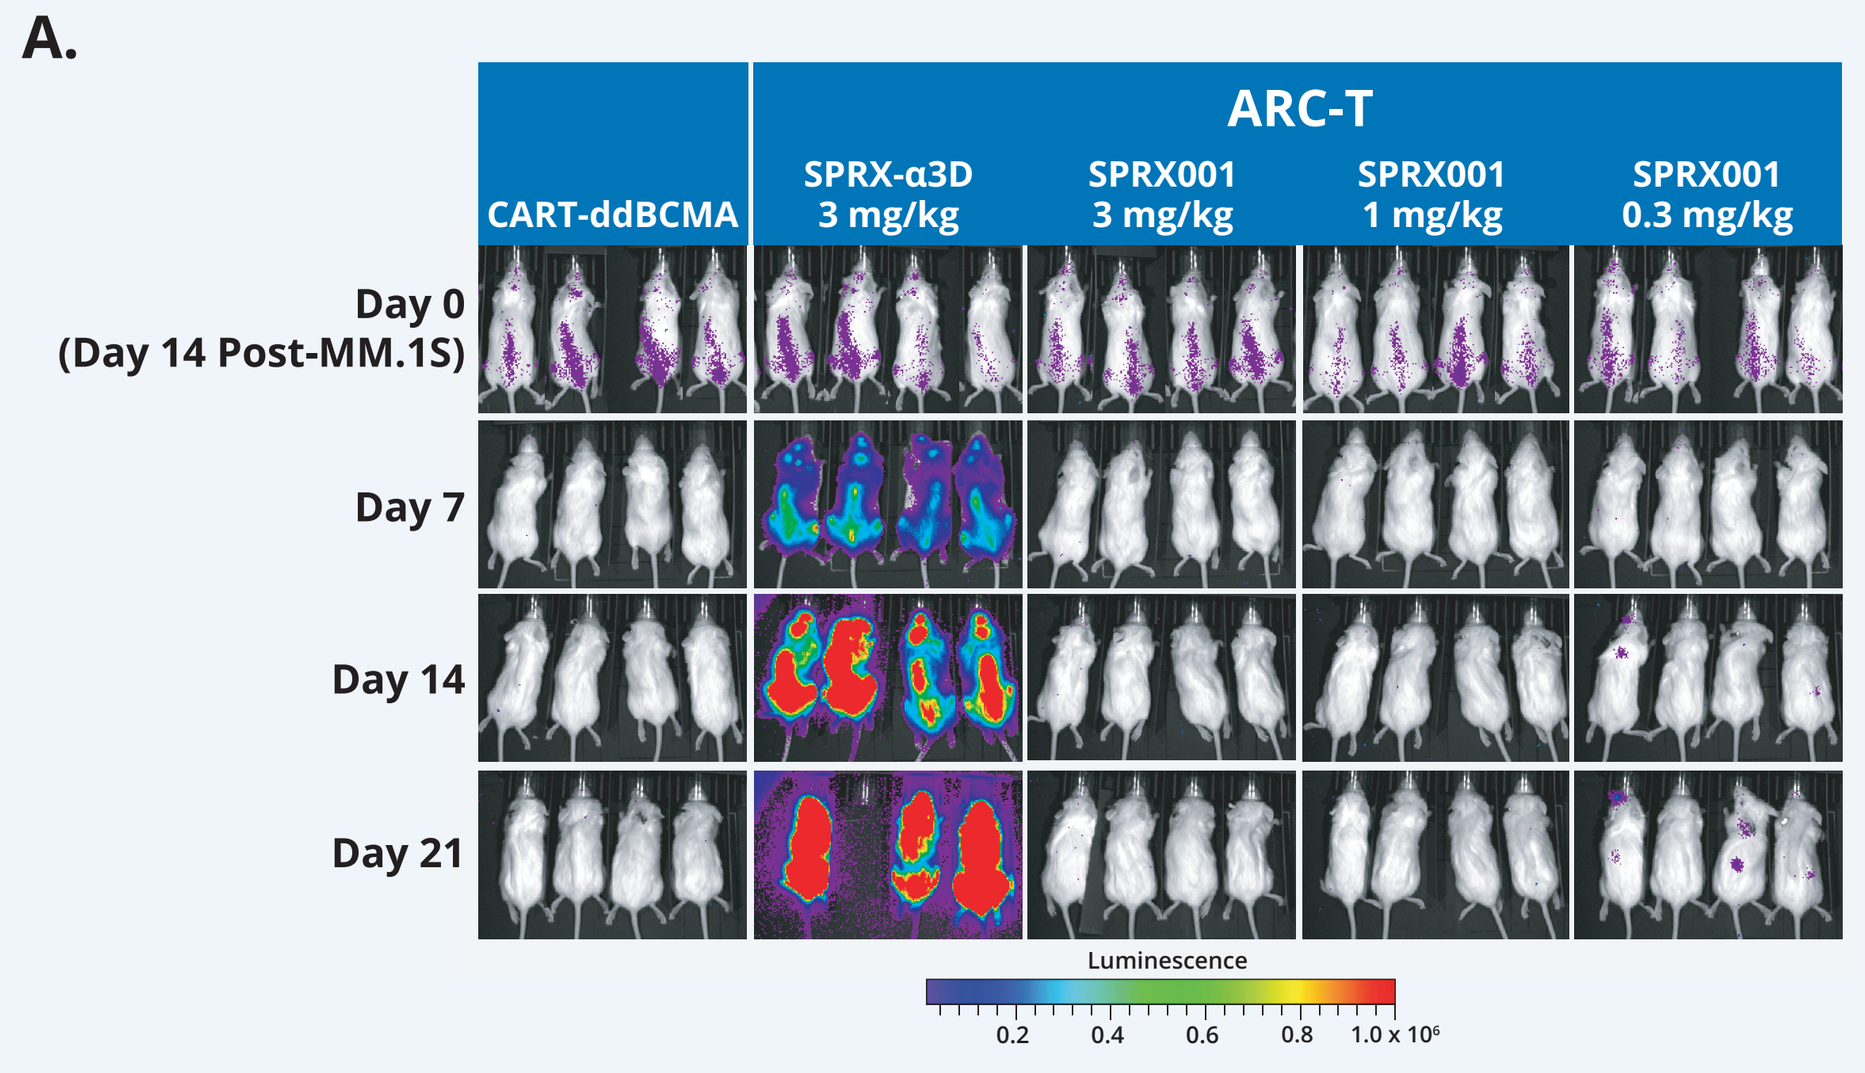

A.
ARC-T
CART-ddBCMA
SPRX-α3D 3 mg/kg
SPRX001 3 mg/kg
SPRX001 1 mg/kg
SPRX001 0.3 mg/kg
Day 0 (Day 14 Post-MM.1S)
Day 7
Day 14
Day 21
Luminescence

