Abstract 3041

Selective Targeting of HER2-overexpressing Solid Tumors with a Next-generation CAR-T Cell Therapy

Jenny Mu, Justin Edwards, Liubov Zaritskaya, Jeffrey Swers, Ankit Gupta, David LaFleur, David Hilbert, Laura Richman

Arcellx, Inc., Gaithersburg, MD

jmu@arcellx.com

Introduction

- Conventional chimeric antigen receptor T cell (CAR-T) therapies have achieved limited clinical success in the treatment of solid tumors, in part due to the challenges of identifying tumor antigen(s) that are uniquely expressed on tumor cells[1,2]
- To enhance safety and efficacy, CAR-T therapies must be engineered to preferentially target tumor cells and mitigate potential on-target off-tumor toxicity to normal cells[3]
- We have developed a novel cell therapy platform comprising Antigen Receptor Complex T (ARC-T) cells that are readily activated and reprogrammed *in vivo* by administration of a novel tumor-targeting soluble protein antigen-receptor X-linker (sparX)[4]
 - sparX proteins can be engineered to target different antigens
 - Universal ARC-T cell product can be paired with any sparX
- Because ARC-T cell activity is entirely dependent on the amount of sparX administered, therapeutic doses of sparX may be defined that preferentially target cells over-expressing a target antigen and thus limit coincident kill of normal cells expressing lower antigen levels

ARC-sparX Platform

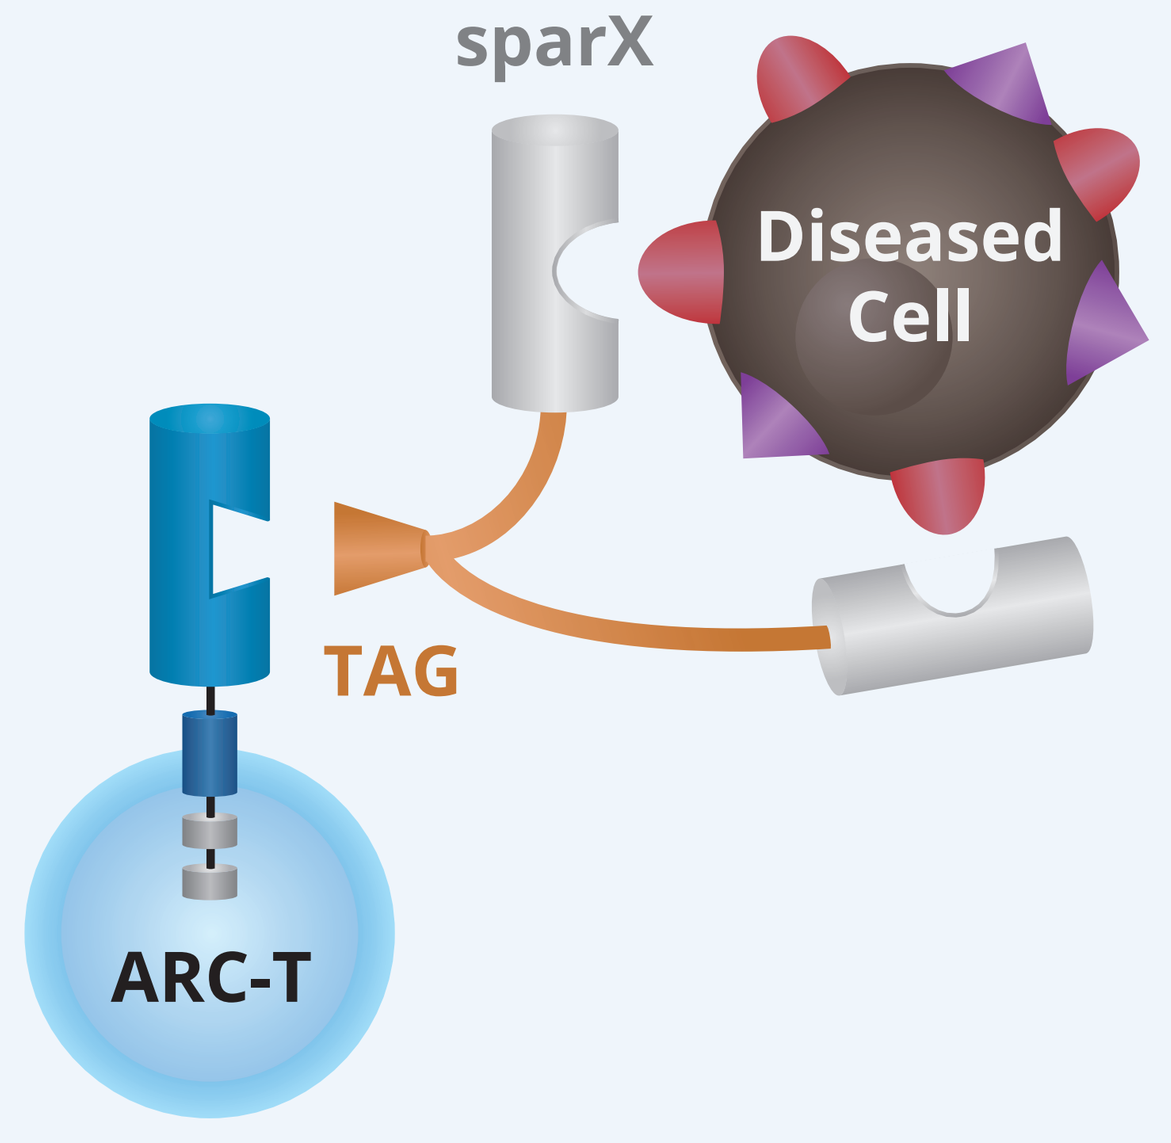

sparX
Diseased Cell
TAG
ARC-T

sparX Protein

- Comprised of one or more novel binding domains fused to the TAG protein, a fragment of human alpha fetoprotein (AFP)
- Designed to recognize specific antigens on diseased cells and flag those cells for destruction
- No inherent therapeutic activity
- Engineered to minimize immunogenic potential

ARC-T Cell

- Utilizes a universal Antigen Receptor Construct (ARC) that can be expressed in T cells and bind sparX proteins to kill flagged cells
- Conventional CAR architecture with novel binding domain specific for TAG
- Same viral vector regardless of antigen target
- ARC is anticipated to function in allogeneic based cell therapies

Tri-complex of ARC-T cell + sparX + diseased cell must be formed to activate cytolytic killing.

Background

Novel Binding Domains and TAG Confer Advantages to ARC-sparX Platform

Novel Binding Domains

- 8kDa/73aa non-scFv binding domain based on a3D scaffold[5]
- Target specific binding domains can be identified from a3D scaffold-based libraries where external facing residues (Blue regions) are randomized[6]
- Target binding affinities comparable to that of scFv
- Engineered to minimize immunogenic potential

sparX TAG

- ARC-T binding domain is specific for the sparX TAG
- AFP demonstrates many features of an ideal tag
 - Non-immunogenic with pre-established tolerance in humans
 - *In vitro* and *in vivo* stability
- ARC-T binds the AFP fragment, TAG, but NOT intact AFP, thereby eliminating potential competition between endogenous AFP and sparX TAG for ARC

