Chimeric Antigen Receptors Incorporating Novel (non-scFv) Binding Domains Targeting CD123 Direct Potent Antitumor Activity of T Cells: Correlation Between Affinity and Activity

ARCELLX

Abstract 3243

David W. LaFleur, Justin P. Edwards, Liubov Zaritskaya, Ankit Gupta, Haiying Qin, Terry J. Fry, Jeffery S. Swers, C. Jenny Mu, Laura K. Richman, David M. Hilbert

Arcellx, Inc., Gaithersburg, MD

DLaFleur@arcellx.com

Introduction

D Domains Constitute a Robust Scaffold for Rapid Development of Targeting Domains

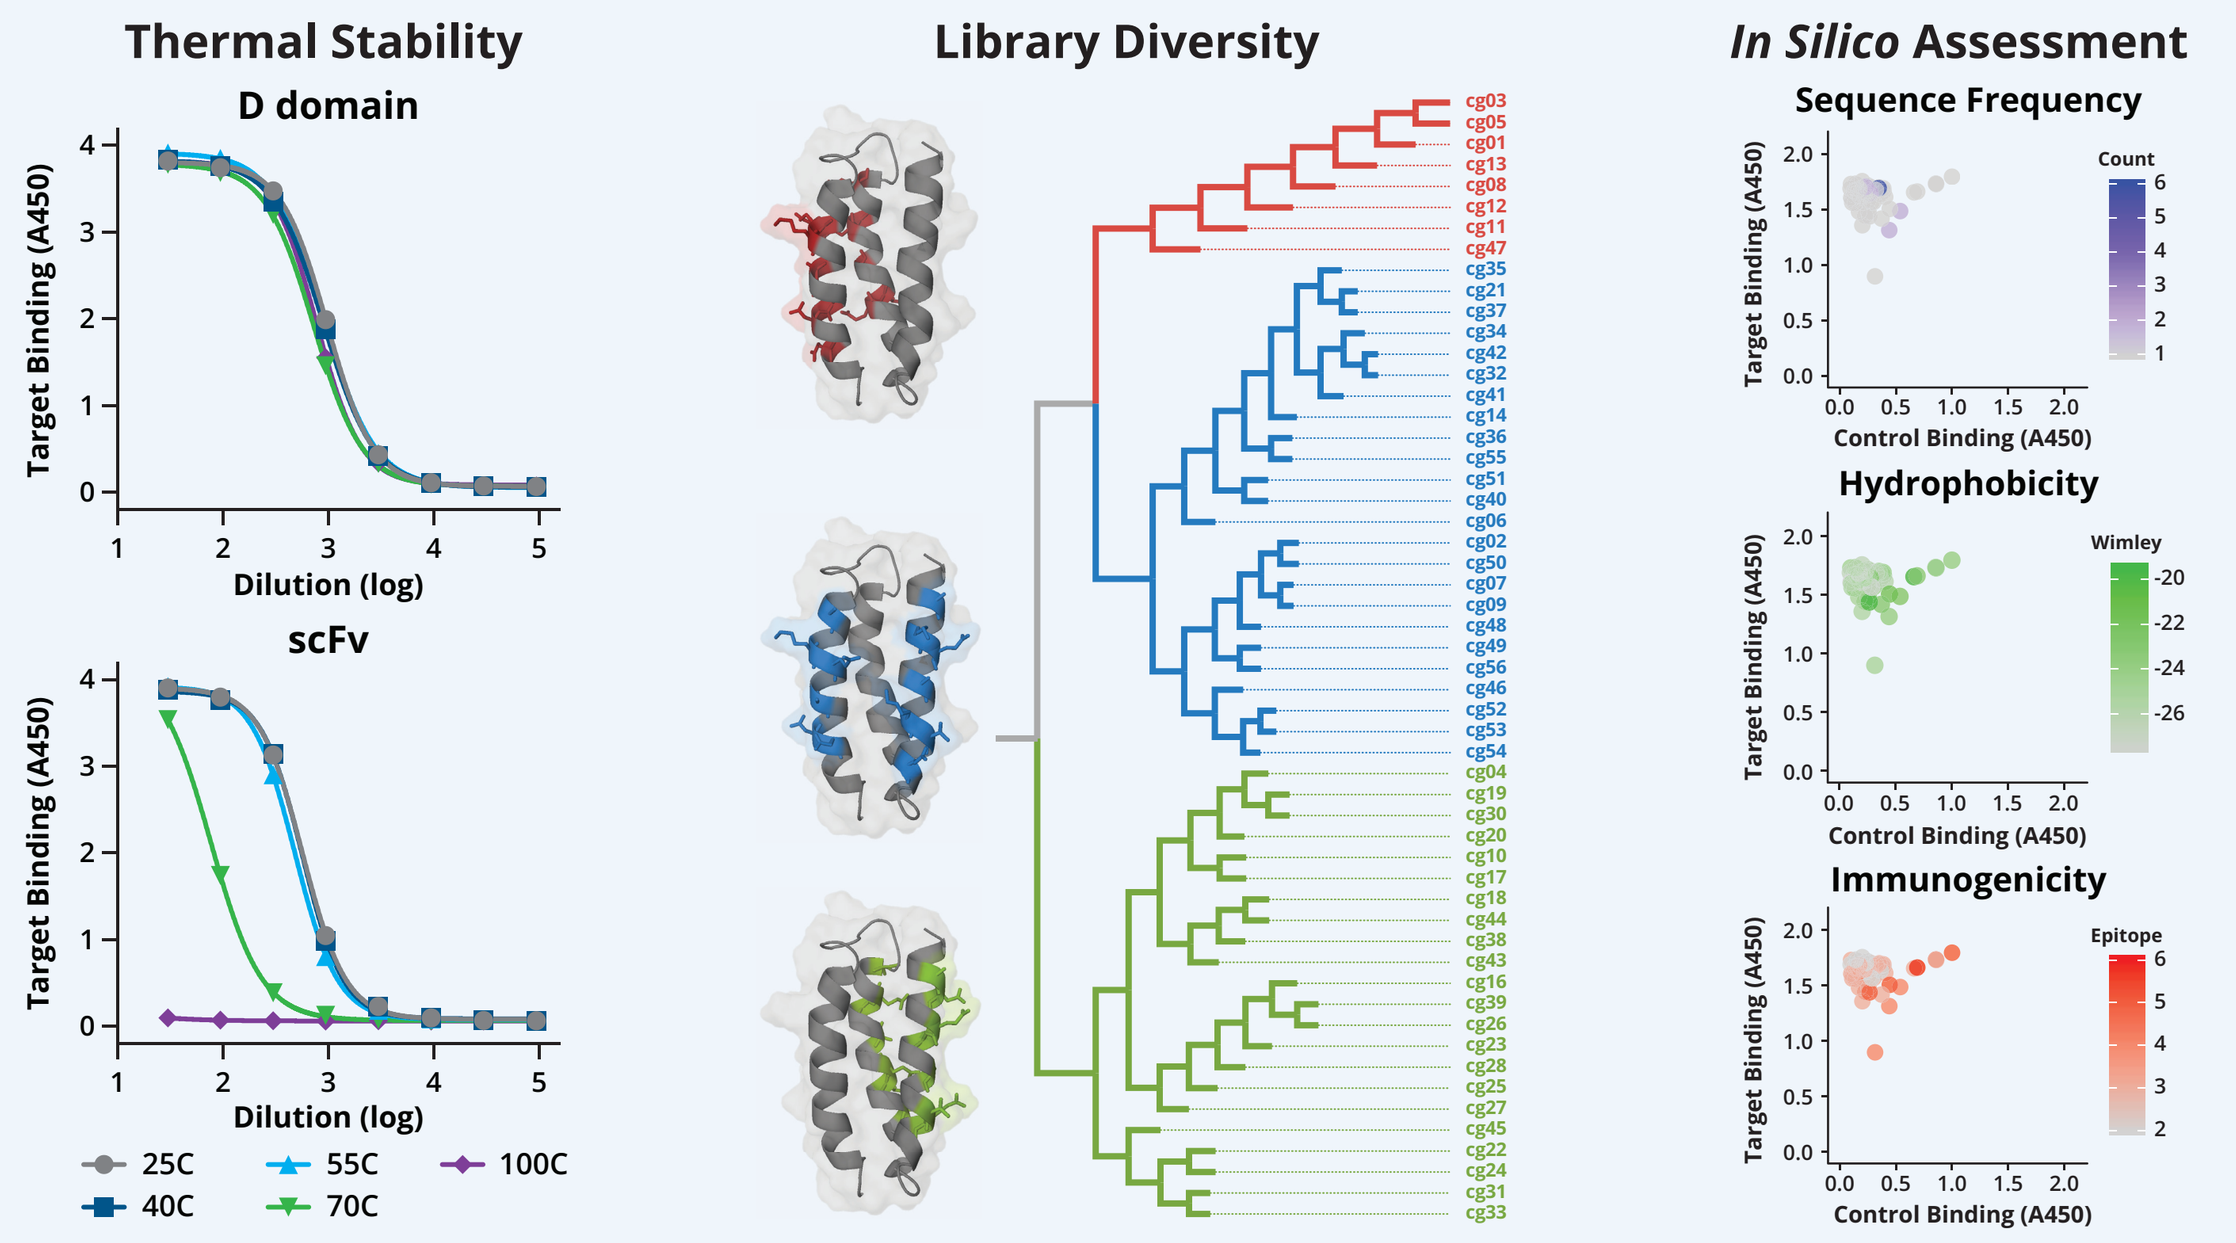

Thermal Stability
D domain
scFv
Target Binding (A450)
Dilution (log)
25C
40C
55C
70C
100C
Library Diversity
In Silico Assessment
Sequence Frequency
Hydrophobicity
Immunogenicity
Control Binding (A450)
Count
Wimley
Epitope