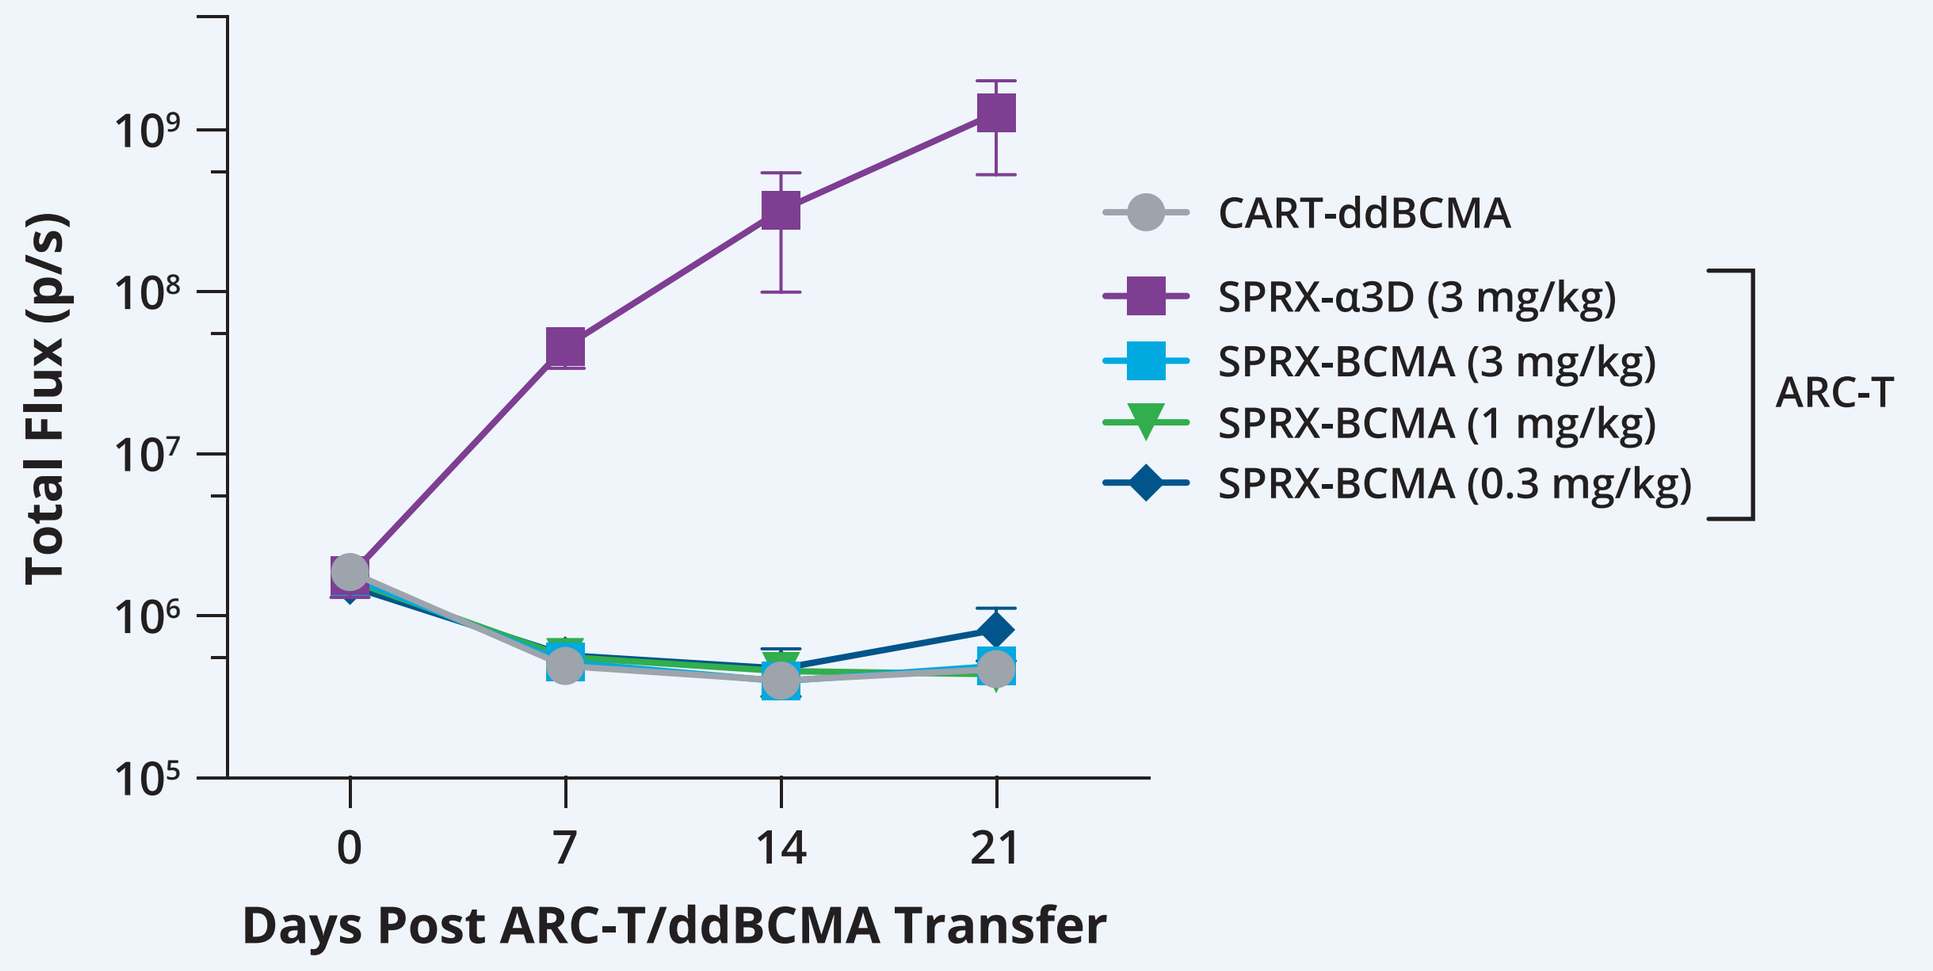

B.
Total Flux (p/s)
CART-ddBCMA
SPRX-α3D (3 mg/kg)
SPRX-BCMA (3 mg/kg)
SPRX-BCMA (1 mg/kg)
SPRX-BCMA (0.3 mg/kg)
ARC-T
Days Post ARC-T/ddBCMA Transfer

A. Both CART-ddBCMA and ACLX001 (ARC-T + SPRX001) cause regression of MM.1S in vivo. NSG mice (NOD-scid IL2Rg^{null}) were engrafted with 2×10^{6} MM.1S cells expressing GFP and Luciferase. 14 days later (Day 0), mice received 5×10^{6} ddBCMA-CAR cells (group 1) or 5×10^{6} ARC-T cells (groups 2-5). Mice that received ARC-T cells were dosed daily with sparX proteins IP at indicated dose levels for 21 days. Animals were subsequently imaged by IVIS throughout.

B. Total tumor burden was quantitated from the images in **(A)**.

ACLX001 exhibits comparable activity in vivo with intermittent dosing of SPRX001 in a disseminated model of BCMA+ B-cell leukemia (NALM6-BCMA)