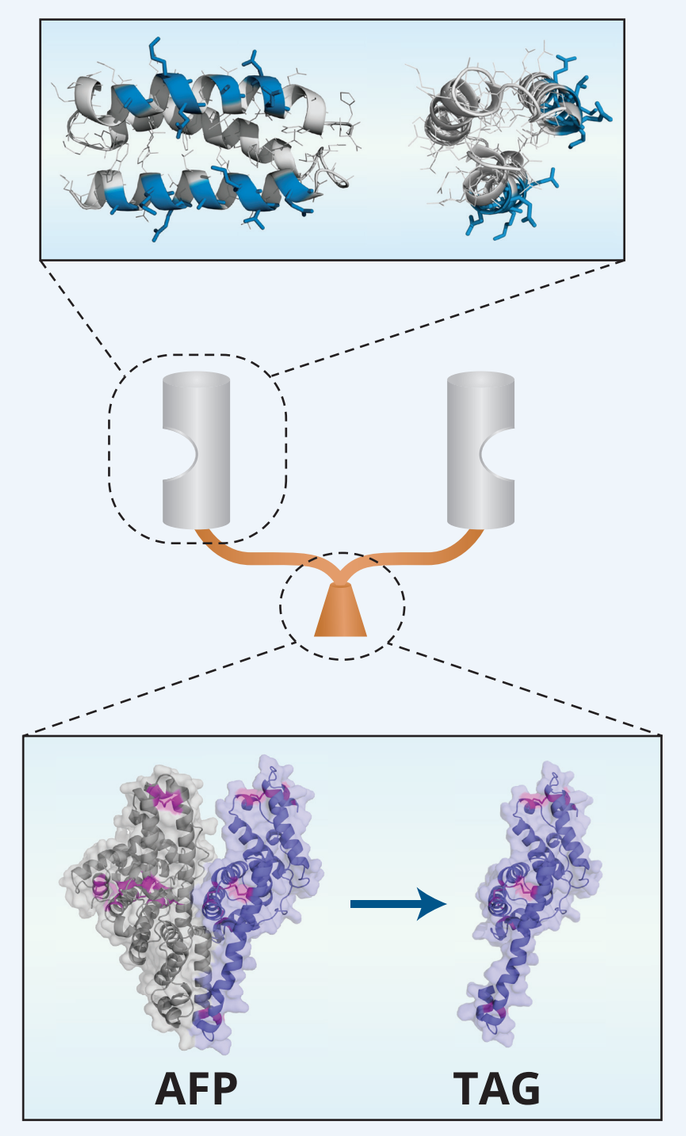

AFP
TAG

sparX Proteins Can Be Engineered in Multi-valent and Multi-specific Configurations to Change Binding Affinity and Permit Targeted Killing

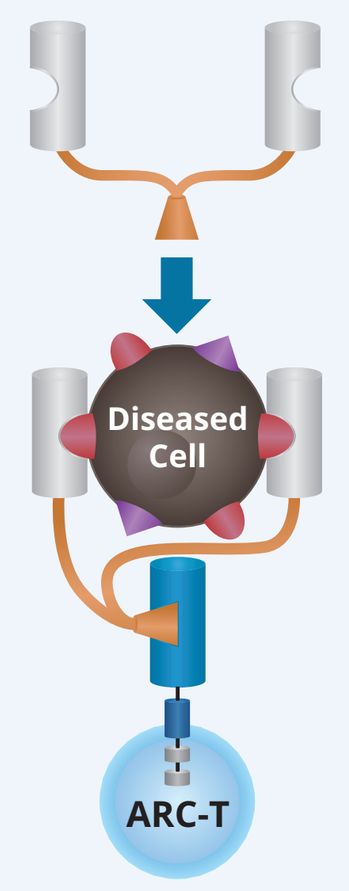

Diseased Cell
ARC-T

Bi-valent sparX Therapy

- Multiple binding regions for the ***same*** target antigen result in higher binding affinity, also referred to as avidity, which can enable:
 - Enhanced tumor killing at lower doses
 - ***Sensitivity to antigen receptor density***[7]

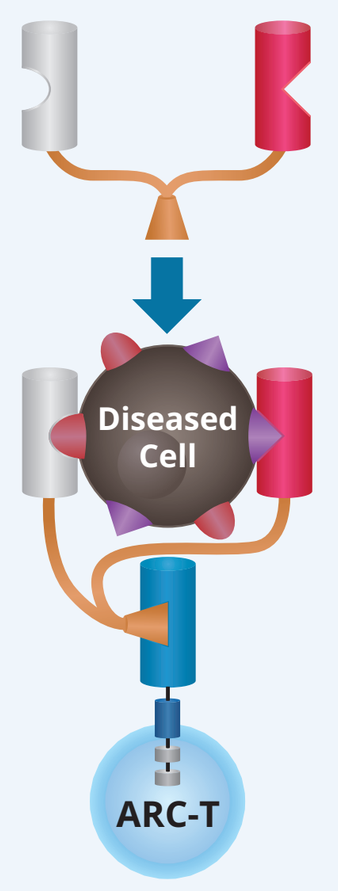

Diseased Cell
ARC-T

Bi-specific sparX Therapy

- Multiple binding regions for ***different*** target antigens can:
 - Mediate higher avidity interaction to direct ARC-T cell killing of only those cells that express two (or more) of the target antigens (***AND*** gating)
 - Address tumor heterogeneity as each binding region can engage at lower affinity with cells expressing only one of the antigens (***OR*** gating)

Results

ARC is Efficiently Expressed on Primary Human T Cells with Comparable Percentages of CAR/ARC-positive T Cells as Conventional scFv-based CAR

