Novel CAR-T Cell Therapy that can be Activated, Silenced, and Reprogrammed *In Vivo* with Soluble Protein Adapters in a Dose Dependent Manner

Abstract 788

Janine Buonato, Justin Edwards, David LaFleur, Jeffrey Swers, Jenny Mu, Liubov Zaritskaya, Ankit Gupta, Hui Wang, Sinnie Ng, Laura Richman, Angela Shen, David Hilbert

Arcellx, Inc., Gaithersburg, MD

Introduction

Arcellx has developed a novel gene-modified cell therapy engineered to address a number of current CAR-T cell limitations such as:

- Fixed antigen targeting is unable to address tumor heterogeneity and antigen escape
- Inability to control rate of cell killing that may result in side effects such as severe CRS, neurotoxicity[1,2] and/or "on-target, off-tumor" toxicities[3]
- Cell exhaustion from being constitutively active[4]
- Limited persistence due to immunogenicity[5]

The ARC-sparX platform is designed to give physicians control of the target specificity and rate of cell killing to potentially increase efficacy and manage toxicities.

The ARC-sparX Platform

The ARC-sparX platform separates the antigen-recognition and killing functions of conventional CAR-T therapy and is comprised of two key components:

1. sparX (**s**oluble **p**rotein **a**ntigen-**r**eceptor **X**-linker) protein: binds specific antigens on diseased cells and flags those cells for destruction.
2. ARC-T Cells (**A**ntigen **R**eceptor **C**omplex **T** Cells): bind sparX proteins and kill flagged cells.

Tri-complex (ARC-T + sparX + target cell) must be formed to activate cytolytic killing.

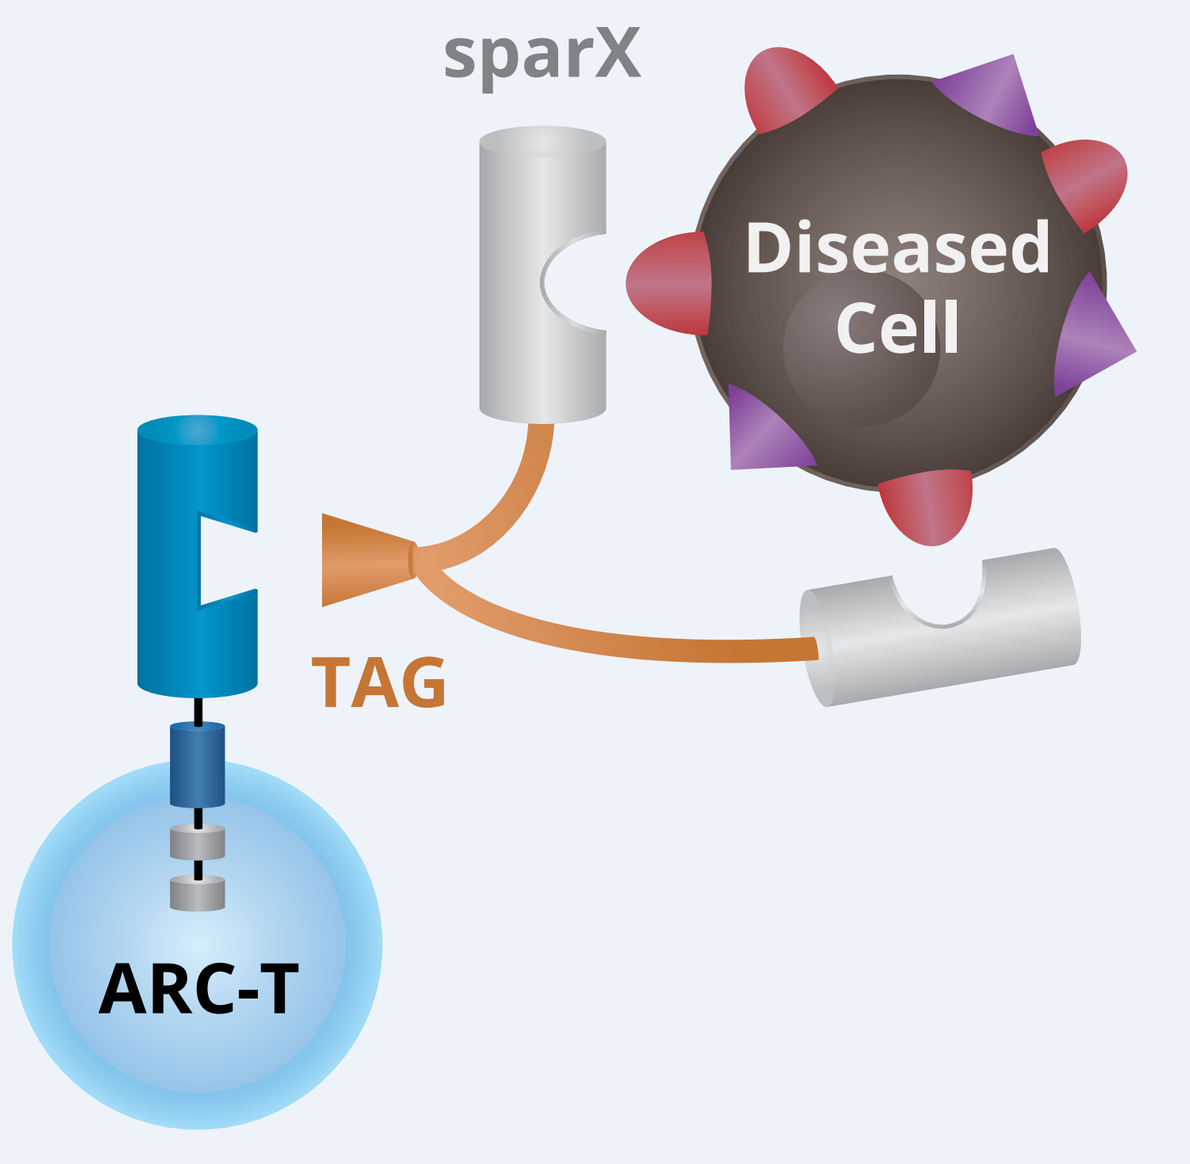

sparX
Diseased Cell
TAG
ARC-T

sparX protein

- No inherent therapeutic activity
- Mono-valent affinity comparable to that of scFv
- Engineered to minimize immunogenic potential
- **"TAG"** is a fragment of human alpha fetoprotein (AFP)
- Predicted human half-life of a few days attributable to "TAG"