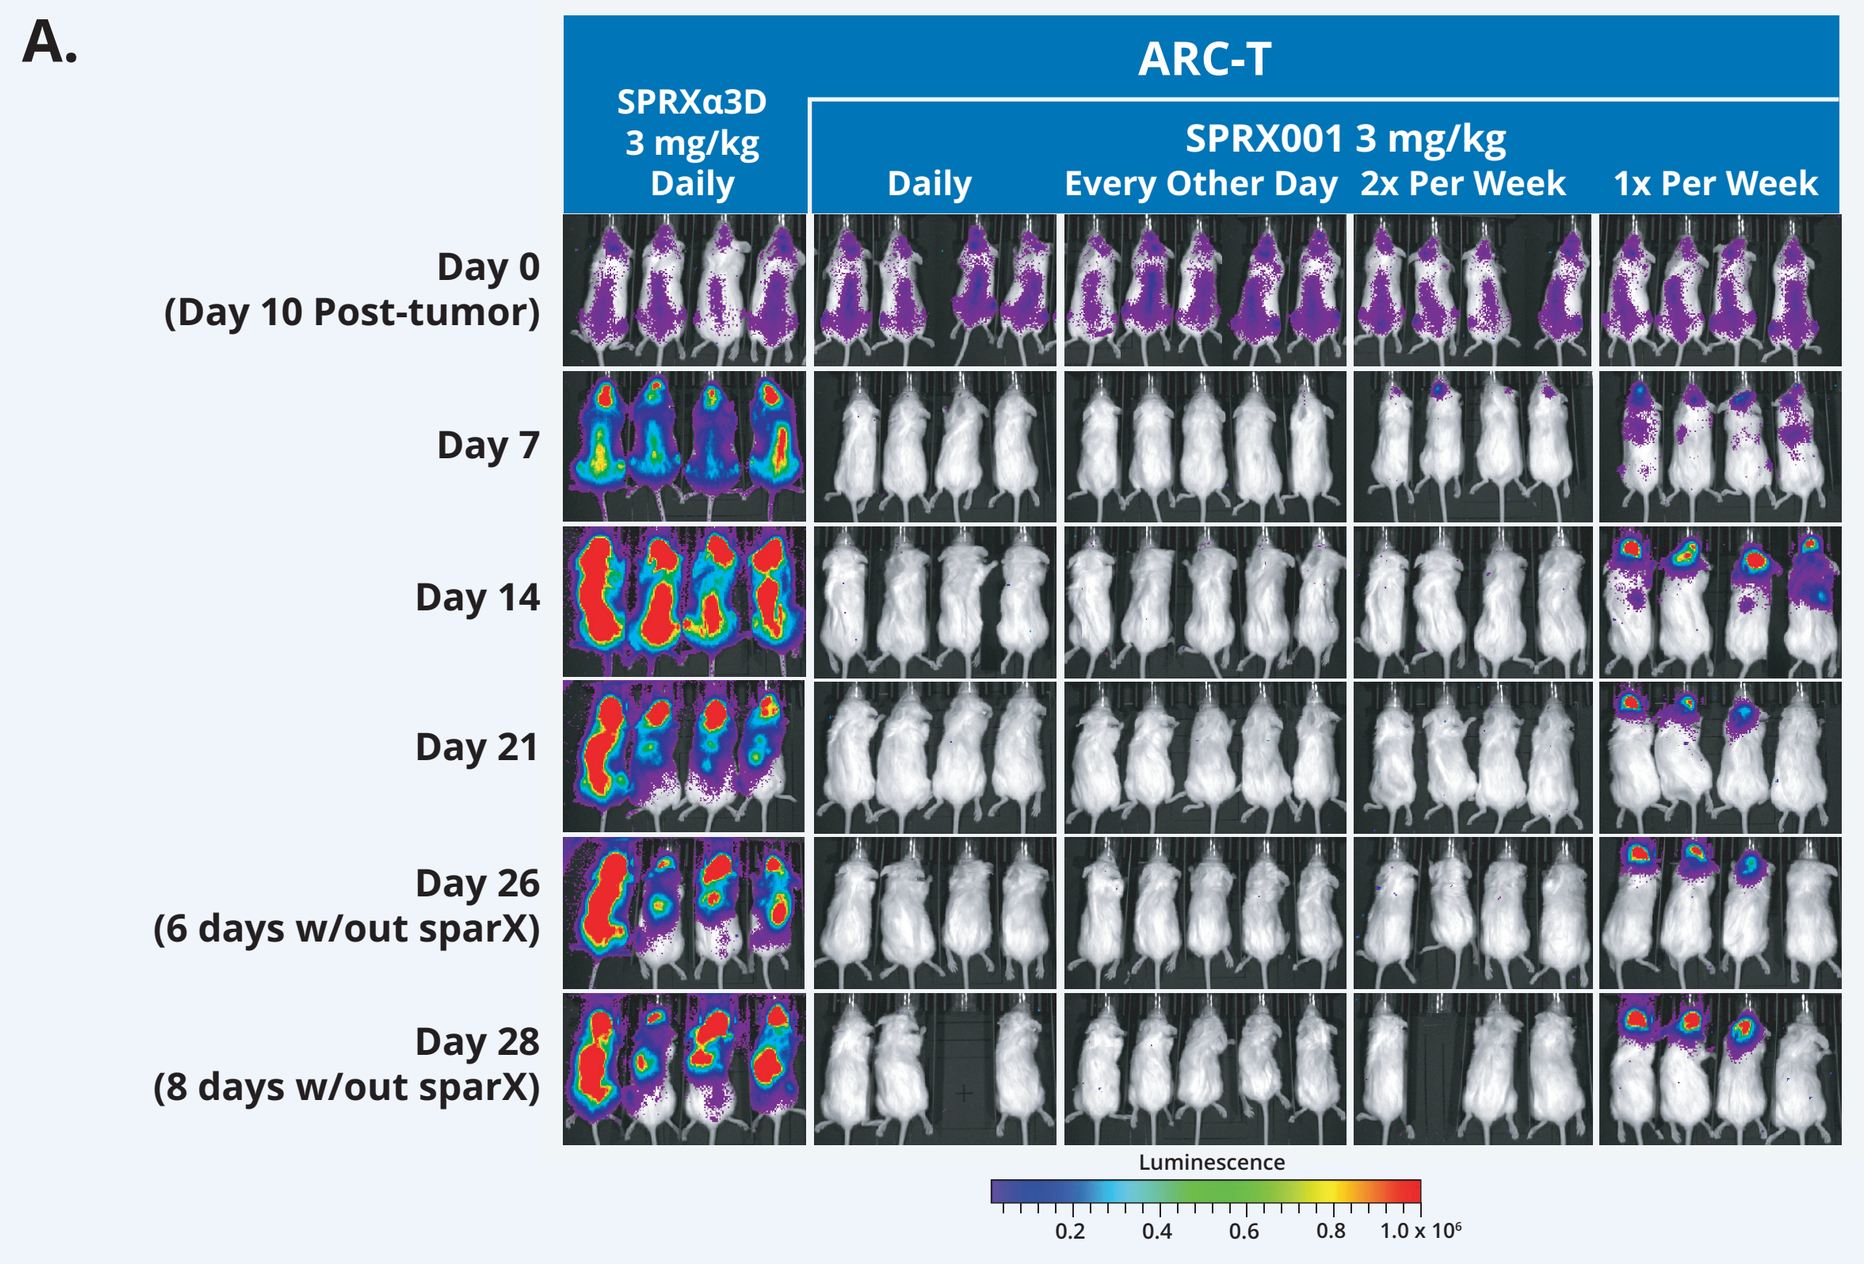

A.
ARC-T
SPRXα3D 3 mg/kg Daily
SPRX001 3 mg/kg
Daily
Every Other Day
2x Per Week
1x Per Week
Day 0 (Day 10 Post-tumor)
Day 7
Day 14
Day 21
Day 26 (6 days w/out sparX)
Day 28 (8 days w/out sparX)
Luminescence