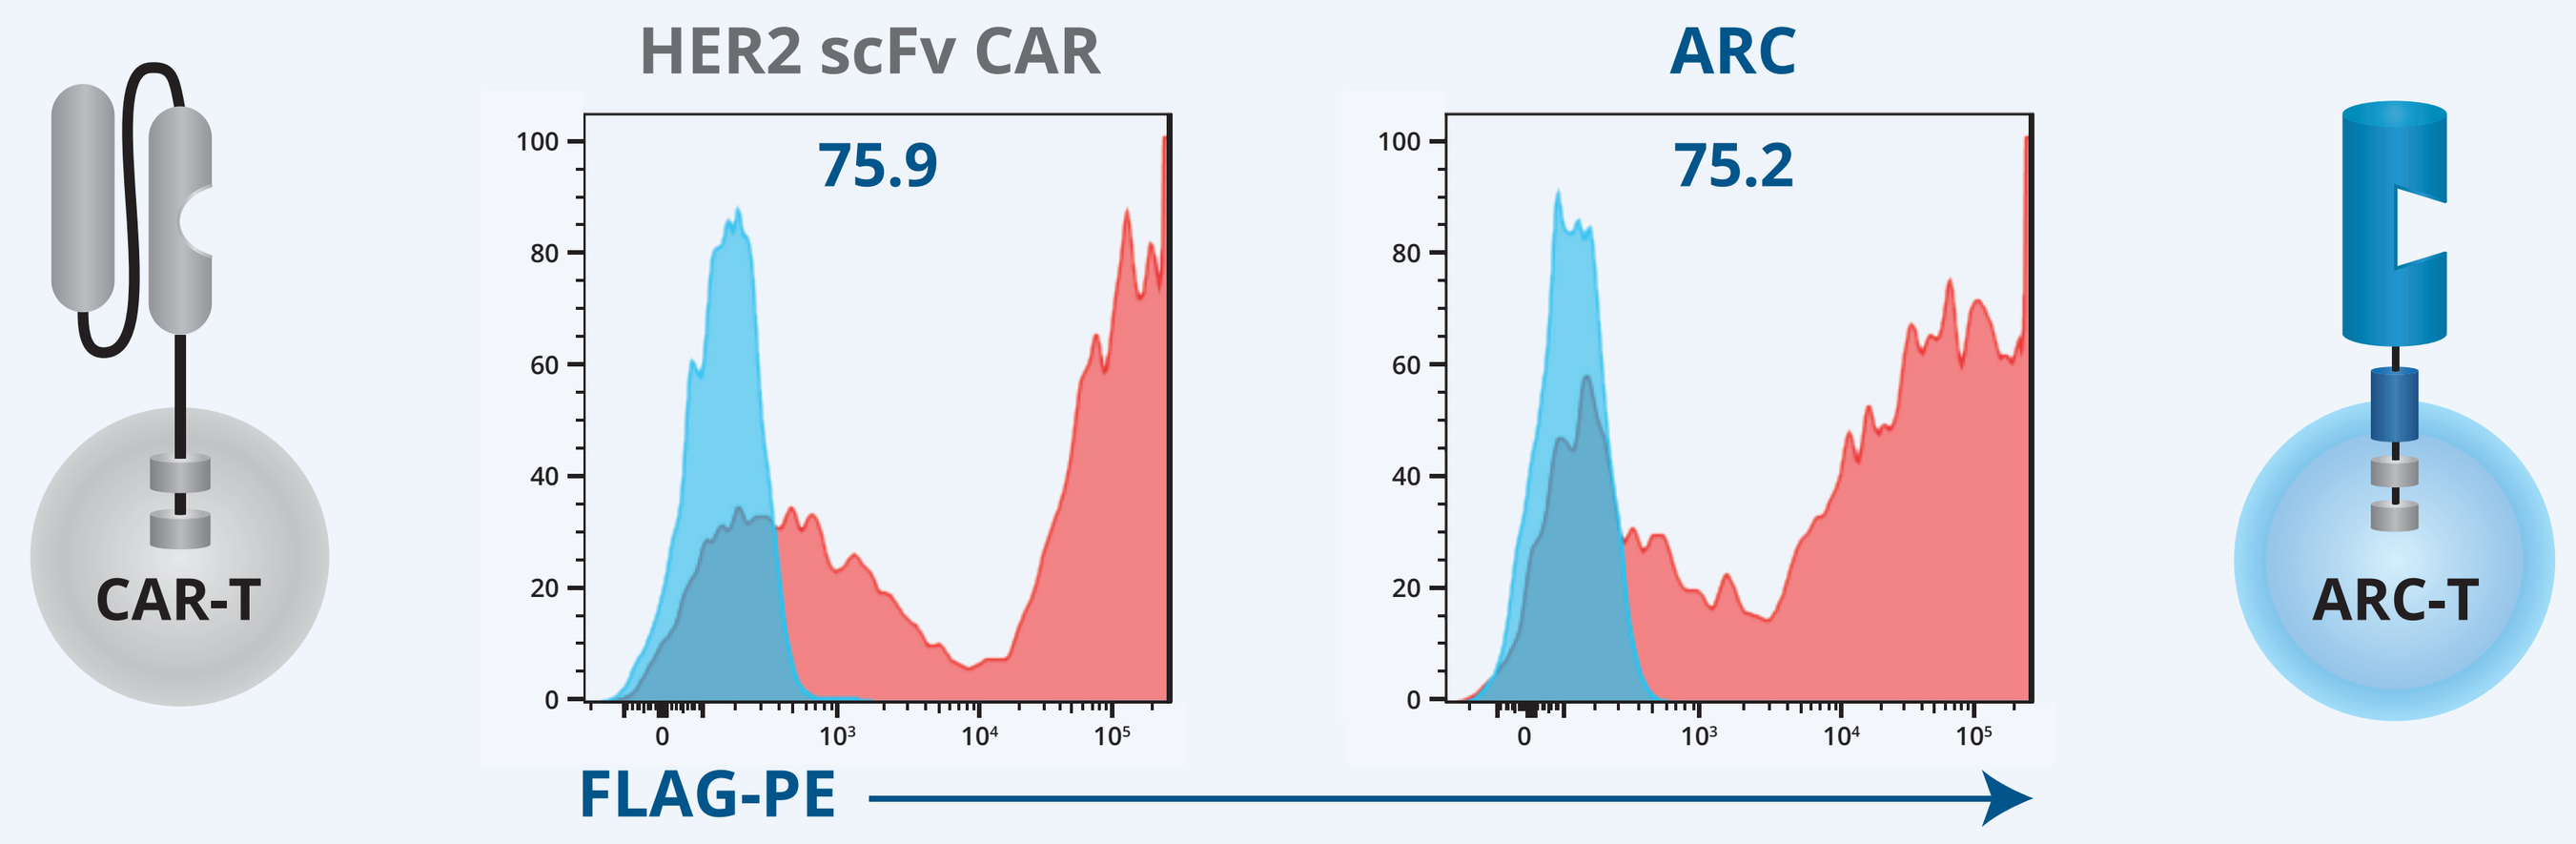

CAR-T
HER2 scFv CAR
75.9
ARC
75.2
FLAG-PE
ARC-T

- ARC and CAR expression was determined by flow cytometry using a PE-labeled antibody against the N-terminal FLAG fused to each receptor

Bi-valent sparX-HER2 Protein Directs More Potent Tumor Cell Killing than its Mono-valent Form