ARC-T cell

- Conventional CAR architecture with our novel binding domain
- Receptor for **"TAG"** has affinity in low nanomolar range
- Only activated upon formation of a triplex of ARC-T + sparX + diseased cell
- Same viral vector regardless of antigen target
- ARC is anticipated to function in allogeneic-based cell therapies

Novel Binding Domains and TAG are Foundational to ARC-sparX Platform

Novel Binding Domains

- Non-scFv binding domains are based on α3D scaffold[6] randomized at 12–14 outward facing residues (**Blue** regions)[7]
- Incorporated as binding domains for both ARC-T and sparX
- 8kDa/73aa protein with no cysteine residues or glycosylation sites
- Stable *in vitro* and *in vivo*
- Mono-valent target binding affinities comparable to that of scFv
- Engineered to minimize immunogenic potential

sparX TAG

- ARC-T binding domain is specific for the sparX TAG
- AFP demonstrates many features of an ideal tag
 - Non-immunogenic with pre-established tolerance in humans
 - Minimal functional role in patients
 - *In vitro* and *in vivo* stability
- Arcellx isolated the C-terminal 26kDa fragment of AFP corresponding to AFP Domain III to avoid competition with endogenous AFP
- ARC-T binds TAG but NOT intact AFP

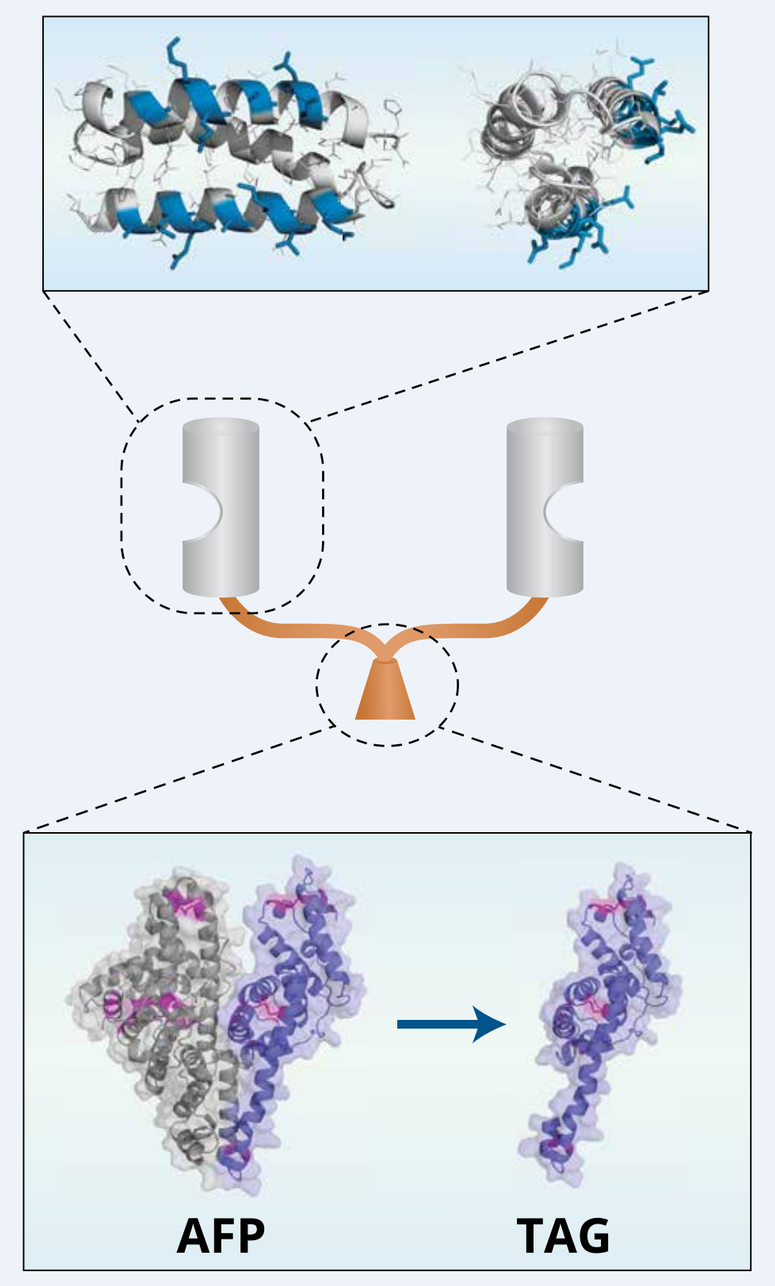

AFP
TAG

sparX Can Be Mono-valent, Multi-valent, or Multi-specific

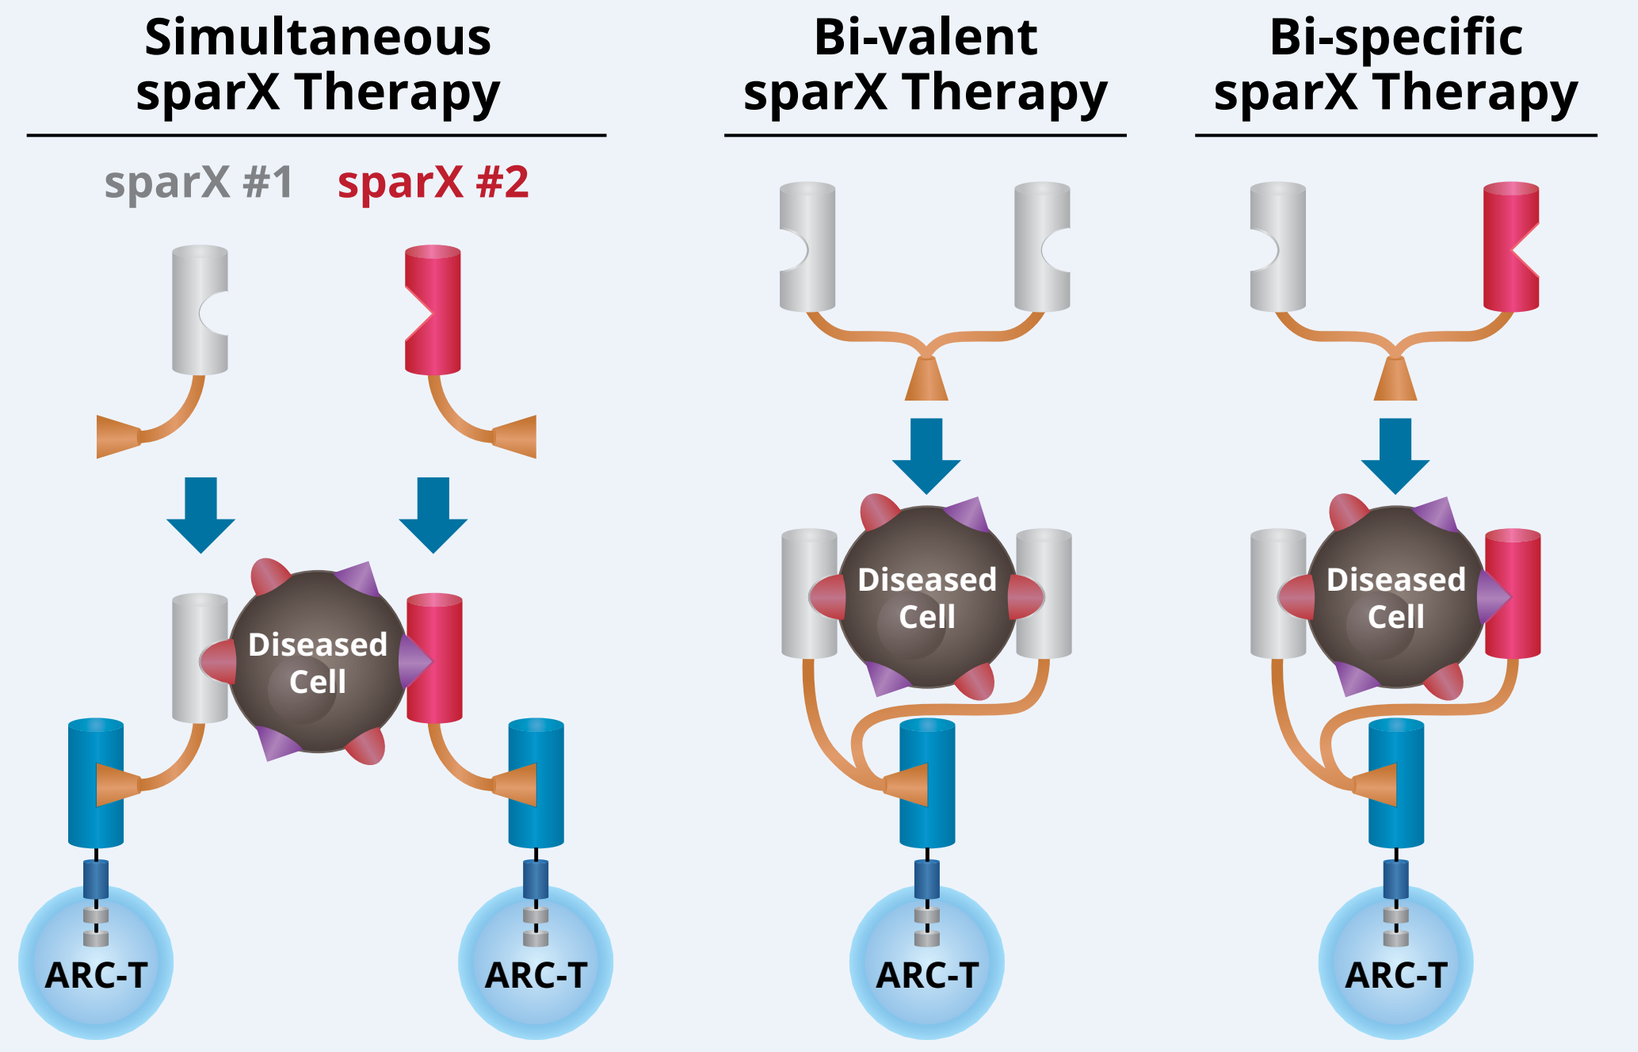

Simultaneous sparX Therapy
Bi-valent sparX Therapy
Bi-specific sparX Therapy
sparX #1
sparX #2
Diseased Cell
ARC-T

- Can be given in combination or in sequence
- Addresses low density or heterogeneous expression of antigen
- Bi-specific sparX proteins support "AND-gated" as well as "OR-gated" targeting of diseased cells