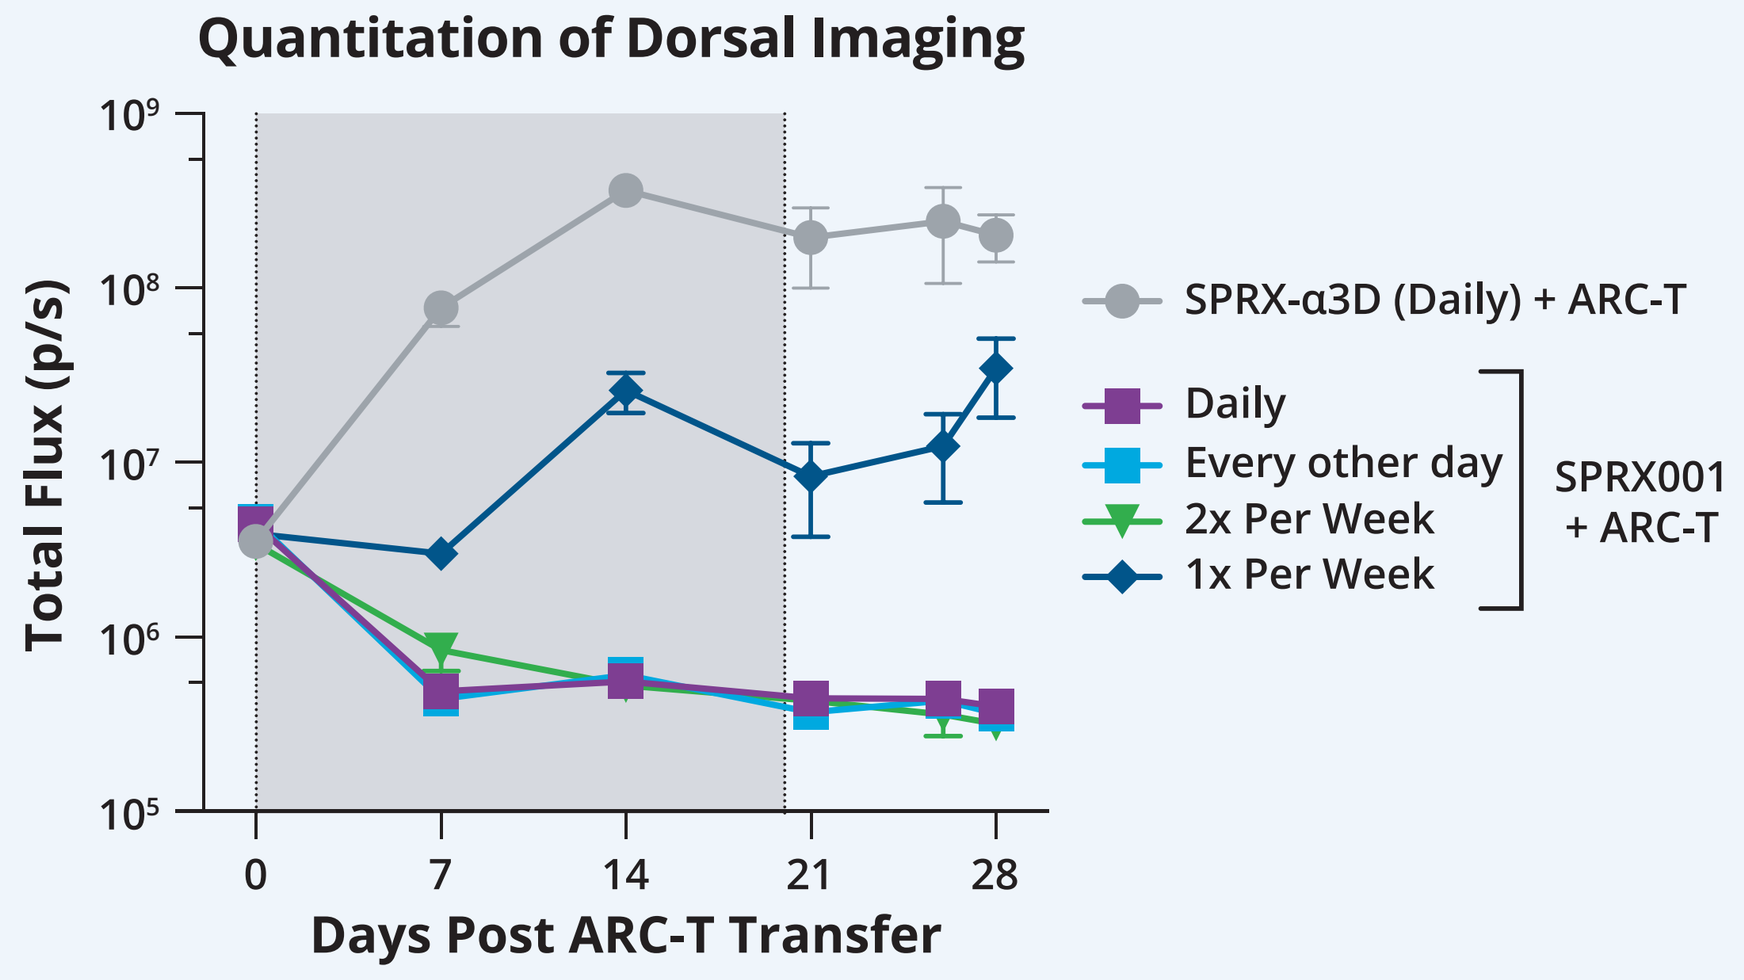

B.
Quantitation of Dorsal Imaging
Total Flux (p/s)
SPRX-α3D (Daily) + ARC-T
Daily
Every other day
2x Per Week
1x Per Week
SPRX001 + ARC-T
Days Post ARC-T Transfer

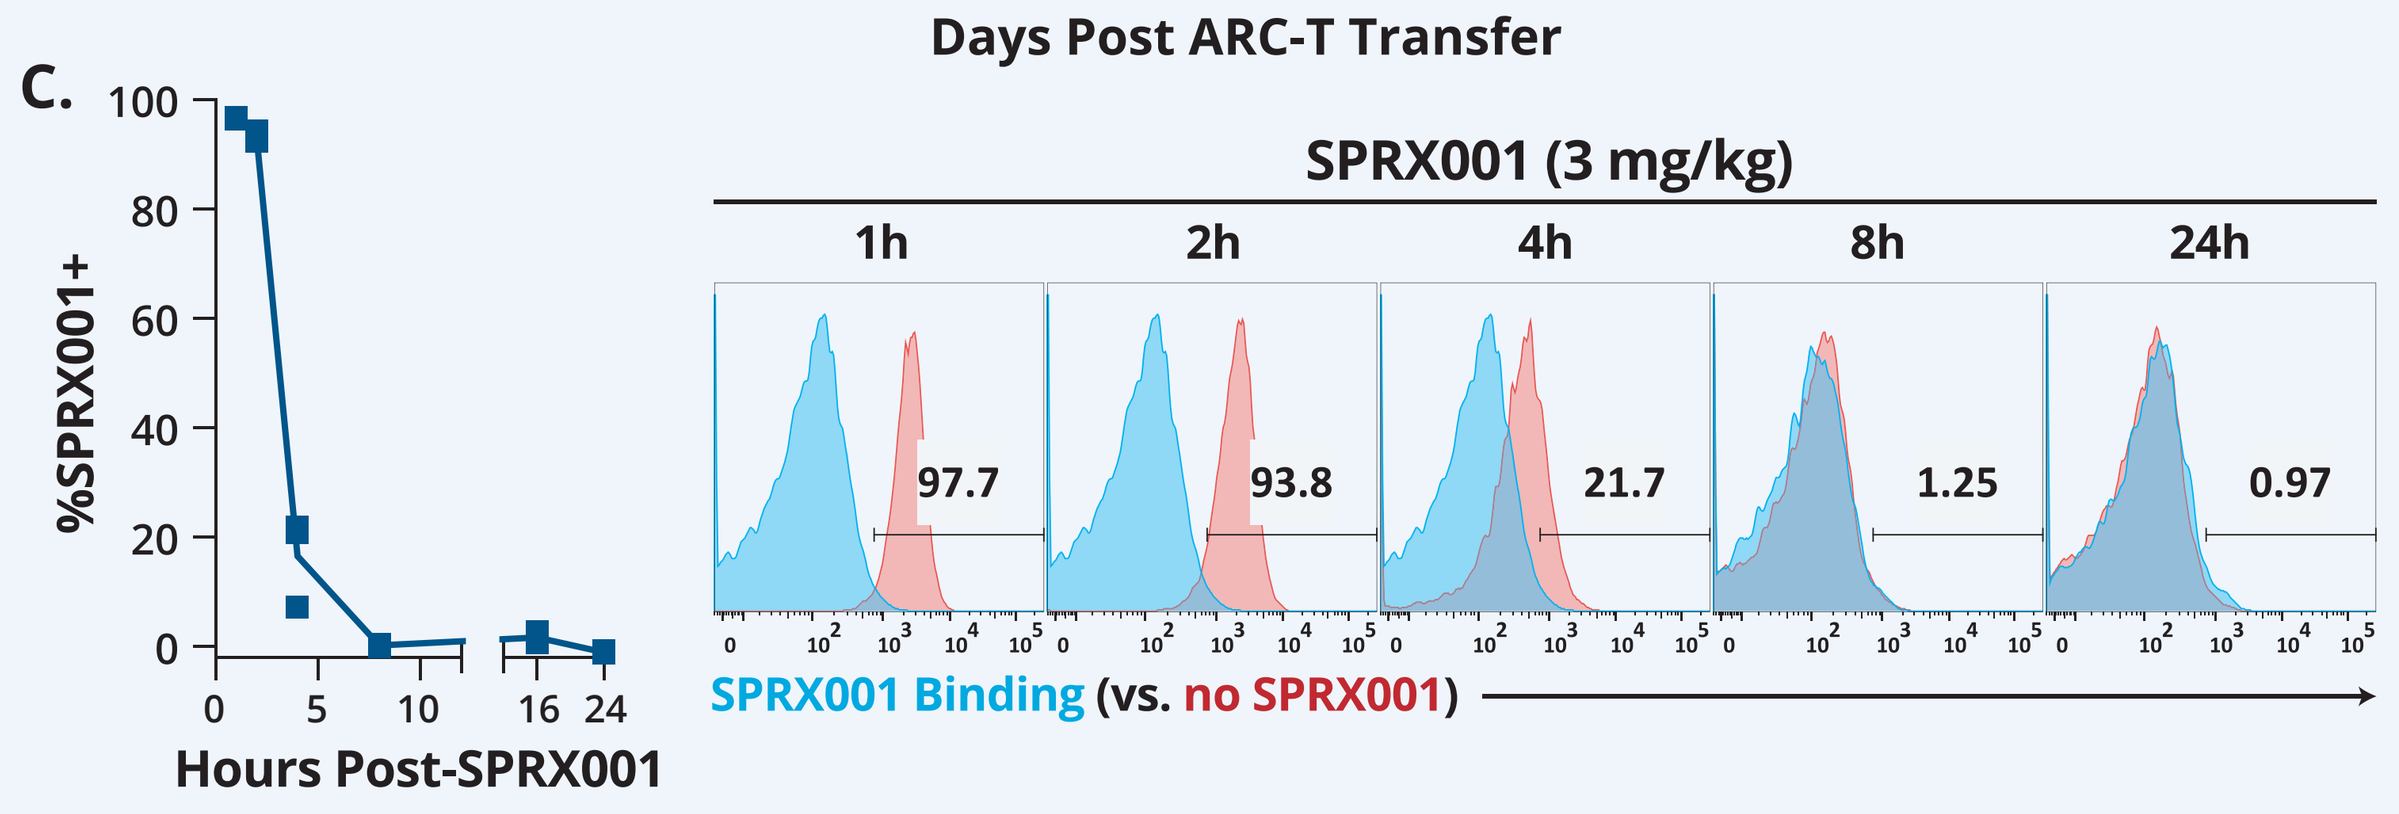

C.
%SPRX001+
Hours Post-SPRX001
SPRX001 (3 mg/kg)
1h
2h
4h
8h
24h
97.7
93.8
21.7
1.25
0.97
SPRX001 Binding (vs. no SPRX001)

A. ACLX001 causes regression of NALM6-BCMA tumors in vivo with intermittent dosing of SPRX001. NSG mice were engrafted with 1×10^{6} NALM6-BCMA cells expressing GFP and Luciferase. 10 days later (Day 0), mice were engrafted with 5×10^{6} ARC-T cells and were dosed with the negative control SPRX-α3D daily or with the SPRX001 daily, every other day, twice per week, or weekly for 21 days. Animals were imaged on IVIS throughout.