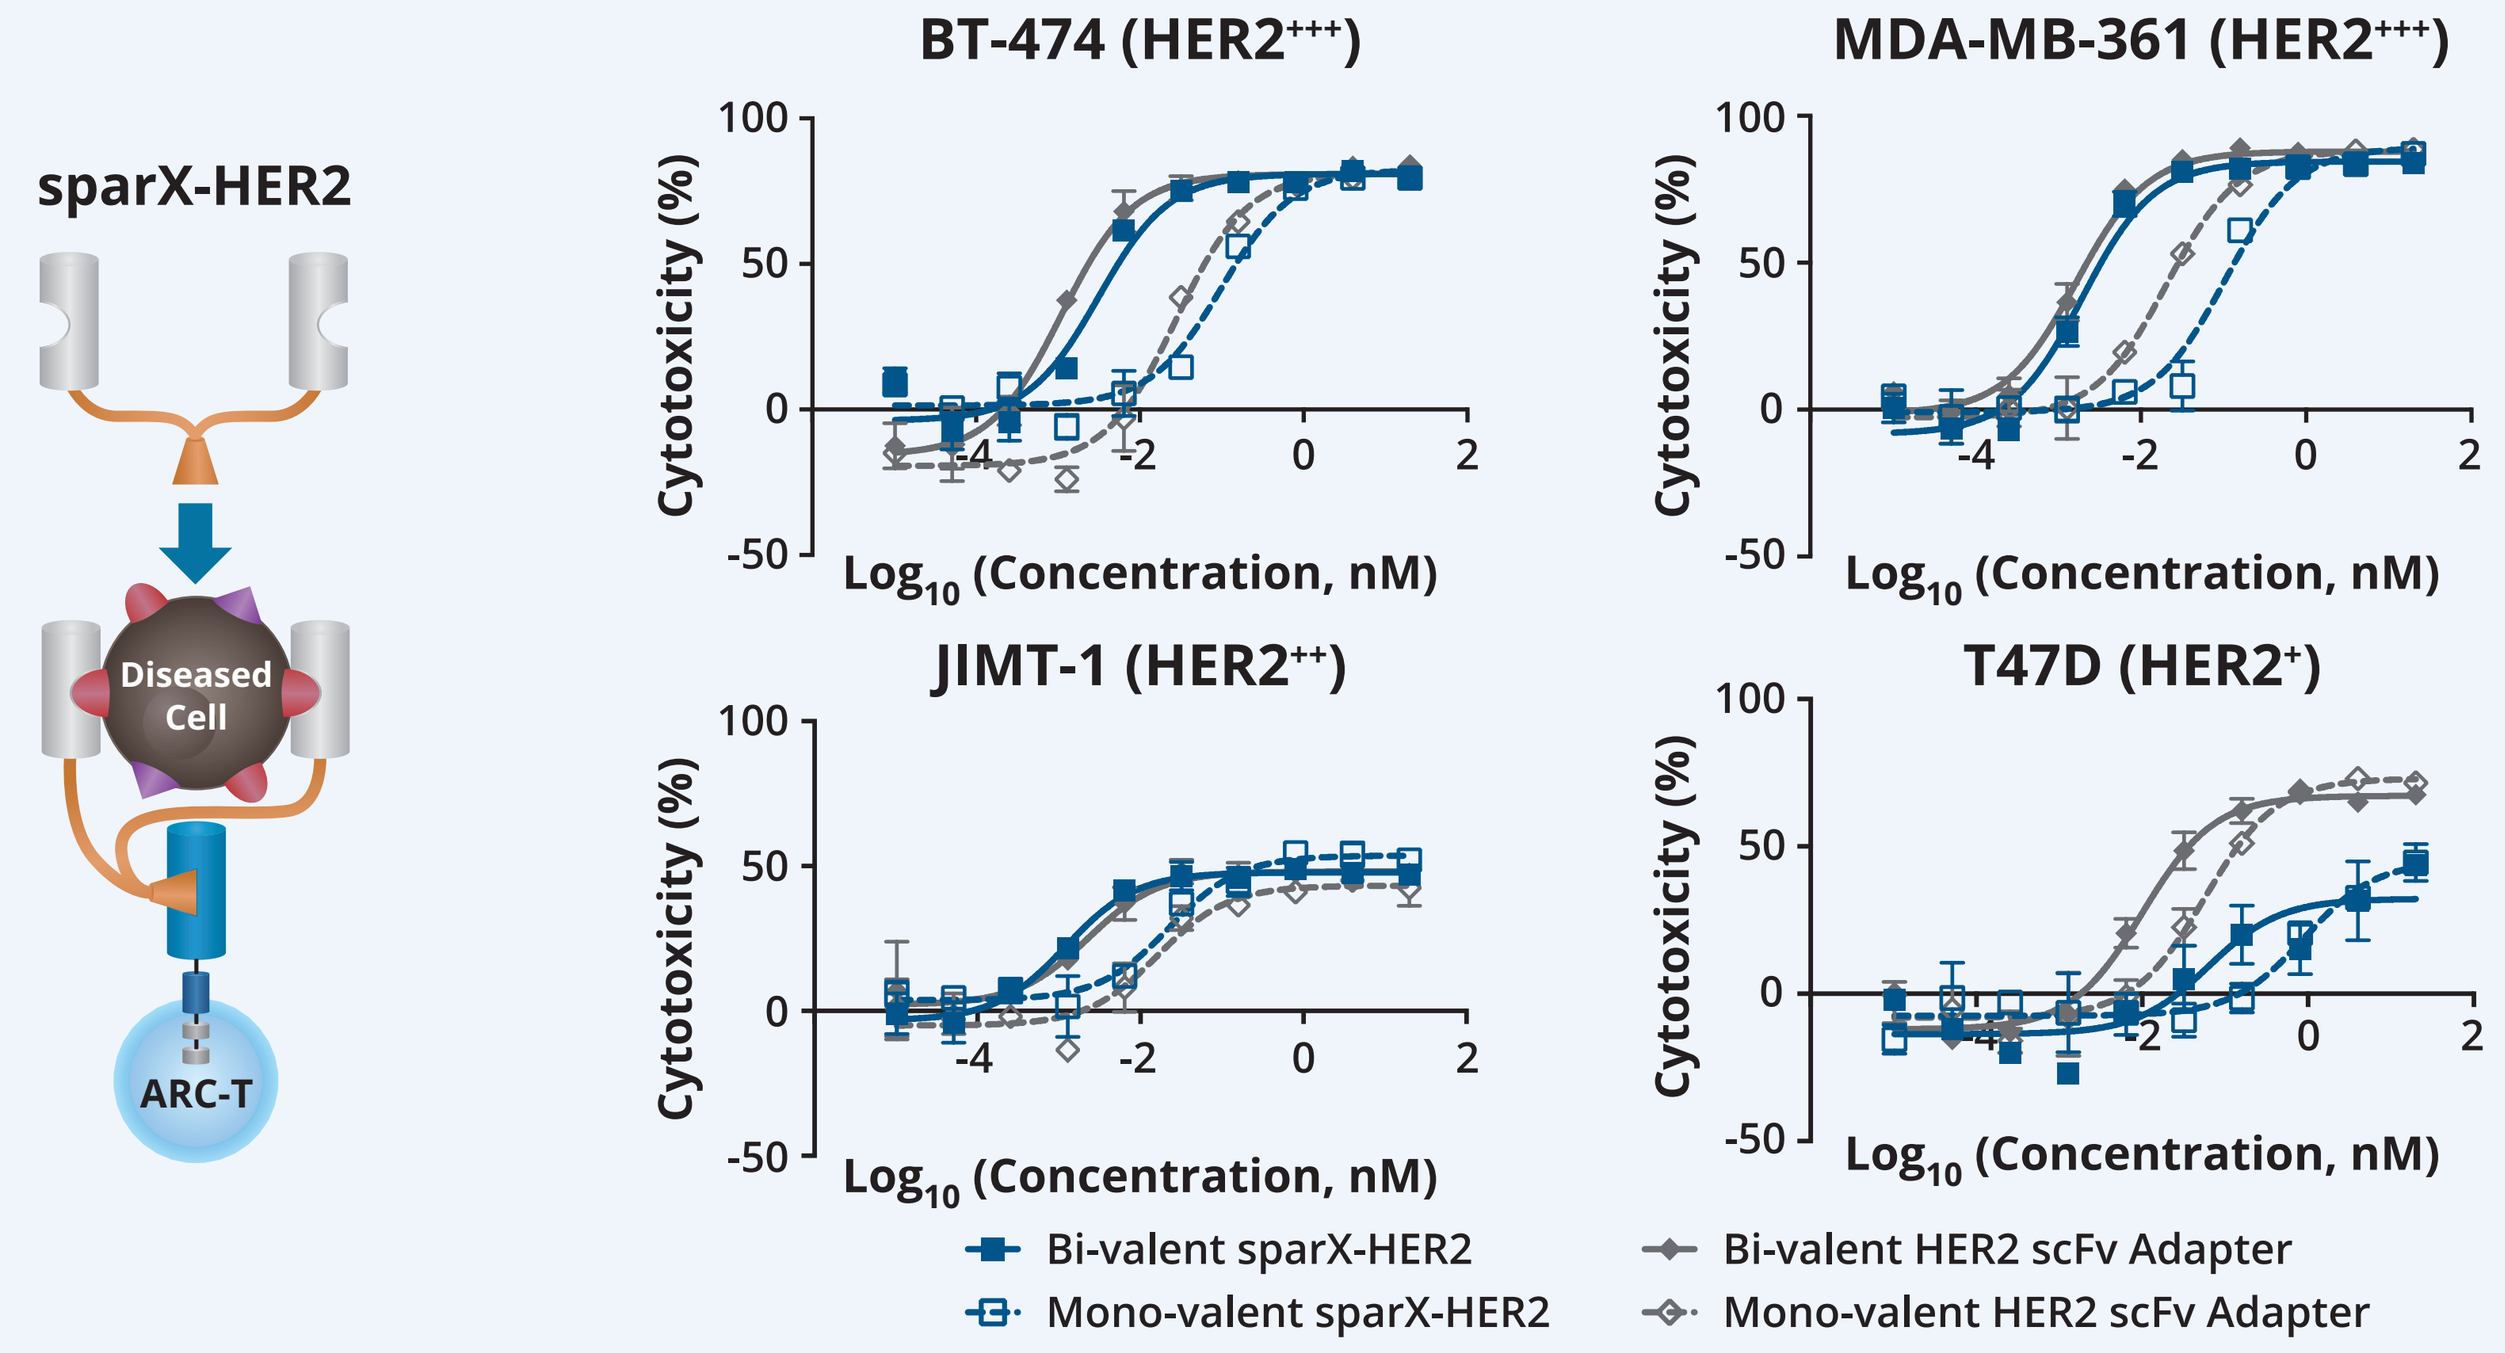

sparX-HER2
Diseased Cell
ARC-T
BT-474 (HER2+++)
MDA-MB-361 (HER2+++)
JIMT-1 (HER2++)
T47D (HER2+)
Cytotoxicity (%)
Log10 (Concentration, nM)
Bi-valent sparX-HER2
Mono-valent sparX-HER2
Bi-valent HER2 scFv Adapter
Mono-valent HER2 scFv Adapter