- Conventional chimeric antigen receptors utilize an antibody-derived scFv as the targeting domain
- D domains are 73 aa in length (approximately 1/3 the size of a scFv) and contain no disulfide bonds or native glycosylation[1]
- Rapid folding of the scaffold[2] imparts exceptional thermal stability **(left)** while simultaneously accommodating a high number of residue substitutions
- Multiple library designs of the triple alpha helical structure, translate into a high degree of paratope and sequence diversity **(center)**
- Combined *in vitro* and *in silico* screens of phage library clones afford predictive assessments of D domain target specificity, sequence diversity, expression and immunogenicity **(right)**
- D domains are selected for low potential immunogenicity and are ultimately engineered to produce an Abzena EpiScreen™ score of zero[3]

The ARC-sparX Platform

Conventional CAR-T cell therapies often target tumors through a mono-specific receptor that is constitutively expressed. To improve the flexibility and control of T-cell response, we developed the ARC-sparX platform, which separates the antigen recognition and killing functions of the conventional CAR-T cell.

The ARC-sparX platform is comprised of two key components:

1. sparX (**s**oluble **p**rotein **a**ntigen-**r**eceptor **X**-linker) protein: binds specific antigens on diseased cells and flags those cells for destruction.
2. ARC-T Cells (**A**ntigen **R**eceptor **C**omplex **T** Cells): bind sparX proteins and kill flagged cells.

Tri-complex (ARC-T + sparX + target cell) must be formed to activate cytolytic killing.

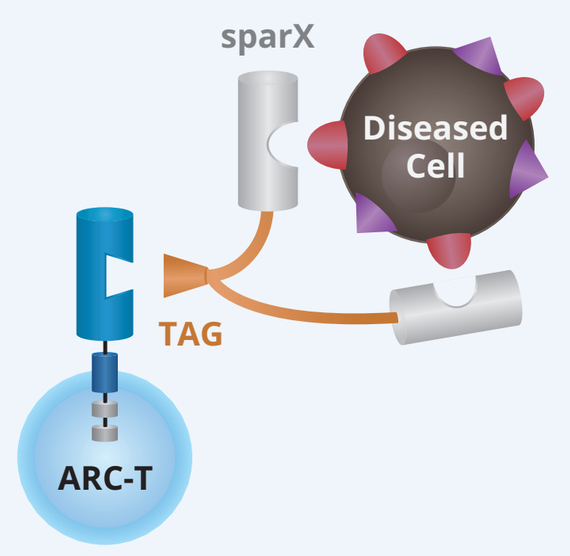

sparX
Diseased Cell
TAG
ARC-T

sparX protein

- No inherent activity
- Mono-valent affinity comparable to that of scFv
- Engineered to minimize immunogenic potential
- **"TAG"** is a fragment of human alpha fetoprotein (AFP)

ARC-T Cell

- Conventional CAR architecture incorporating an anti-TAG D domain
- Receptor for **"TAG"** has affinity in low nanomolar range
- Only activated upon formation of tri-complex (ARC-T + sparX + antigen-expressing target cell)
- Same viral vector regardless of antigen target

sparX Can Be Mono-valent, Multi-valent, or Multi-specific

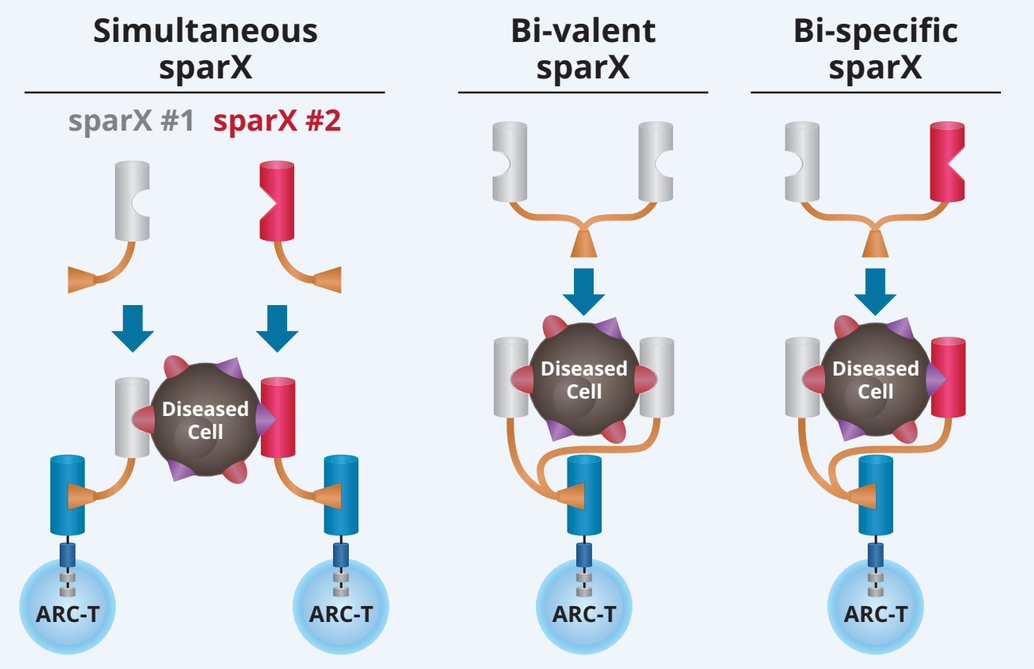

Simultaneous sparX
Bi-valent sparX
Bi-specific sparX
sparX #1
sparX #2
Diseased Cell
ARC-T

- Can be given in combination or in sequence
- Addresses low density or heterogeneous expression of antigen
- Bi-specific sparX proteins support "AND-gated" as well as "OR-gated" targeting of diseased cells