Results

ARC-sparX Tumor Killing is Dose Dependent

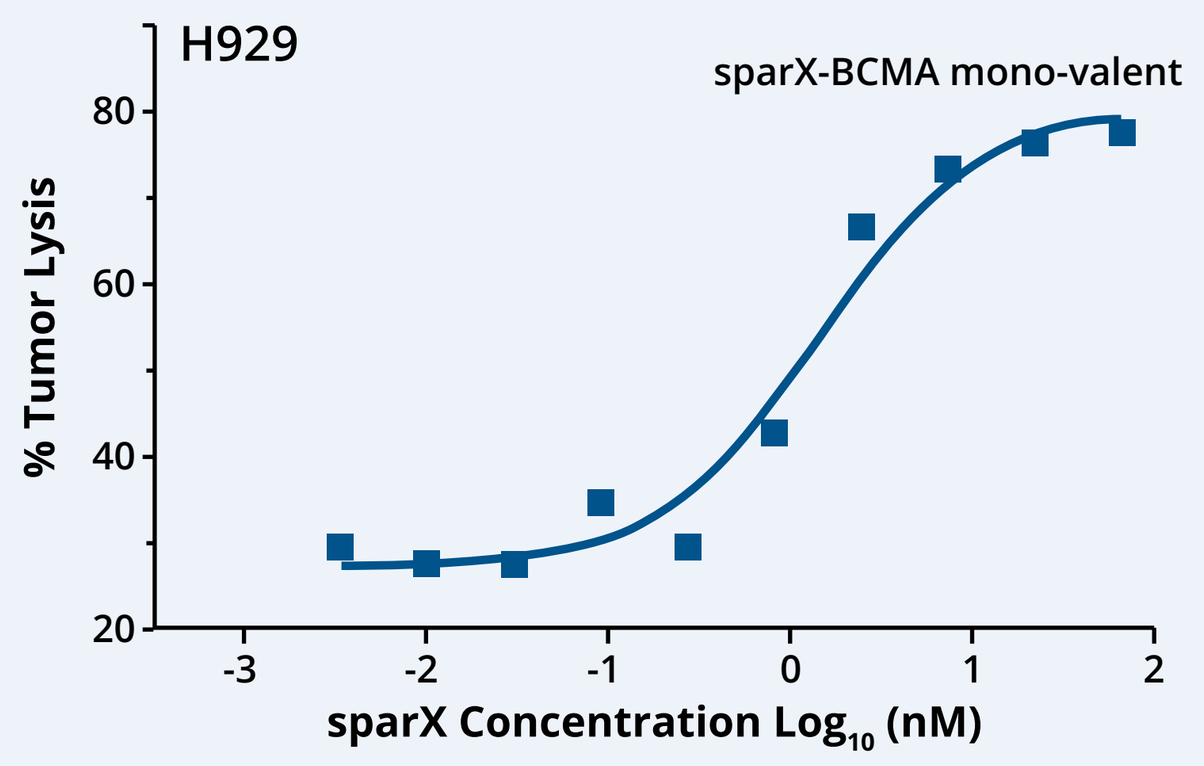

H929
sparX-BCMA mono-valent
% Tumor Lysis
sparX Concentration Log10 (nM)