- HER2 scFv Adapter is comprised of a HER2 mAb-derived scFv as the binding domain fused to the TAG as in the sparX protein
- sparX-HER2 achieves the same maximum cytolytic effect as the HER2 scFv adapter in response to cancer cell lines with HER2 expression levels greater than IHC 1+

Bi-valent sparX-HER2 Mediates ARC-T Cell Cytokine Release and Proliferation Only in Response to Cells with High HER2 Expression

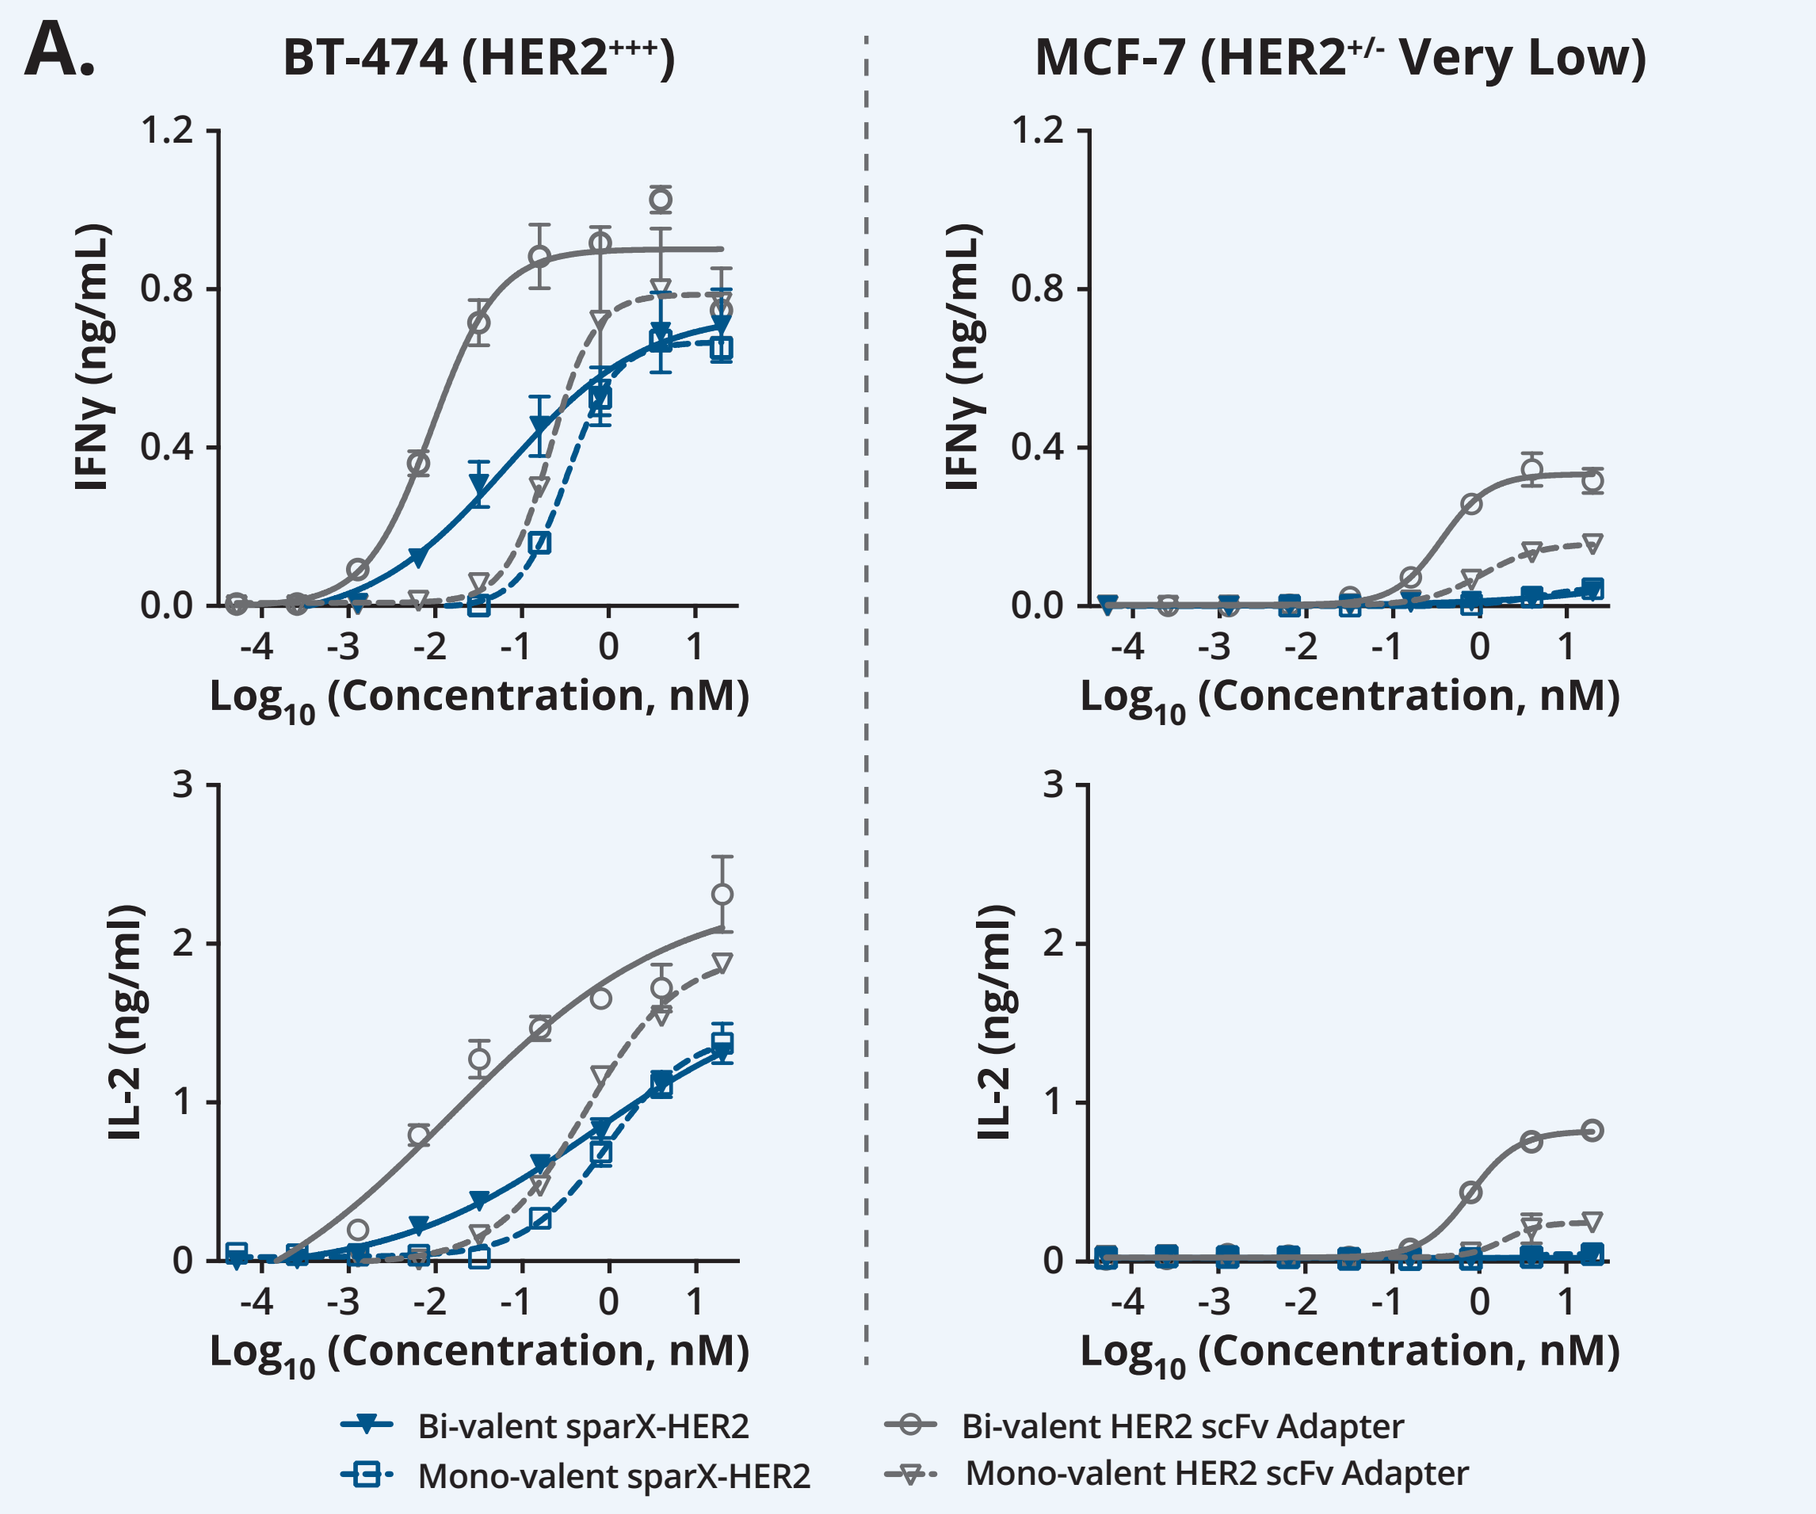

A.
BT-474 (HER2+++)
MCF-7 (HER2+/- Very Low)
IFNγ (ng/mL)
IL-2 (ng/ml)
Log10 (Concentration, nM)
Bi-valent sparX-HER2
Mono-valent sparX-HER2
Bi-valent HER2 scFv Adapter
Mono-valent HER2 scFv Adapter