Results

CAR T Cells Incorporating D Domains Control AML Tumor Growth *In Vivo*

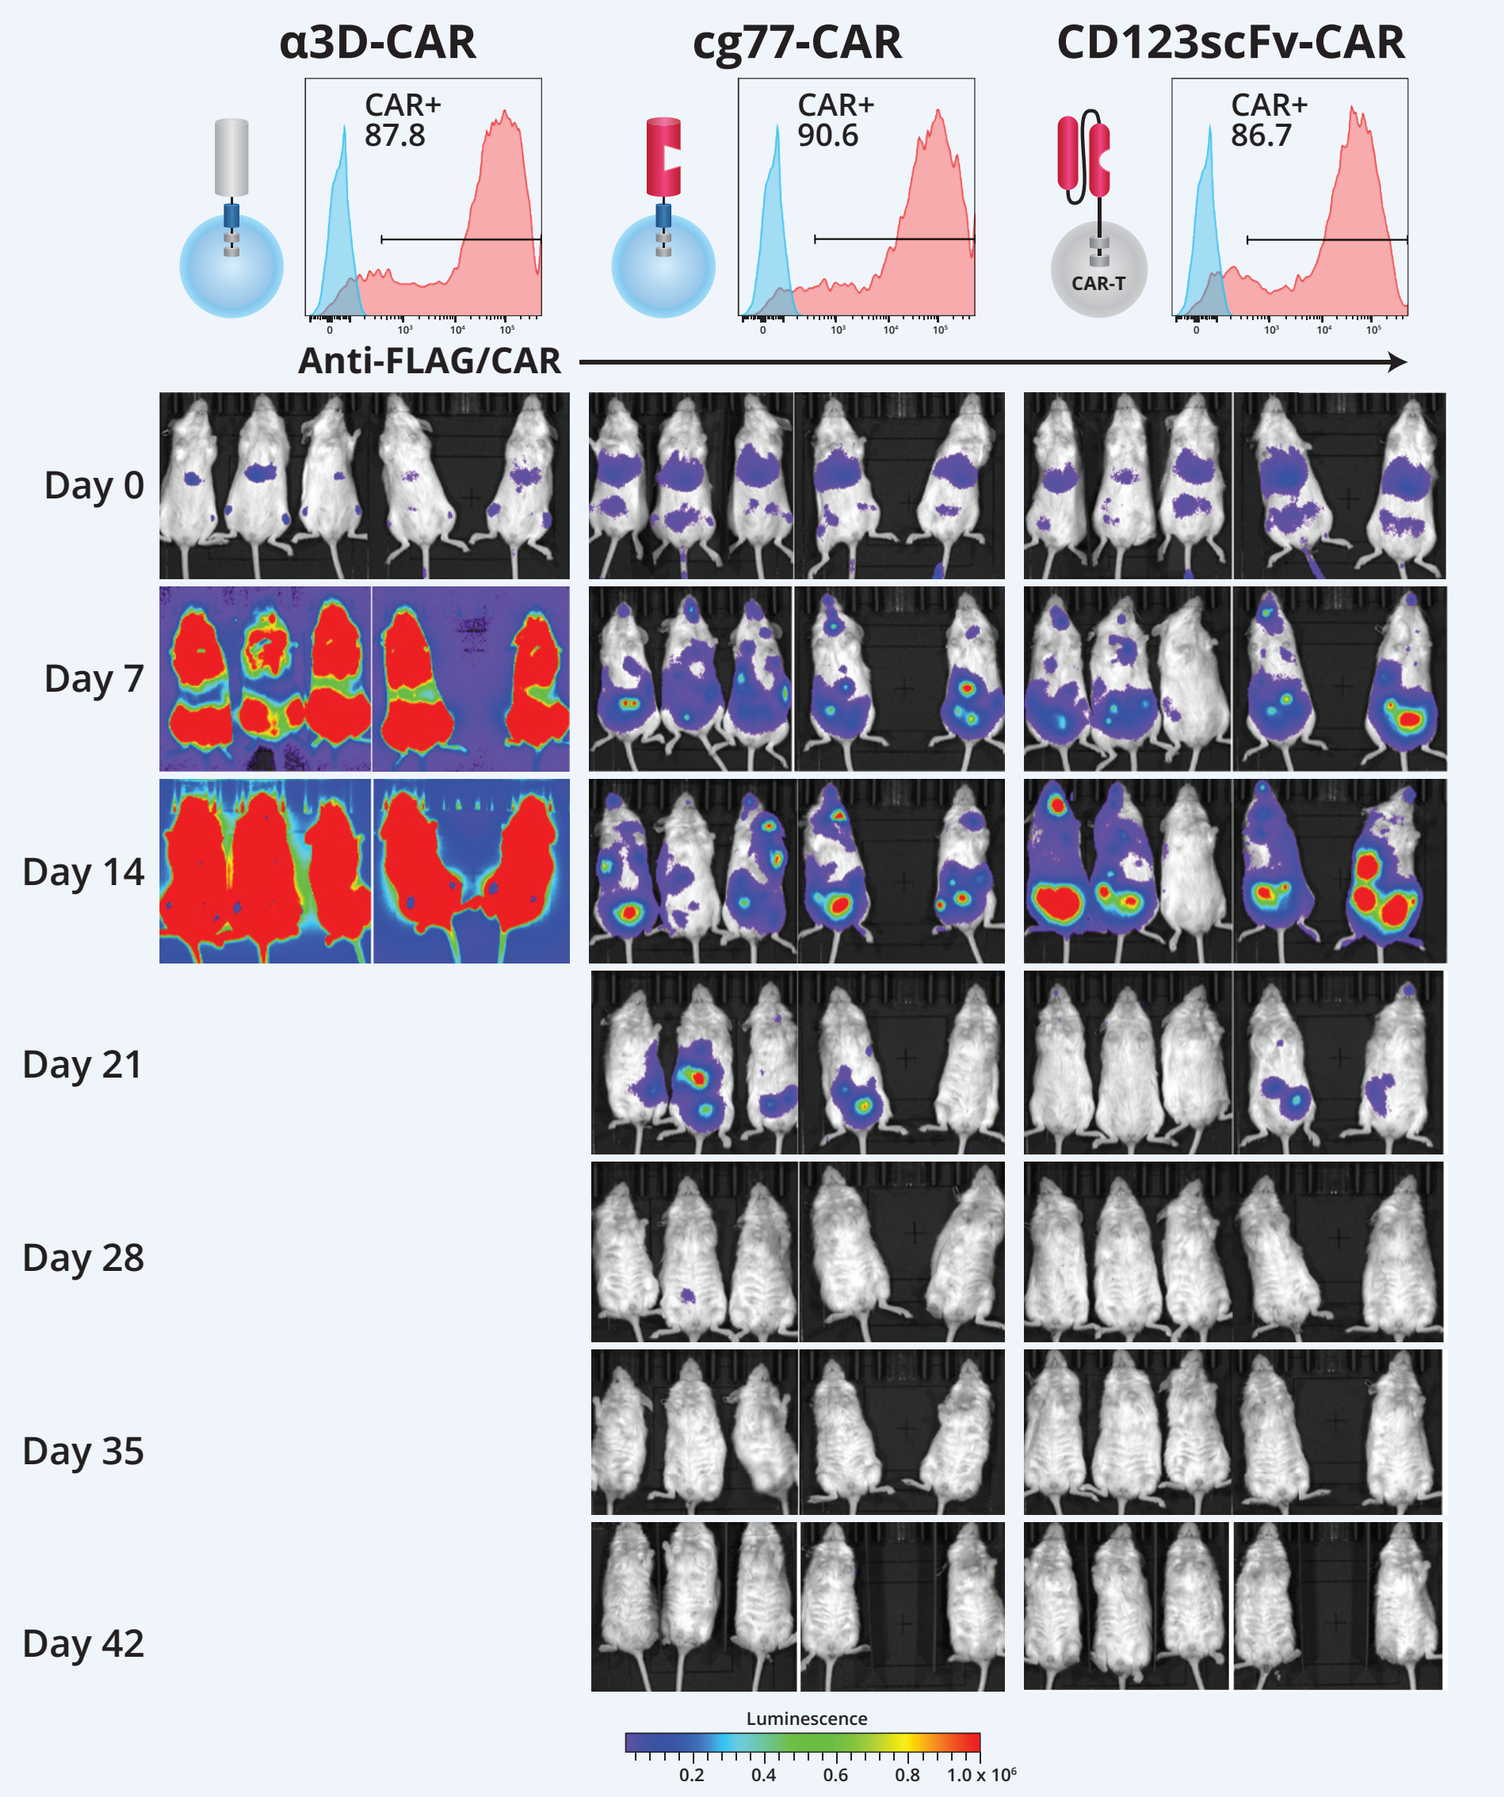

α3D-CAR
cg77-CAR
CD123scFv-CAR
CAR+ 87.8
CAR+ 90.6
CAR+ 86.7
Anti-FLAG/CAR
Day 0
Day 7
Day 14
Day 21
Day 28
Day 35
Day 42
Luminescence

- A CD123-binding D domain was identified through phage display methods and further deimmunized to remove predicted T cell epitopes (cg77)[1]
- Detection of N-terminal Flag epitope by flow cytometry demonstrated comparable percent positive CAR expression on T cells between non-binding D domain CAR (a3D-CAR), cg77-CAR and a 32716-scFv-based CAR targeting CD123 (CD123scFv-CAR)
- AML-derived, CD123-positive MOLM14 tumor cells ($3x10^6$) were administered systemically 1 day prior to administration of $3x10^6$ a3D-CAR, cg77-CAR and CD123scFv-CAR transduced primary human T cells
- Both cg77-CAR and CD123scFv-CAR eliminated detectible levels of tumor cells as measured by IVIS imaging