B. Total tumor burden was quantitated from the images in **(A)**. Grey shading indicates the period of sparX-protein administration.

C. The in vivo residence time of SPRX001 on NALM6-BCMA tumor cells was assessed in mice following I.V. SPRX001 administration. Femurs were collected at various timepoints post-injection, flushed, samples fixed, and SPRX001-binding was detected on tumor cells via an anti-tag antibody. Binding is presented as percent positive **(left)**. Representative histograms for each time point are also presented **(right)**.

ARC-T cell kinetics and memory cell differentiation vary widely in vivo

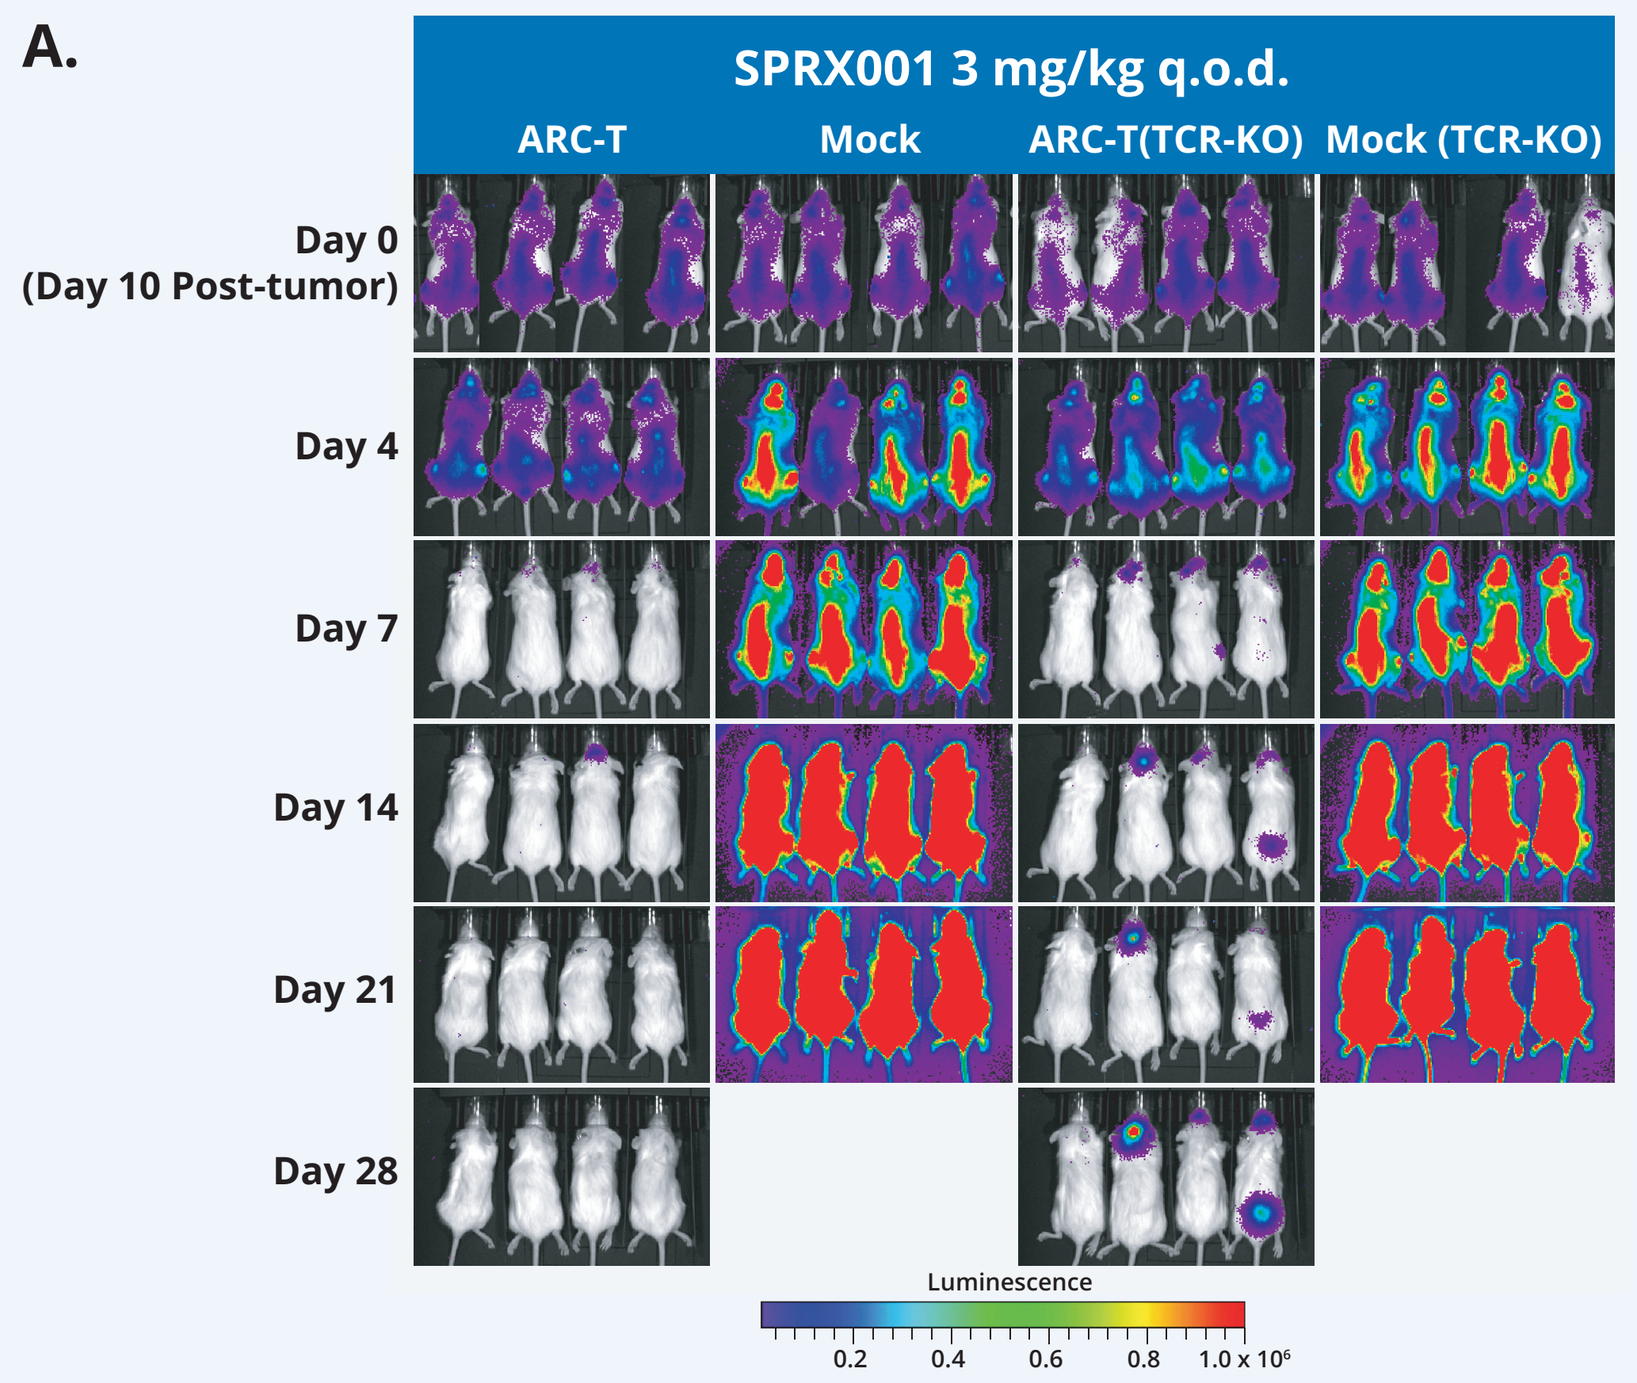

A.
SPRX001 3 mg/kg q.o.d.
ARC-T
Mock
ARC-T(TCR-KO)
Mock (TCR-KO)
Day 0 (Day 10 Post-tumor)
Day 4
Day 7
Day 14
Day 21
Day 28
Luminescence

A. ACLX001 (ARC-T + SPRX001) halts expansion and causes regression of NALM6-BCMA in vivo. NSG mice (NOD-scid IL2Rg^{null}) were engrafted with 1×10^{6} NALM6-BCMA cells expressing GFP and Luciferase. 10 days later (Day 0), animals were imaged on IVIS and randomized. Mice were engrafted with ARC-T cells (T-cell receptor wild type or CRISPR-KO) or Mock transduced T cells (T-cell receptor wild type or CRISPR-KO). All mice were dosed with SPRX001 every other day (q.o.d.) for 28 days. Animals were imaged by IVIS throughout.