- HER2 scFv Adapter directs ARC-T cell cytokine production in response to cells without regard to HER2 amplification status
- Bi-valent sparX-HER2 imparts selectivity to ARC-T cells to ensure cytokine production only in the presence of HER2-overexpressing cells

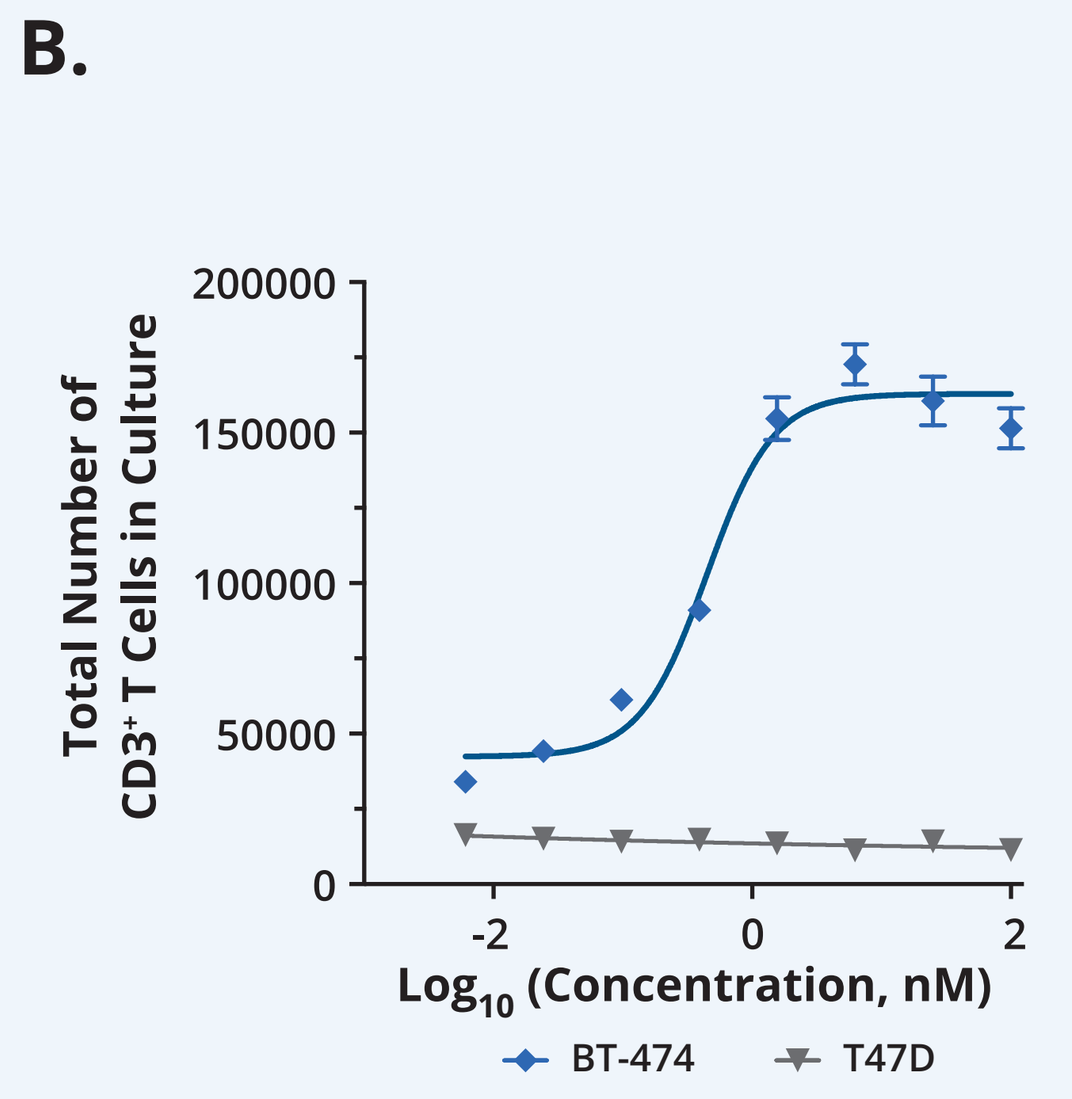

B.
Total Number of CD3+ T Cells in Culture
Log10 (Concentration, nM)
BT-474
T47D

- Bi-valent sparX-HER2 induces T cell expansion in a dose-dependent manner when co-incubated with HER2-overexpressing BT-474 cells and not in the presence of T47D cells, which exhibit much lower HER2 expression

Affinity-tuned Bi-valent sparX-HER2 Can Potentiate ARC-T Cells to Preferentially Kill HER2-overexpressing Tumor Cells While Sparing Cells with Low HER2 Expression