13 days post ARC-T

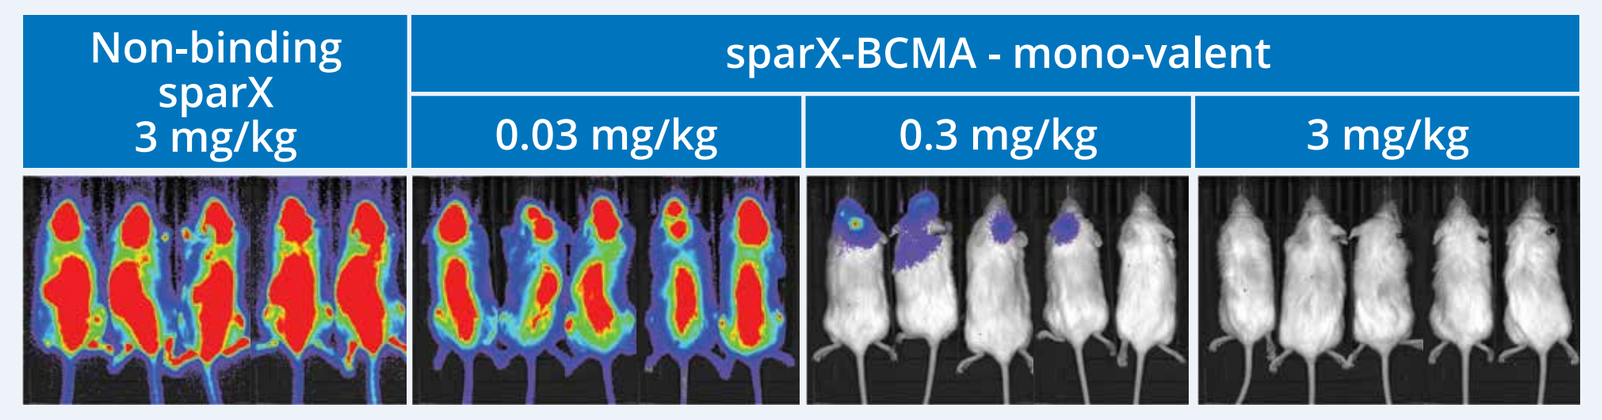

Non-binding sparX 3 mg/kg
sparX-BCMA - mono-valent
0.03 mg/kg
0.3 mg/kg
3 mg/kg

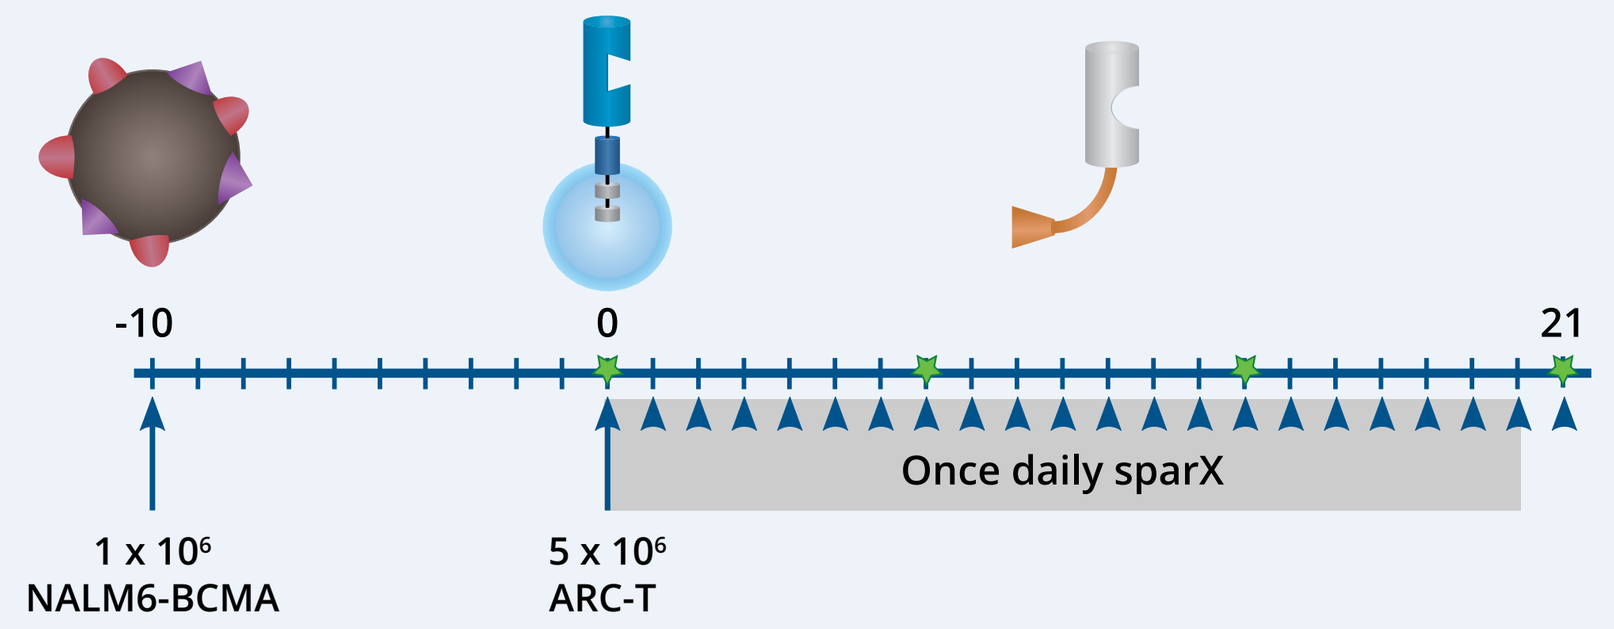

-10
0
21
Once daily sparX
1 x 10^6 NALM6-BCMA
5 x 10^6 ARC-T

- ARC-T cells induced killing of GFP/Luciferase expressing H929 target cells dependent on the dose of mono-valent sparX-BCMA **(top)**
- ARC-T cells with mono-valent sparX BCMA demonstrated dose dependent elimination of tumors generated with BCMA-expressing NALM6 cells *in vivo* with daily sparX administration **(middle)**
- As shown in bottom schematic, disseminated disease was induced by intravenous transplant of 1 x 10^6 BCMA-expressing NALM6-GFP/Luciferase cells for 10 days prior to administration of 5 x 10^6 ARC-T cells and the subsequent first dose of sparX (D0). Daily intraperitoneal sparX injections continued for 21 days. Tumor growth was monitored via bioluminescence imaging once or twice weekly throughout the experiment. *In vivo* experiments throughout were performed similarly except as noted
- ARC-T cells throughout were generated via lentiviral transduction of T-cells enriched from healthy human donor PMBCs