ARC-T cell kinetics and memory cell differentiation vary widely in vivo

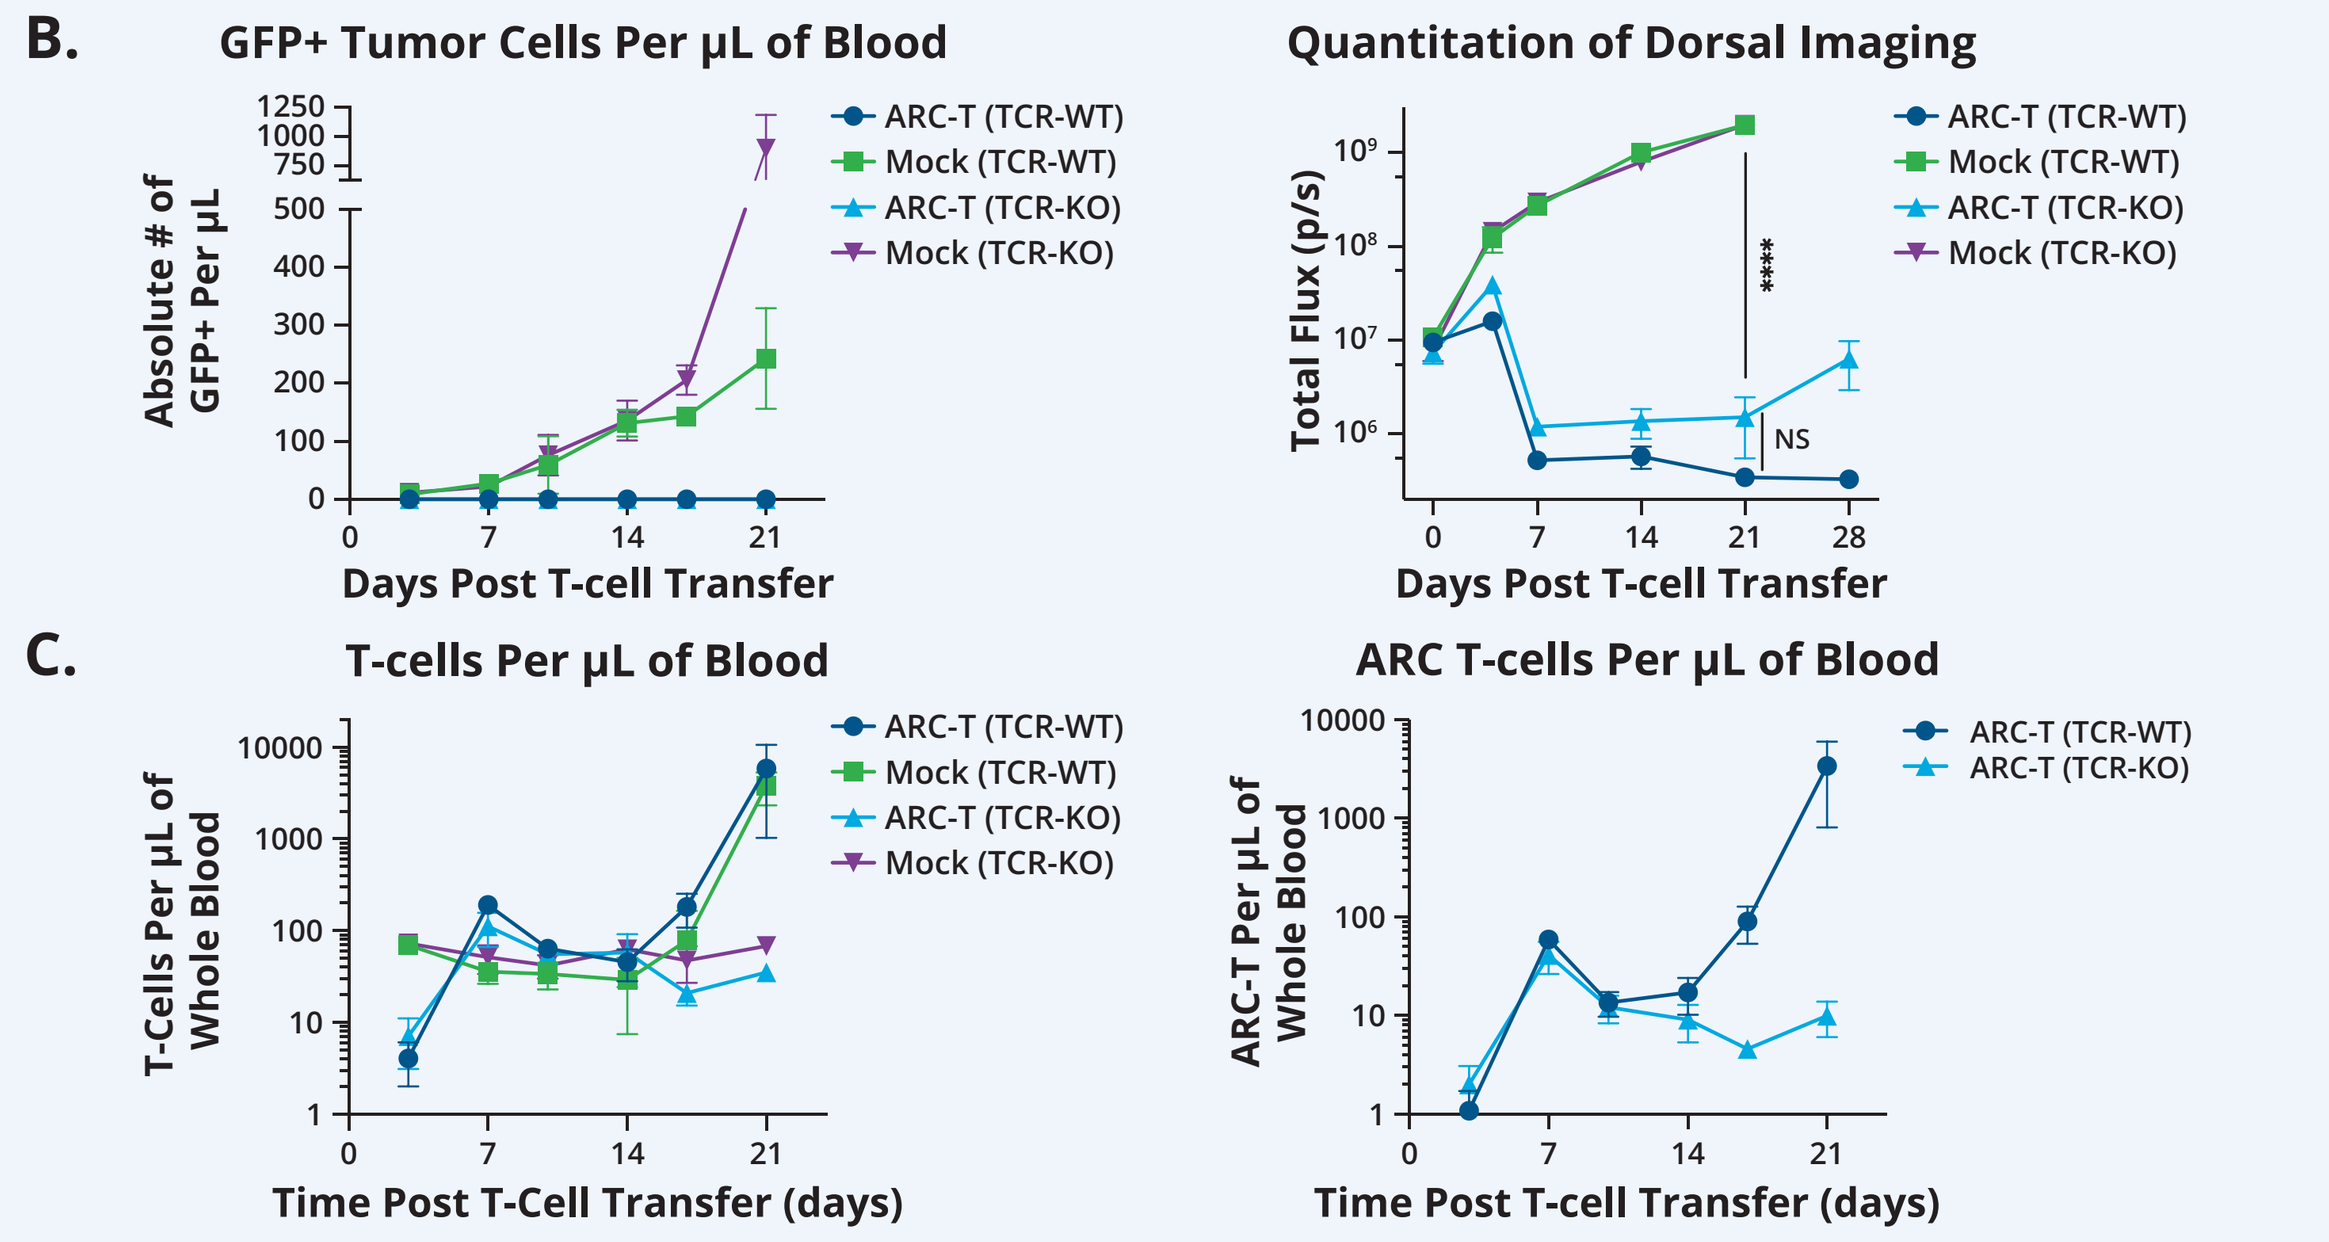

B.
GFP+ Tumor Cells Per μL of Blood
Absolute # of GFP+ Per μL
Days Post T-cell Transfer
Quantitation of Dorsal Imaging
Total Flux (p/s)
NS
ARC-T (TCR-WT)
Mock (TCR-WT)
ARC-T (TCR-KO)
Mock (TCR-KO)
C.
T-cells Per μL of Blood
T-Cells Per μL of Whole Blood
Time Post T-Cell Transfer (days)
ARC T-cells Per μL of Blood
ARC-T Per μL of Whole Blood
Time Post T-cell Transfer (days)