ARC-sparX Platform Incorporating D Domains Control AML Tumor Growth *In Vivo*

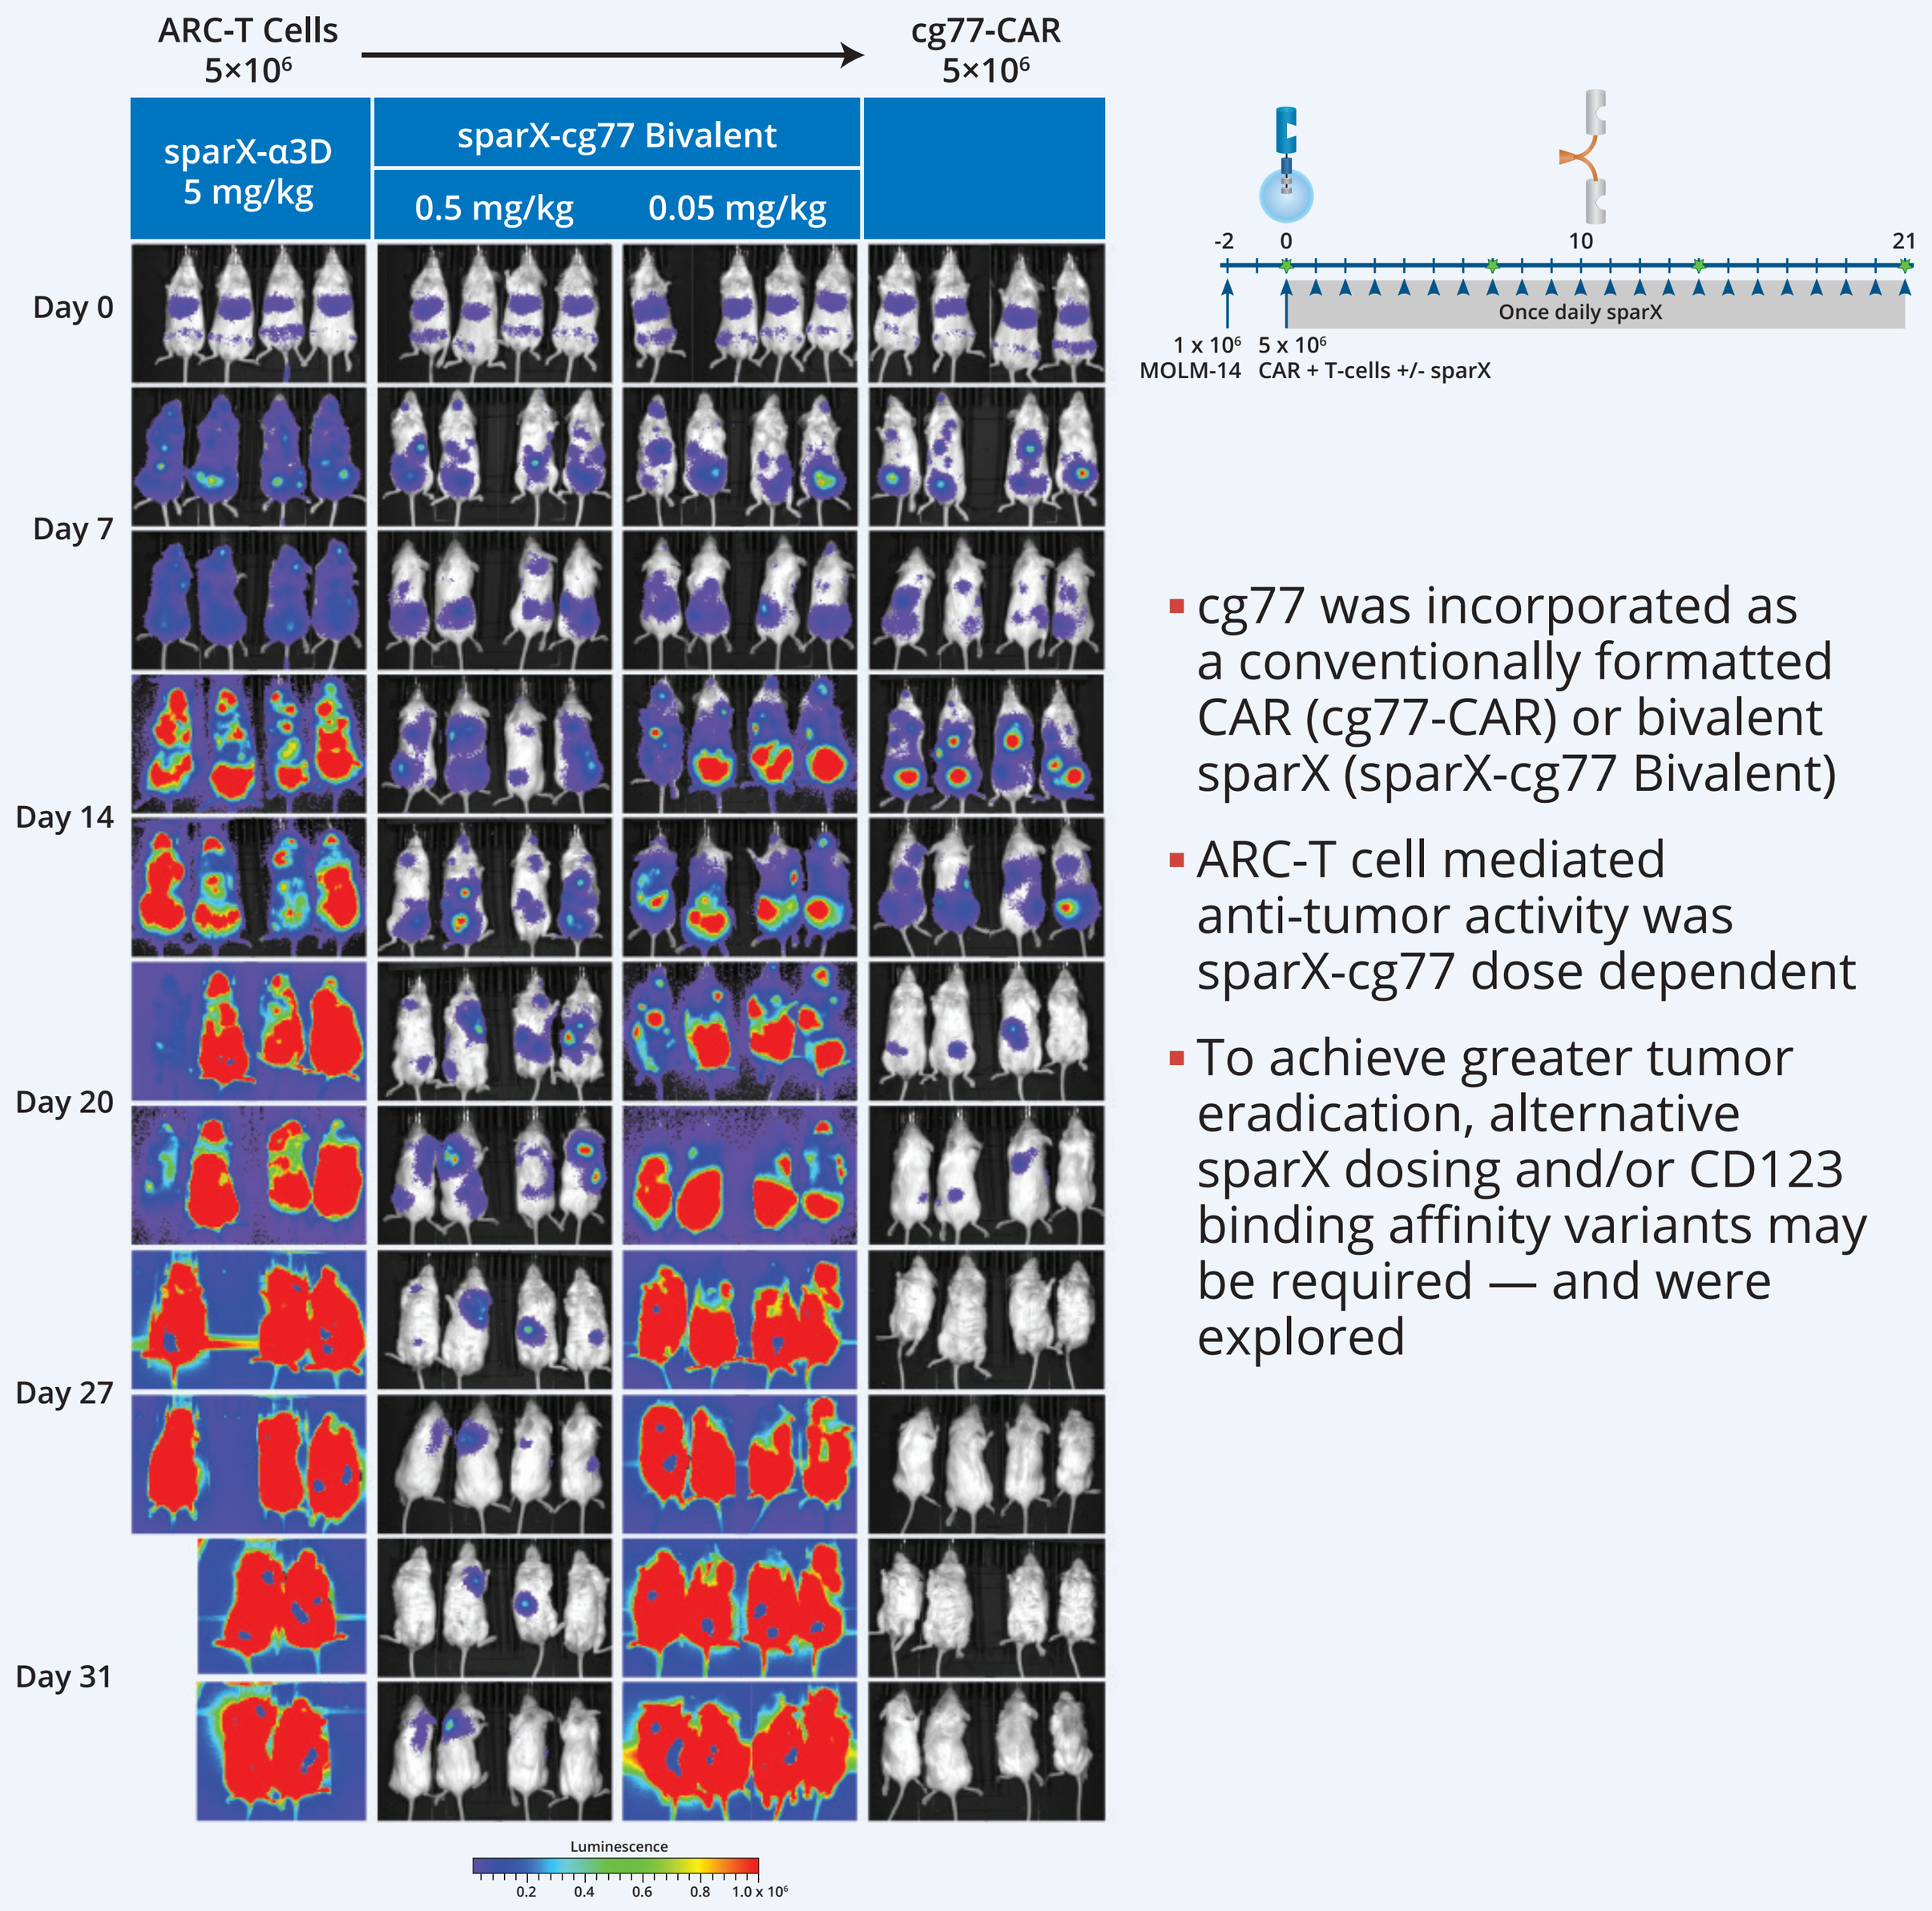

ARC-T Cells 5×10⁶
cg77-CAR 5×10⁶
sparX-α3D 5 mg/kg
sparX-cg77 Bivalent
0.5 mg/kg
0.05 mg/kg
Day 0
Day 7
Day 14
Day 20
Day 27
Day 31
Once daily sparX
1 x 10⁶ MOLM-14
5 x 10⁶ CAR + T-cells +/- sparX
Luminescence

- cg77 was incorporated as a conventionally formatted CAR (cg77-CAR) or bivalent sparX (sparX-cg77 Bivalent)
- ARC-T cell mediated anti-tumor activity was sparX-cg77 dose dependent
- To achieve greater tumor eradication, alternative sparX dosing and/or CD123 binding affinity variants may be required — and were explored

D Domains are Affinity Matured Through Mutagenesis

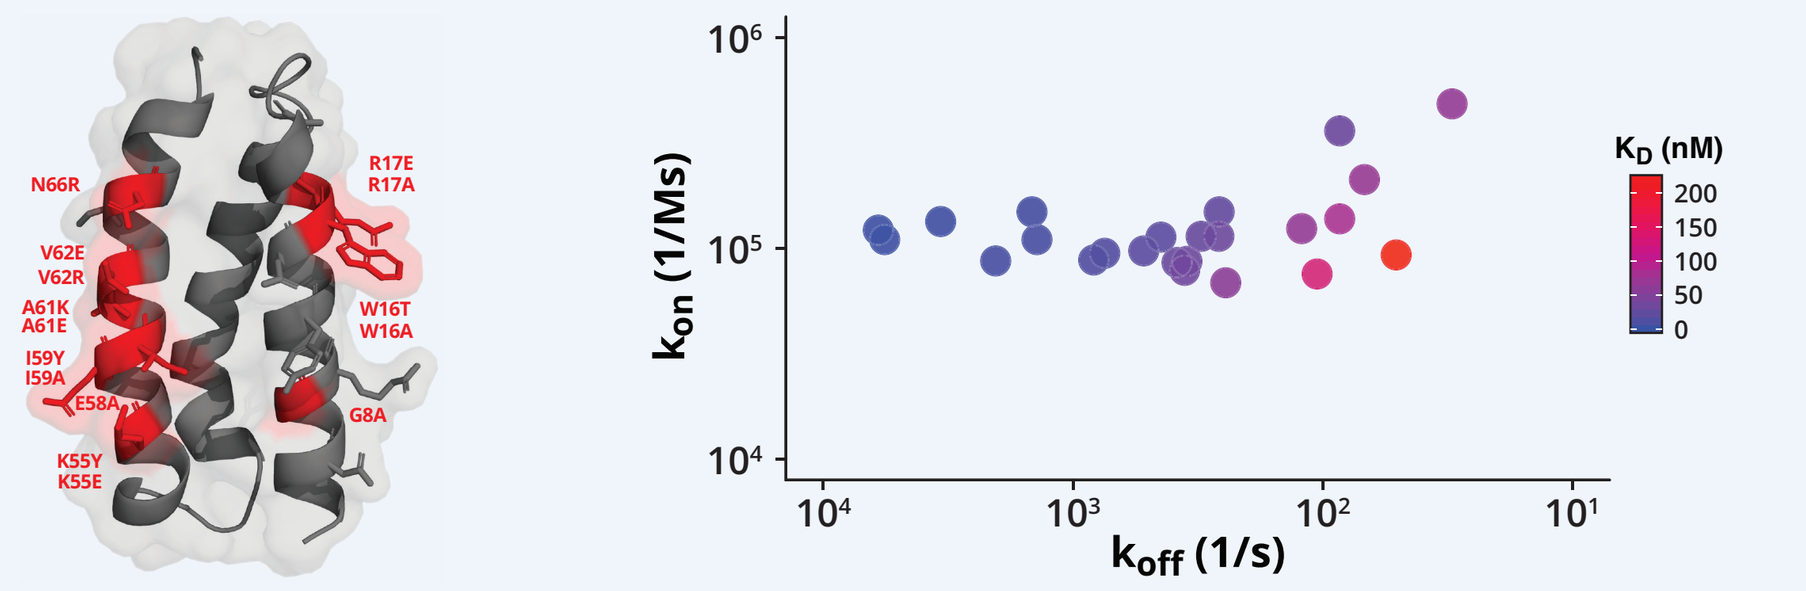

N66R
V62E V62R
A61K A61E
I59Y I59A
E58A
K55Y K55E
R17E R17A
W16T W16A
G8A
k_{on} (1/Ms)
k_{off} (1/s)
K_D (nM)

- A CD123-binding D domain was subjected to mutagenesis to identify point mutations that could either increase or decrease binding affinity
- A homology model of the scaffold illustrates the location of representative mutations in red **(left)**
- D domain mutants were expressed as monovalent MBP-fusions and binding kinetics were assessed by surface plasmon resonance **(right)**
- K_D values for binding of CD123-HIS to D domains spanned a 150-fold range (1.4 nM to 212 nM), with off-rate contributing more to the distribution than on-rate, suggesting these mutations impact stabilization of the complex, once formed