ARC-sparX Tumor Clearance Activity Is Comparable with Conventional CAR-T

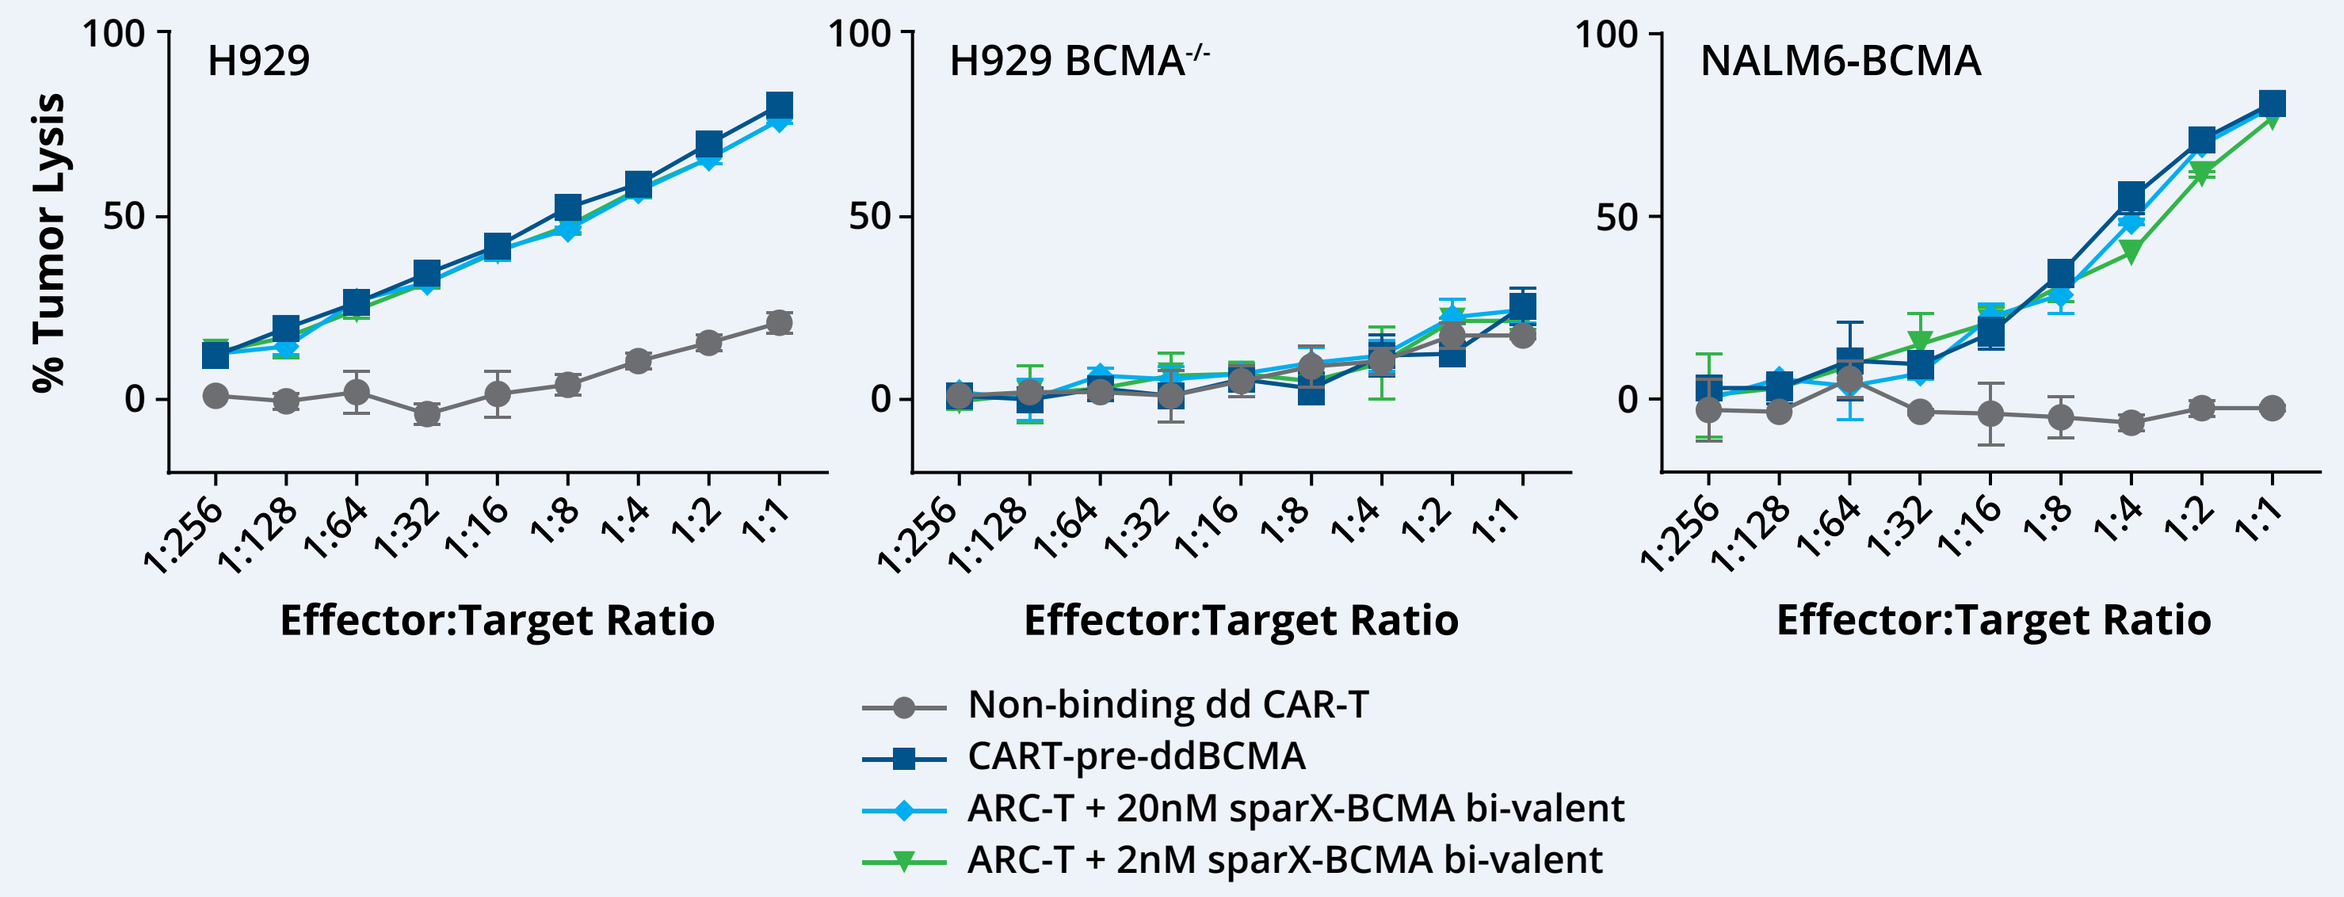

H929
H929 BCMA-/-
NALM6-BCMA
% Tumor Lysis
Effector:Target Ratio
Non-binding dd CAR-T
CART-pre-ddBCMA
ARC-T + 20nM sparX-BCMA bi-valent
ARC-T + 2nM sparX-BCMA bi-valent