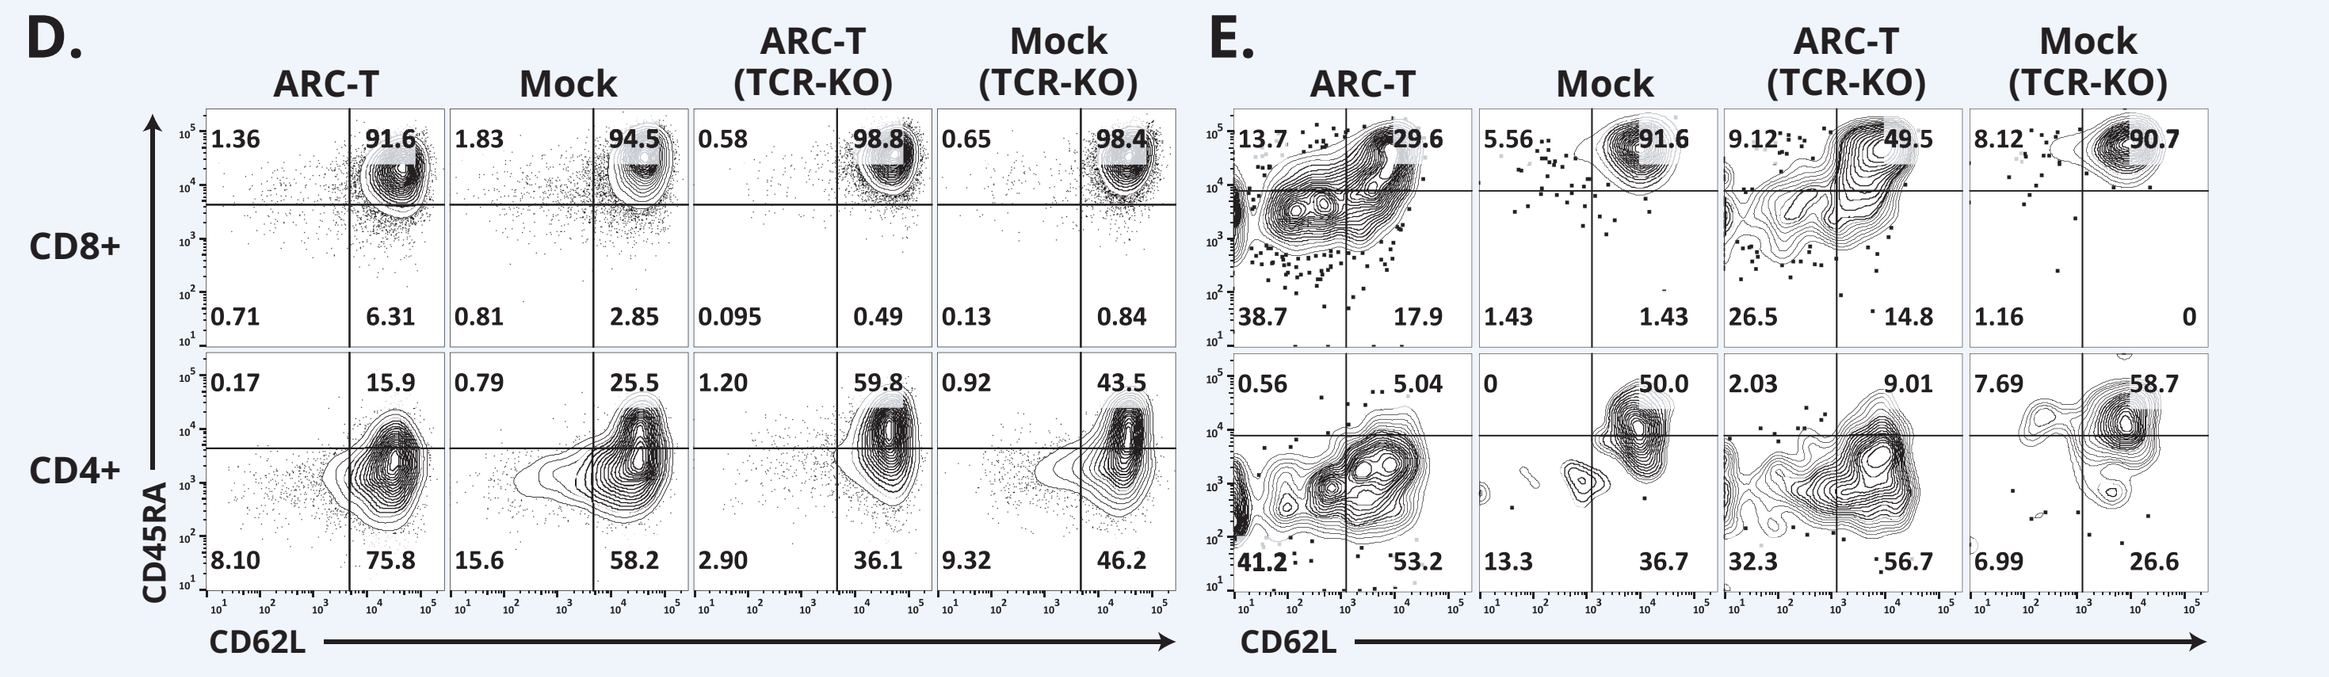

D.
ARC-T
Mock
ARC-T (TCR-KO)
Mock (TCR-KO)
CD8+
CD4+
CD45RA
CD62L
E.
ARC-T
Mock
ARC-T (TCR-KO)
Mock (TCR-KO)
CD62L

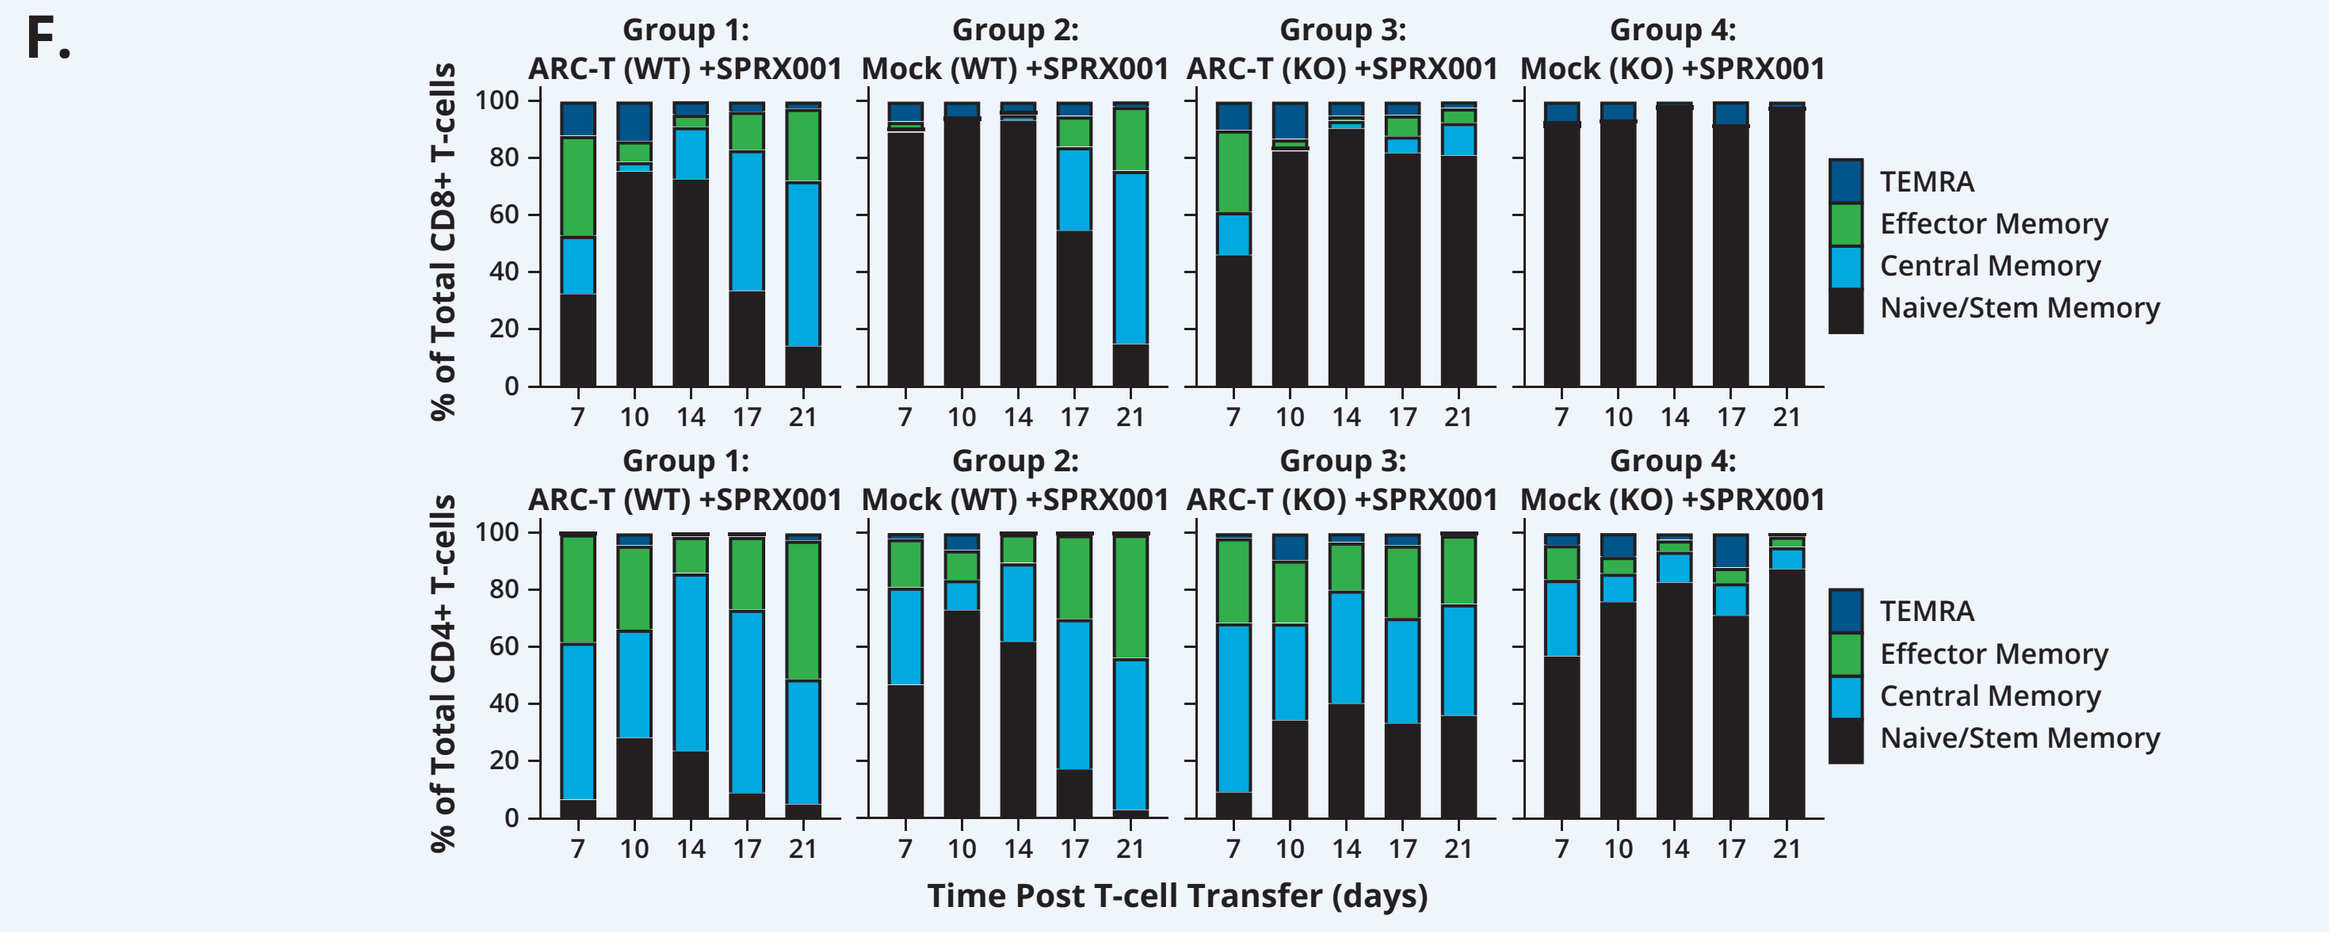

F.
Group 1: ARC-T (WT) +SPRX001
Group 2: Mock (WT) +SPRX001
Group 3: ARC-T (KO) +SPRX001
Group 4: Mock (KO) +SPRX001
% of Total CD8+ T-cells
% of Total CD4+ T-cells
TEMRA
Effector Memory
Central Memory
Naive/Stem Memory
Time Post T-cell Transfer (days)

B. (Left) Absolute number of circulating GFP+ tumor cells per μl of whole blood were monitored throughout the course of the experiment. **(Right)** Total tumor burden was quantitated from the dorsal images in **(A)**.

C. Absolute number of circulating total T cells **(left)** or ARC+ T cells **(right)** per μL of whole blood were monitored throughout the course of the experiment. After day 14 post-T-cell transfer, T-cell expansion is driven primarily through GvHD in the TCR-WT groups in this experiment.

D. T cells were assessed for their expression of CD45RA and CD62L to determine their phenotype prior to transfer. CD8 T cells were primarily of the naïve/stem memory phenotype, while CD4 T cells were a mixture of naïve/stem memory and central memory.

E. SPRX001 drives effector and memory phenotype differentiation of ARC-T cells as they are activated and expand in vivo. T cells were monitored for their differentiation into effector and memory phenotype cells seven days post-transfer based on their expression of CD45RA and CD62L.