sparX Incorporating High Affinity D Domain Enhances Potency of ARC-T Cells

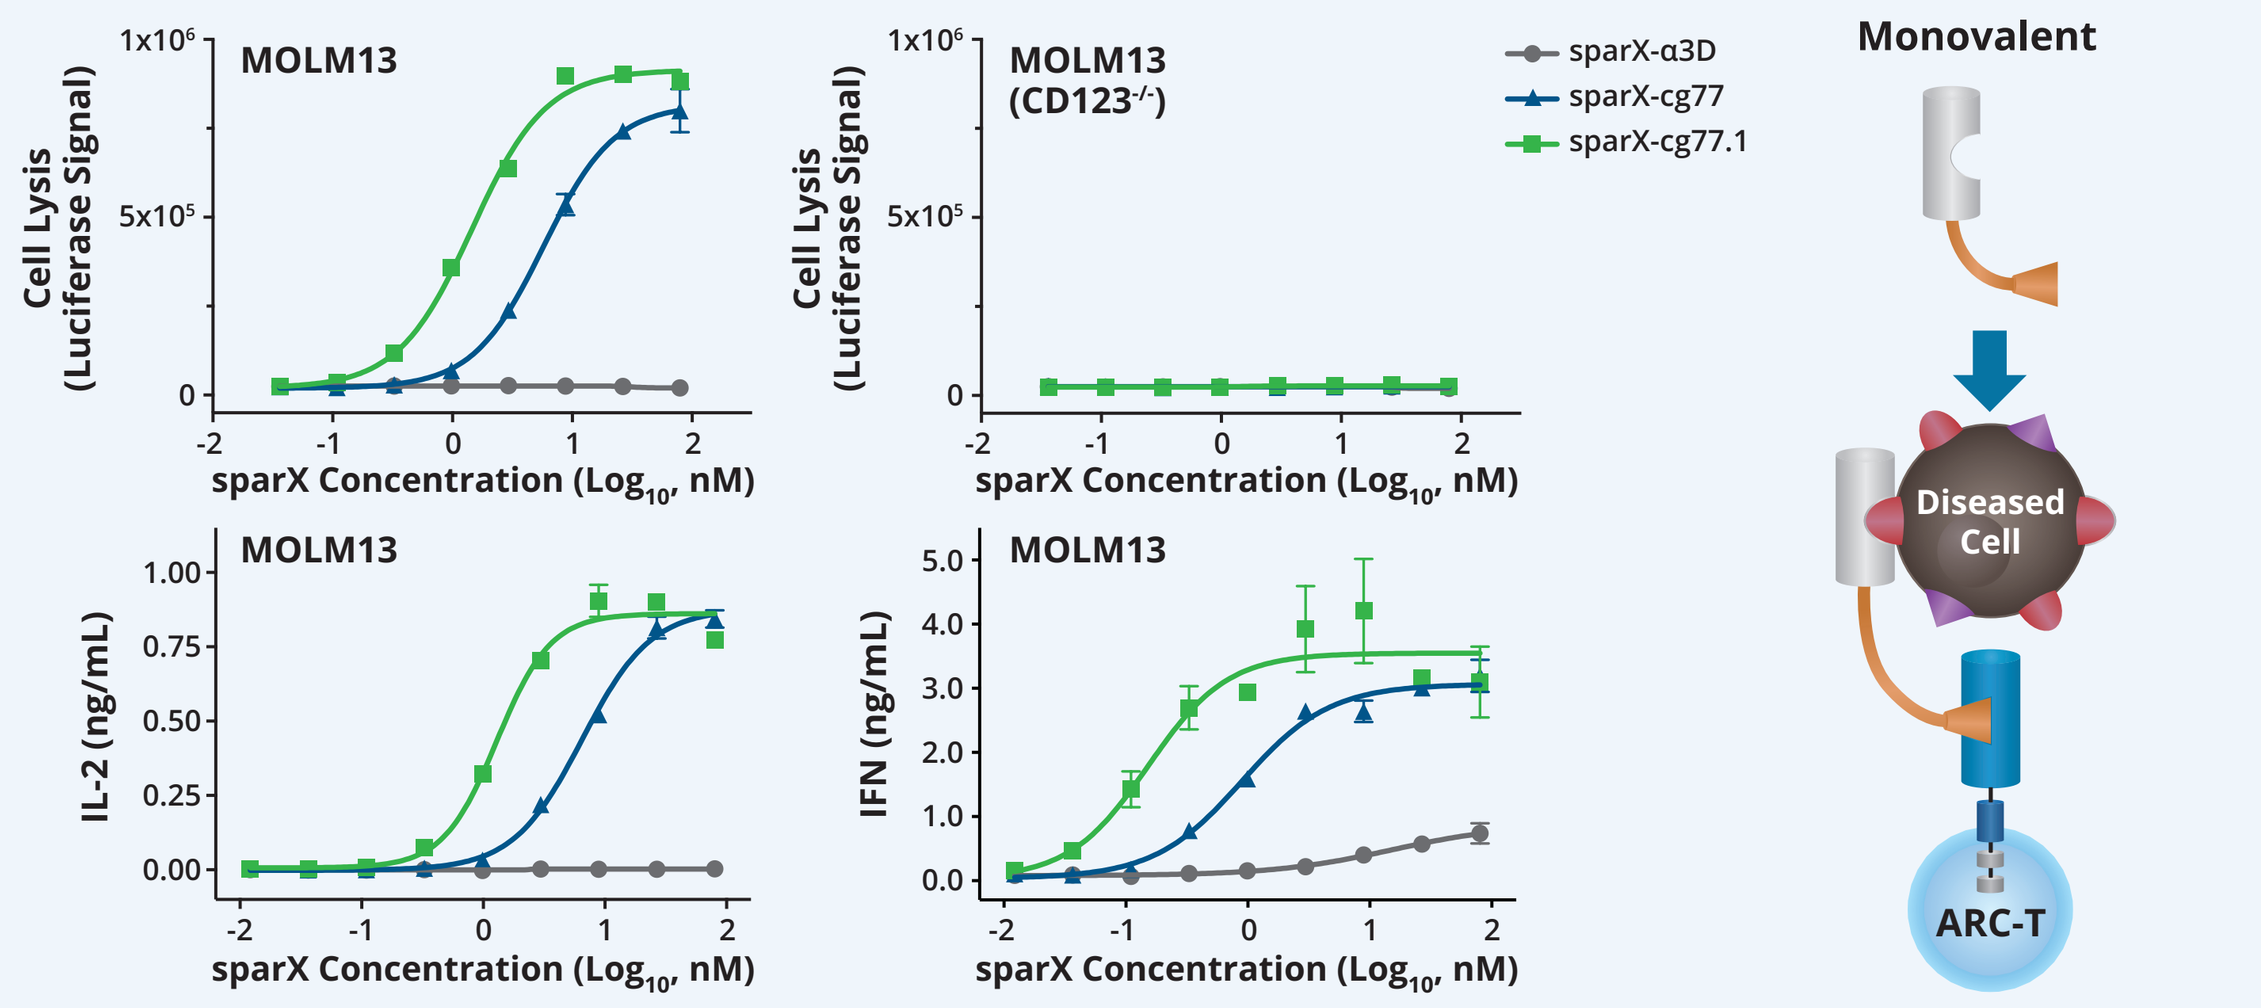

MOLM13
MOLM13 (CD123⁻/⁻)
Cell Lysis (Luciferase Signal)
IL-2 (ng/mL)
IFN (ng/mL)
sparX Concentration (Log10, nM)
sparX-α3D
sparX-cg77
sparX-cg77.1
Monovalent
Diseased Cell
ARC-T

- Monovalent sparX were generated with the cg77 D domain or a high-affinity variant (sparX-cg77.1), with an approximately 10-fold lower K_D, as measured by SPR
- As measured by luciferase signal, ARC-T cell mediated lysis of AML-derived MOLM13/luciferase cells requires CD123-binding sparX **(top left)**, CD123 expression on target cells **(top right)** and ARC-T cells (not shown)
- ARC-T cell mediated target cell lysis **(top left)**, IL-2 (bottom left) release and IFN release **(bottom right)** are dose and affinity dependent
- As measured by EC50 and as compared to the parental sparX-cg77 the high-affinity variant (sparX-cg77.1) demonstrated enhanced potency for target cell lysis (4-fold increase), IL-2 release (5-fold increase) and IFN release (6-fold increase)

Combinations of Affinity and Valency of D Domain Can Be Used to Further Broaden Dynamic Range of sparX