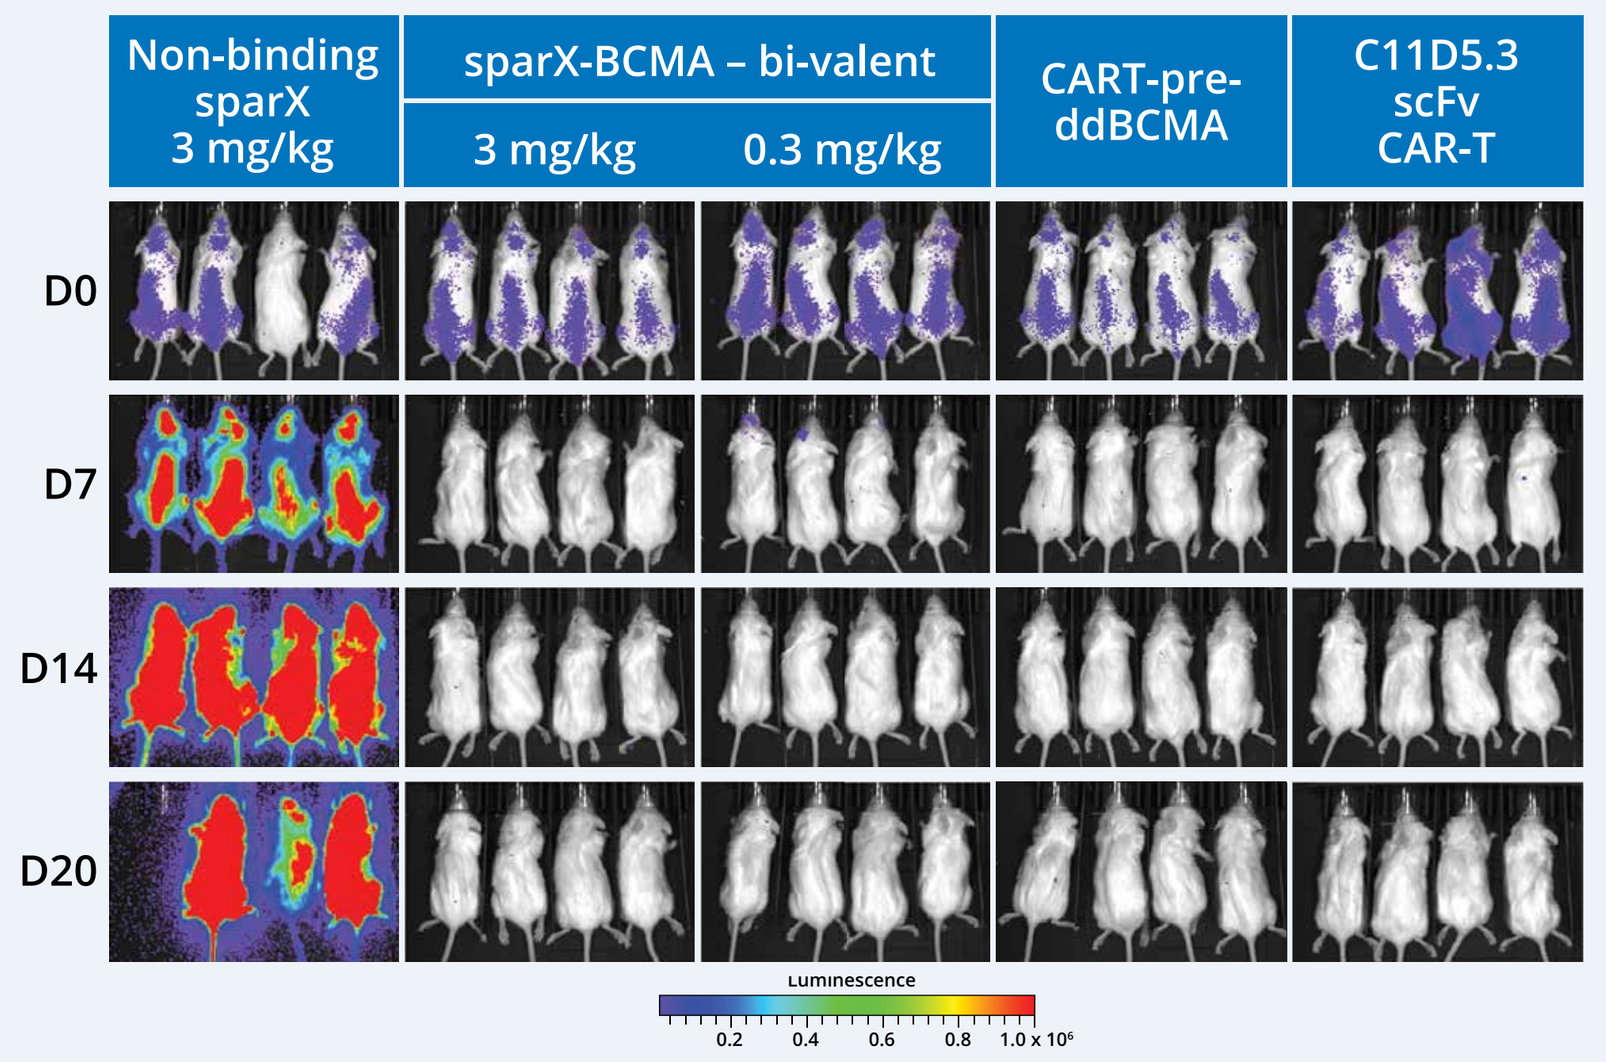

Non-binding sparX 3 mg/kg
sparX-BCMA – bi-valent
3 mg/kg
0.3 mg/kg
CART-pre-ddBCMA
C11D5.3 scFv CAR-T
D0
D7
D14
D20