F. Effector and memory phenotype differentiation of ARC-T or Mock transduced (TCR-WT or TCR-KO) were tracked throughout the course of experiment. As in **(D)**, T cells were monitored for their differentiation into effector and memory phenotype based on their expression of CD45RA and CD62L throughout the course of the experiment. CD8+ T cells are presented on the top and CD4+ T cells are presented on the bottom. After day 14 post-T-cell transfer, T-cell differentiation is driven primarily through GvHD in the TCR-WT groups as compared to the TCR-KO groups.

Conclusions

- ACLX001 is a novel cell therapy for BCMA+ malignancies
 - The SPRX001 protein imparts BCMA-binding and -targeting capacity to ARC-T Cells
 - Only when SPRX001 bridges ARC-T cells to BCMA on target cells are ARC-T cells activated to produce cytokines, have cytotoxic activity against BCMA+ targets, or proliferate in response to BCMA+ target cells.
- ACLX001 eliminates MM.1S tumor cells in vivo comparable to a BCMA-targeted CAR-T
- SPRX001 dosed daily, every other day, or twice per week eliminated measurable NALM6-BCMA tumor burden
 - Despite rapid reduction in receptor occupancy on the BCMA+ tumor cells, intermittent dosing still achieves tumor clearance
- ARC-T cell kinetics and memory cell differentiation vary widely in vivo when driven by different stimuli

*The data in this poster were generated with related but distinct reagents from the ACLX001 clinical reagents and are meant to support the clinical development of this product.

Presented at the American Association for Cancer Research 2021 Virtual Annual Meeting, April 10–15, 2021

©2021 Arcellx, Inc. All rights reserved.