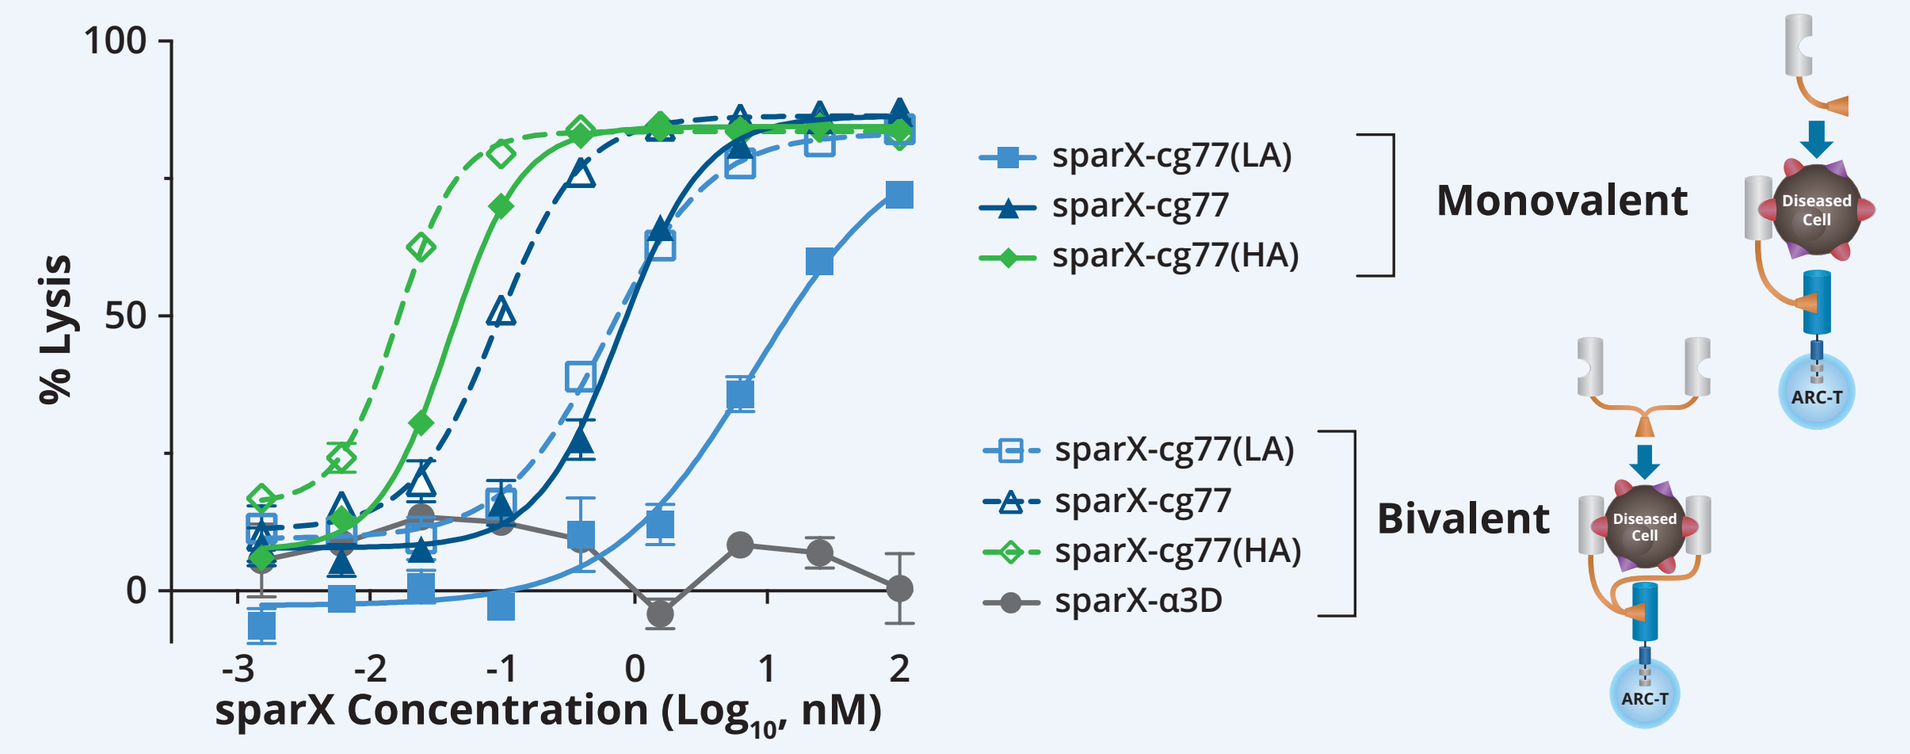

% Lysis
sparX Concentration (Log10, nM)
sparX-cg77(LA)
sparX-cg77
sparX-cg77(HA)
Monovalent
sparX-α3D
Bivalent

- sparX were generated in both mono and bivalent formats, utilizing the parent cg77 D domain or variants incorporating high affinity (HA) or low affinity (LA) mutations
- ARC-T cell mediated lysis of CD123-expressing, NALM6 cells is modulated by CD123-binding sparX
- The extent of target cell lysis is dose-dependent for each sparX and collectively the EC_{50}s span 3 logs
- Low affinity sparX manifest greatest gain in potency when formatted as a bivalent
- The high affinity, monovalent sparX (sparX-cg77 [HA]) outperforms the bivalent sparX-cg77