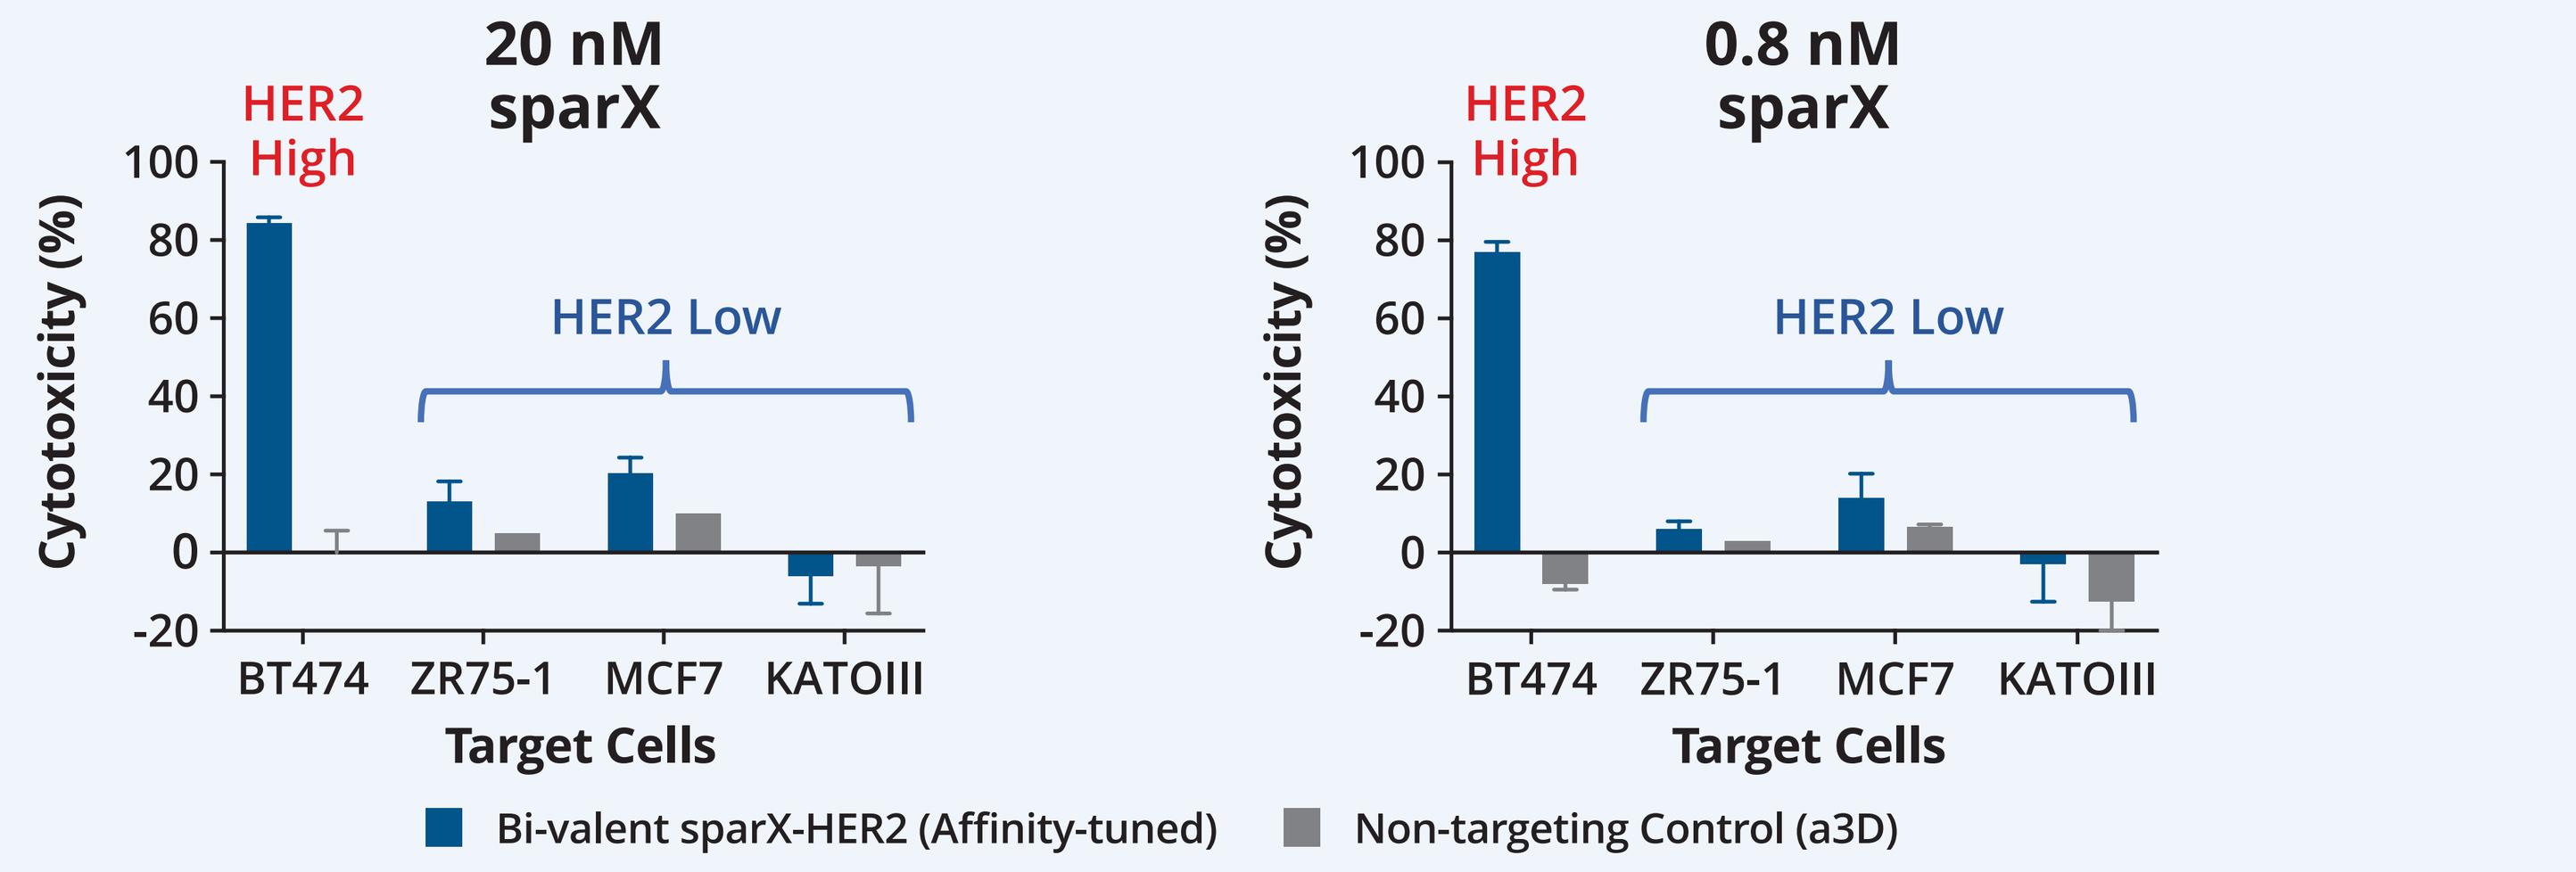

20 nM sparX
0.8 nM sparX
HER2 High
HER2 Low
Cytotoxicity (%)
BT474
ZR75-1
MCF7
KATOIII
Target Cells
Bi-valent sparX-HER2 (Affinity-tuned)
Non-targeting Control (a3D)

- Many solid tumor targets, including HER2, are expressed at lower levels on normal tissues
- sparX-HER2 was engineered for enhanced sensitivity to HER2 receptor density by mutating the target-specific binding domains to the desired affinity range
- Affinity-tuned bi-valent sparX-HER2 demonstrates *in vitro* selectivity for HER2-overexpressing cells across a 25-fold change in concentration

Bi-valent sparX-HER2 Enables ARC-T Cells to Eliminate Subcutaneously Implanted SK-OV-3 Tumors