Monovalent, High-Affinity CD123 sparX Eliminates Detectable AML Tumors *In Vivo*

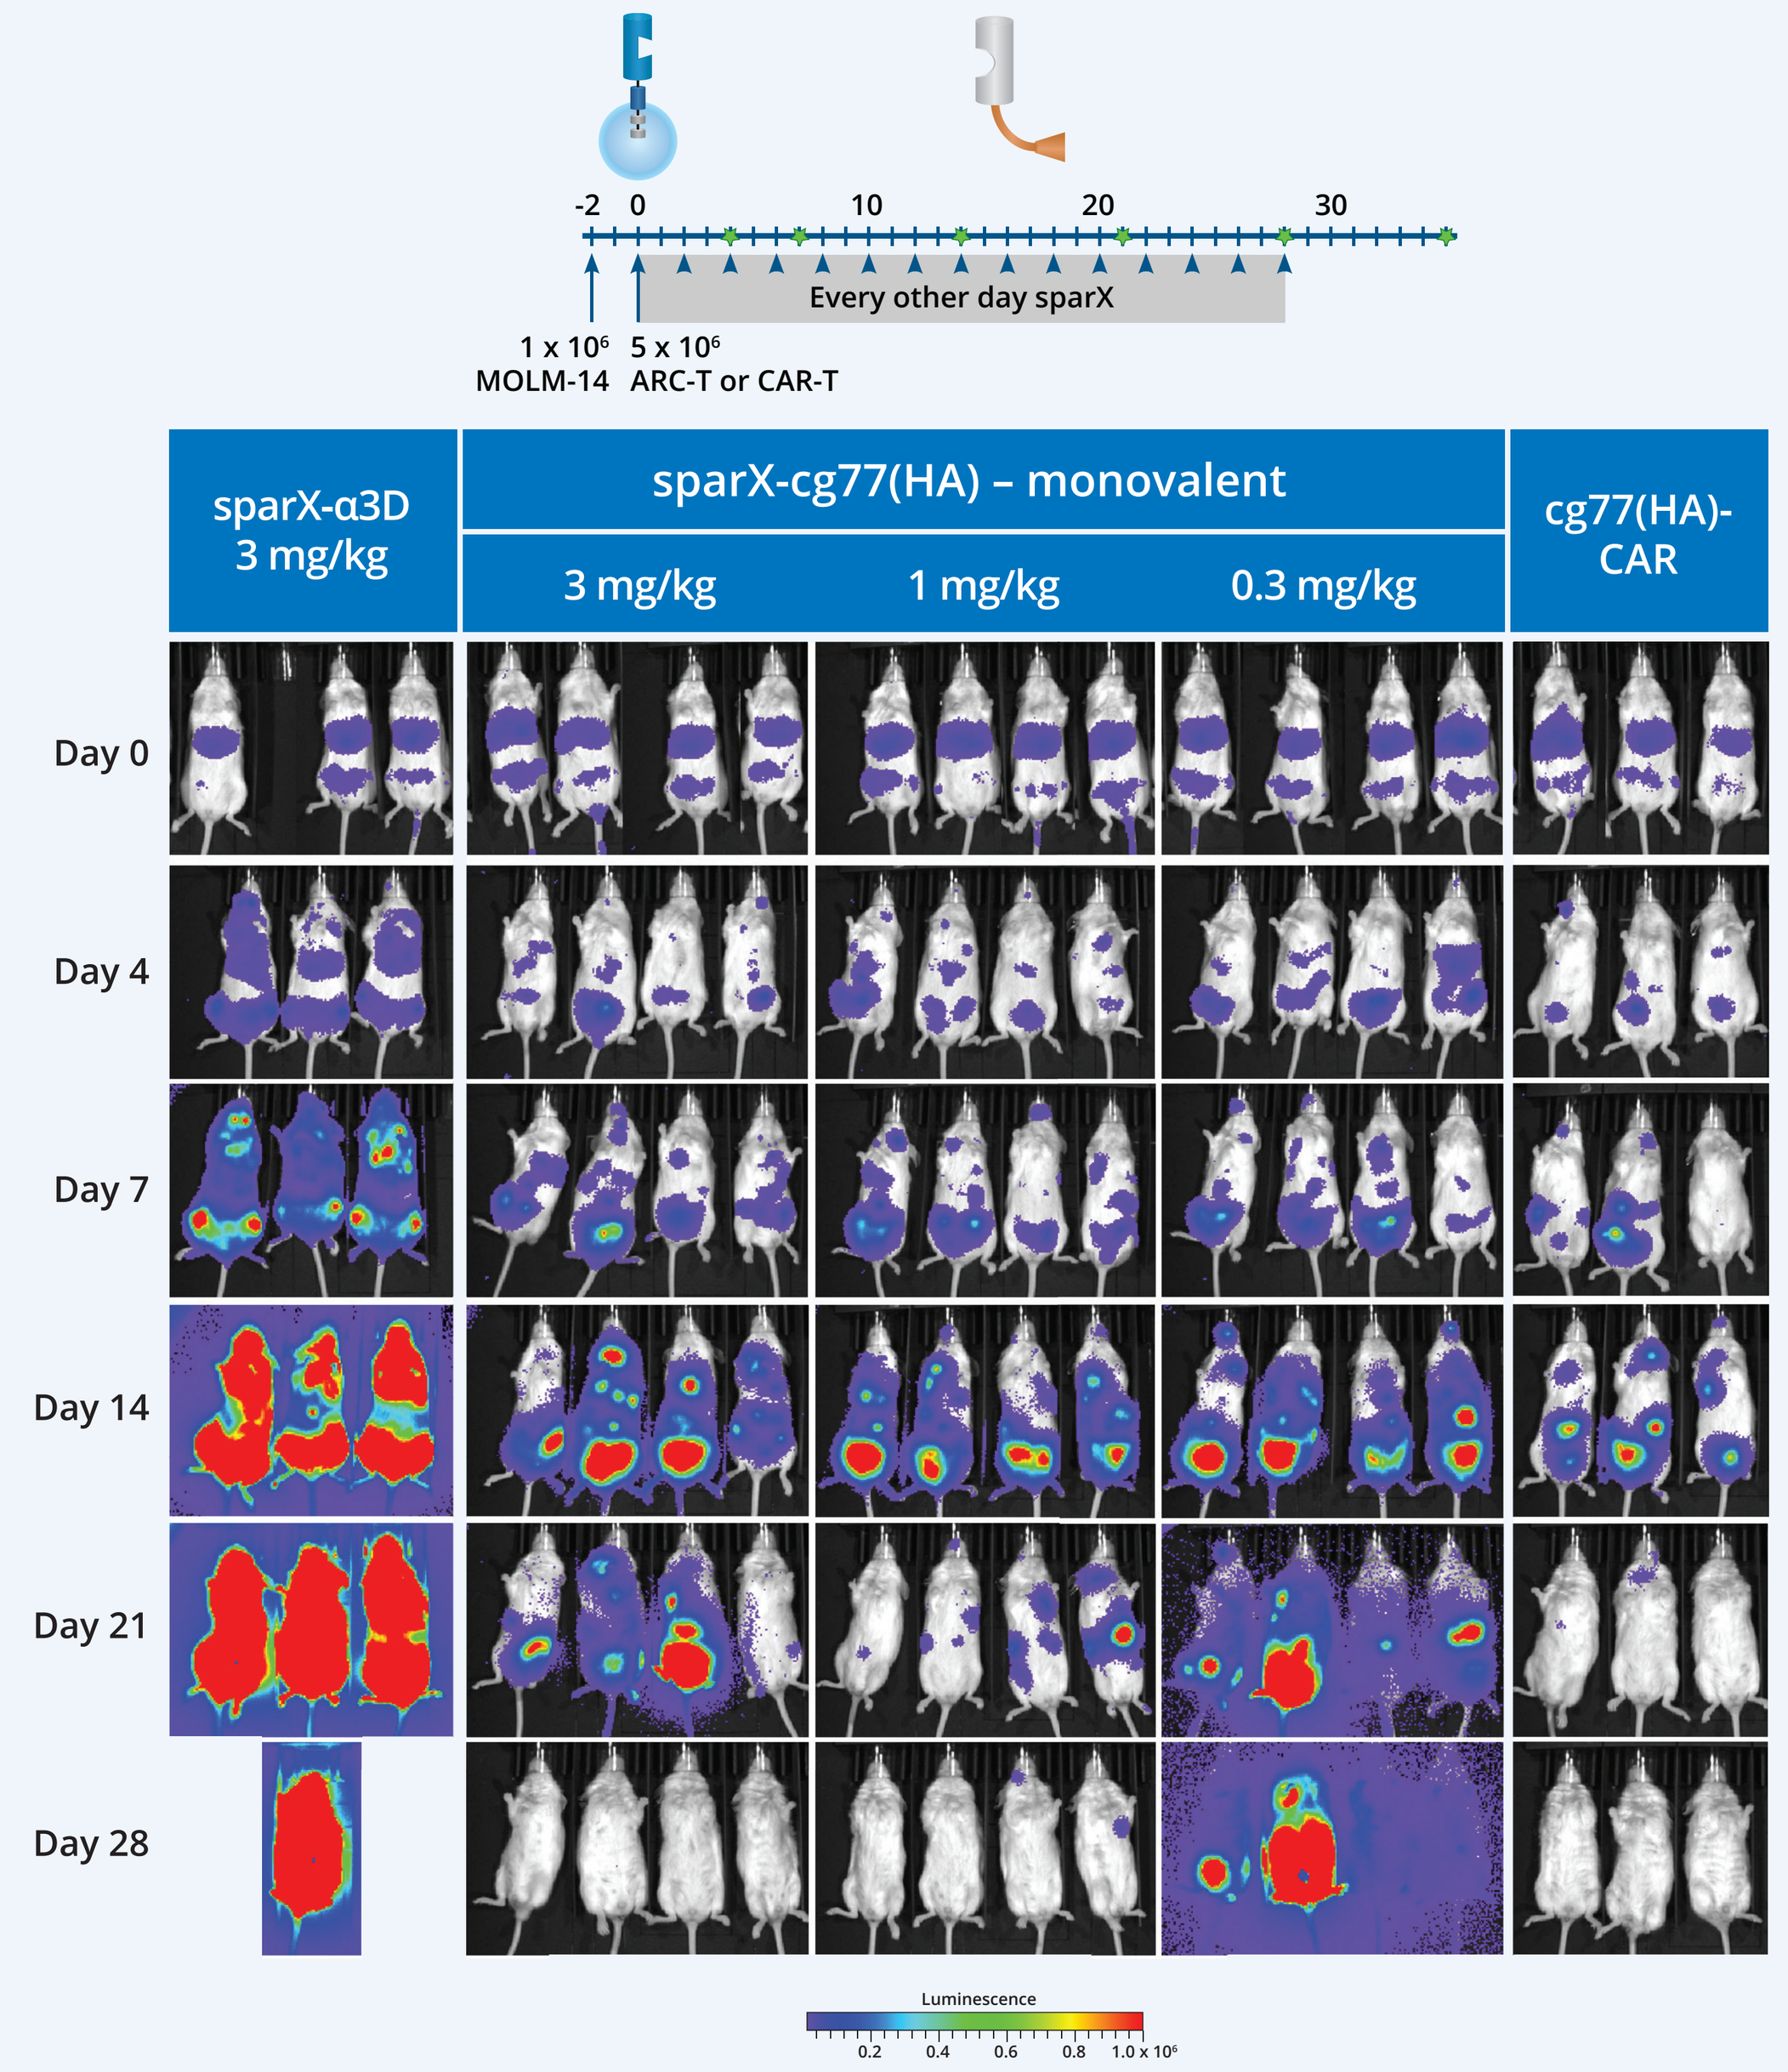

Every other day sparX
1 x 10⁶ MOLM-14
5 x 10⁶ ARC-T or CAR-T
sparX-α3D 3 mg/kg
sparX-cg77(HA) – monovalent
3 mg/kg
1 mg/kg
0.3 mg/kg
cg77(HA)-CAR
Day 0
Day 4
Day 7
Day 14
Day 21
Day 28
Luminescence

- A high affinity CD123 D domain is incorporated into monovalent sparX (sparX-cg77[HA]) and a conventionally formatted CAR (cg77[HA]-CAR)
- *In vivo* anti-tumor activity of MOLM14 tumors by ARC-T is dependent on sparX dose
- Cohorts receiving 1 and 3 mg/kg sparX-cg77(HA), every other day, achieved comparable anti-tumor activity to that of the conventionally formatted CAR

Conclusions

- As exemplified by D domains discovered and optimized to bind CD123, D domains constitute a robust scaffold for development of therapeutic targeting domains
- CAR T cells comprised of D domains targeting CD123, direct potent *in vivo* anti-tumor activity of AML-derived tumors comparable to that of a conventional, scFv-based CAR
- The ARC-sparX platform affords the ability to modulate T-cell activity by controlling the specificity, dose, affinity and valency of the sparX protein
- sparX proteins targeting CD123, in combination with ARC-T cells, inhibit the growth of AML-derived tumors *in vivo* and may offer a novel therapeutic approach for treating AML
- More broadly, the ARC-sparX platform affords an opportunity for treating other challenging cell therapy indications where greater T-cell control may be warranted

References

1. Qin, Haiying, et al. "Chimeric antigen receptors incorporating D domains targeting CD123 direct potent mono- and bi- specific antitumor activity of T cells." *Molecular Therapy* 27.7 (2019): 1262-1274.
2. Zhu Y, Alonso DO, Maki K, et al. Ultrafast folding of alpha3D: a de novo designed three-helix bundle protein. *Proc Natl Acad Sci* U S A. 003;100(26):15486-15491. doi:10.1073/pnas.2136623100.
3. Baker MP, Jones TD. Identification and removal of immunogenicity in therapeutic proteins. *Curr Opin Drug Discov Devel.* 2007;10(2): 219-227.

Presented at the American Association for Cancer Research 2020 Virtual Annual Meeting II, June 22-24, 2020

©2020 Arcellx, Inc. All rights reserved.