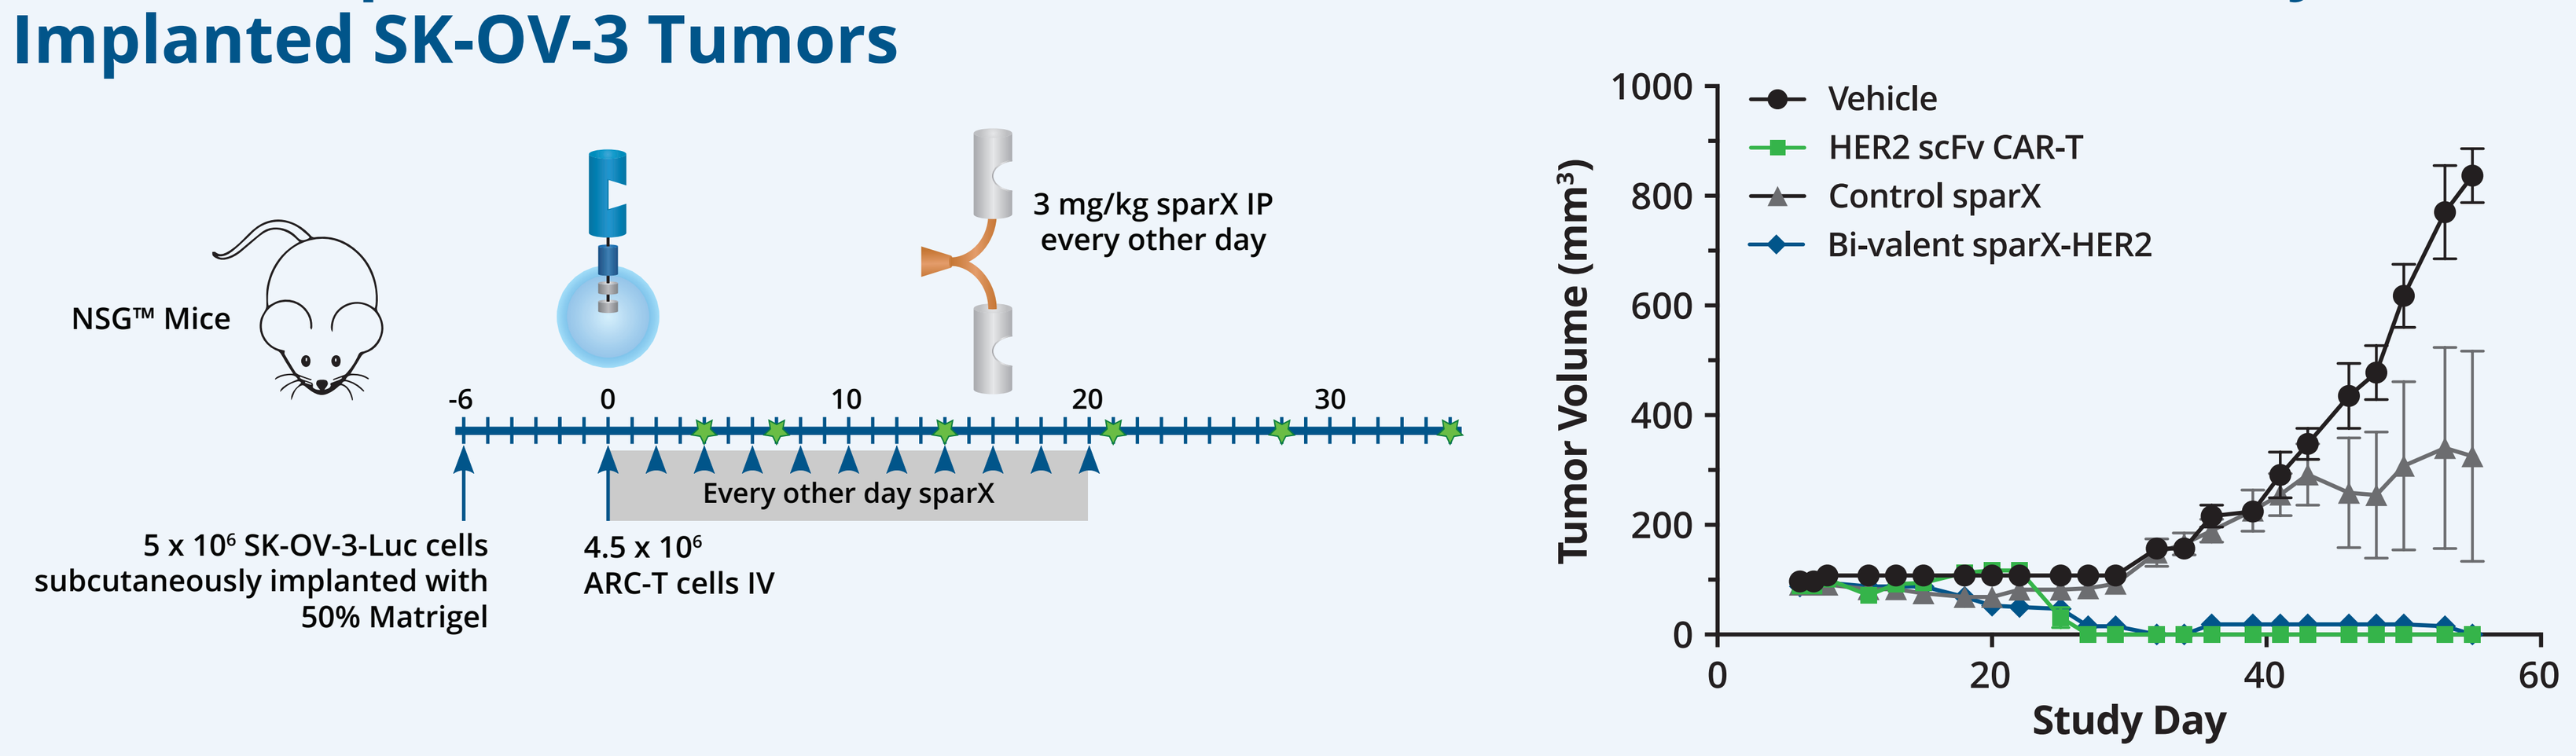

NSG™ Mice
3 mg/kg sparX IP every other day
Every other day sparX
5 x 10⁶ SK-OV-3-Luc cells subcutaneously implanted with 50% Matrigel
4.5 x 10⁶ ARC-T cells IV
Tumor Volume (mm³)
Study Day
Vehicle
HER2 scFv CAR-T
Control sparX
Bi-valent sparX-HER2

- ARC-sparX treatment began once tumors reached approximately 100 mm^3
- Tumor growth attenuation in the non-targeting control sparX group may be attributed to T cell alloreactivity, as donor T cells used in ARC-T cell generation were not HLA-matched with the human tumor cells
- sparX-HER2 enabled ARC-T cells are as effective as conventional HER2 scFv CAR-T cells at inducing complete tumor regression
- A single intravenous dose of ARC-T cells can traffic to the solid tumor site and induce tumor clearance upon systemic administration and co-localization of tumor-targeting sparX

Conclusions

- ARC expression on primary human T cells is comparable to conventional scFv-based CAR expression as characterized by percentages of CAR/ARC-positive T cells
- ARC-sparX activity can be tuned through engineering sparX valency and dose titration
- Affinity-tuned bi-valent sparX can direct ARC-T cells to preferentially kill and proliferate in response to target cells with high antigen expression while sparing those with low/normal levels of antigen expression
- sparX-HER2 directed ARC-T cells perform comparably to HER2 scFv CAR-T cells in eliminating a subcutaneously implanted solid tumor
- Ability to redirect ARC-T cells with sparX proteins may enable physicians to adapt treatments to disease trajectory by addressing tumor heterogeneity and antigen escape

References

1. Schmidts, A. & Maus, M. V. Making CAR T cells a solid option for solid tumors. *Front Immunol.* 9, 2593 (2018).
2. Srivastava, S. & Riddell, S. R. Chimeric antigen receptor T cell therapy: challenges to bench-to-bedside efficacy. *J Immunol.* 200, 459-468 (2018).
3. Morgan, R. A. *et al.* Case report of a serious adverse event following the administration of T cells transduced with a chimeric antigen receptor recognizing ERBB2. *Mol Ther.* 18, 843-851 (2010).
4. Buonato, J. *et al.* Novel CAR-T cell therapy that can be activated, silenced, and reprogrammed in vivo with soluble protein adapters in a dose dependent manner. Poster presented at: ASGCT 23rd Annual Meeting; May 12-15, 2020; Virtual Format.
5. Qin, H. *et al.* Chimeric antigen receptors incorporating D domains targeting CD123 direct potent mono- and bi-specific antitumor activity of T cells. *Mol Ther.* 27, 1262-1274 (2019).
6. Walsh, S. T. *et al.* Solution structure and dynamics of a de novo designed three-helix bundle protein. *Proc Natl Acad Sci USA.* 96, 5486-5491 (1999).
7. Slaga, D. *et al.* Avidity-based binding to HER2 results in selective killing of HER2-overexpressing cells by anti-HER2/CD3. *Sci Transl Med.* 10, eaat5775 (2018).

Presented at the ASCO20 Virtual Scientific Program, May 29–31, 2020