- CART-pre-ddBCMA is a preclinical prototype of the clinical candidate CART-ddBCMA. Both contain conventional CAR architecture utilizing our novel binding domain that has specificity for BCMA
- *In vitro* lysis of GFP/Luciferase expressing target cells was shown to be equivalent for ARC-T + sparX-BCMA and CART-pre-ddBCMA, with target-specific activity demonstrated using H929 cells with CRISPR-mediated BCMA knockout (H929-BCMA$^{-/-}$) **(top)**
- Tumors generated with BCMA-expressing NALM6 cells were grown for 10 days prior to administration of 5 x 10^6 CART-pre-ddBCMA cells, C11D5.3 scFv CAR-T cells, or co-administration of ARC-T cells and first dose of sparX. Daily sparX continued for 21 days **(bottom)**
- ARC-T with bi-valent sparX-BCMA demonstrates *in vivo* tumor control that is equivalent to both conventionally-formatted CAR-T cells – CART-pre-ddBCMA and C11D5.3 scFv CAR-T cells

Bivalency Improves on Mono-valent sparX-BCMA Efficacy *In Vitro* and *In Vivo*

sparX-BCMA Format	k_{on} ($M^{-1}s^{-1}$)	k_{off} (s^{-1})	K_D (M)
Mono-valent	1.6E6	7.3E-2	4.6E-8
Bi-valent	8.2E6	7.6E-4	9.2E-11

Target Cell Lysis

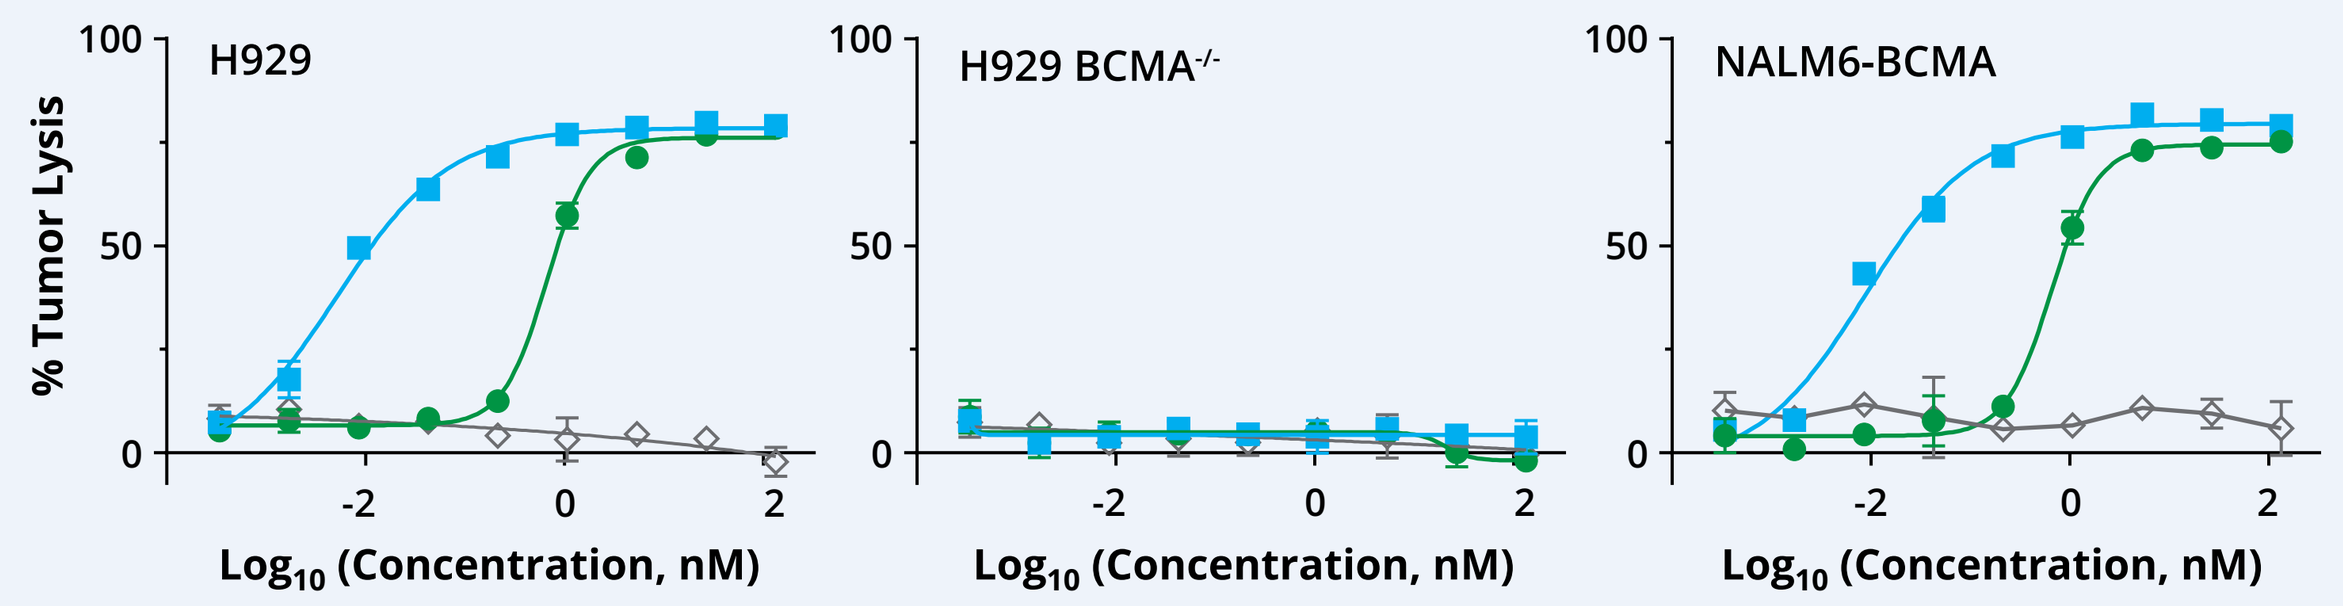

H929
H929 BCMA-/-
NALM6-BCMA
% Tumor Lysis
Log10 (Concentration, nM)

Cytokine Release

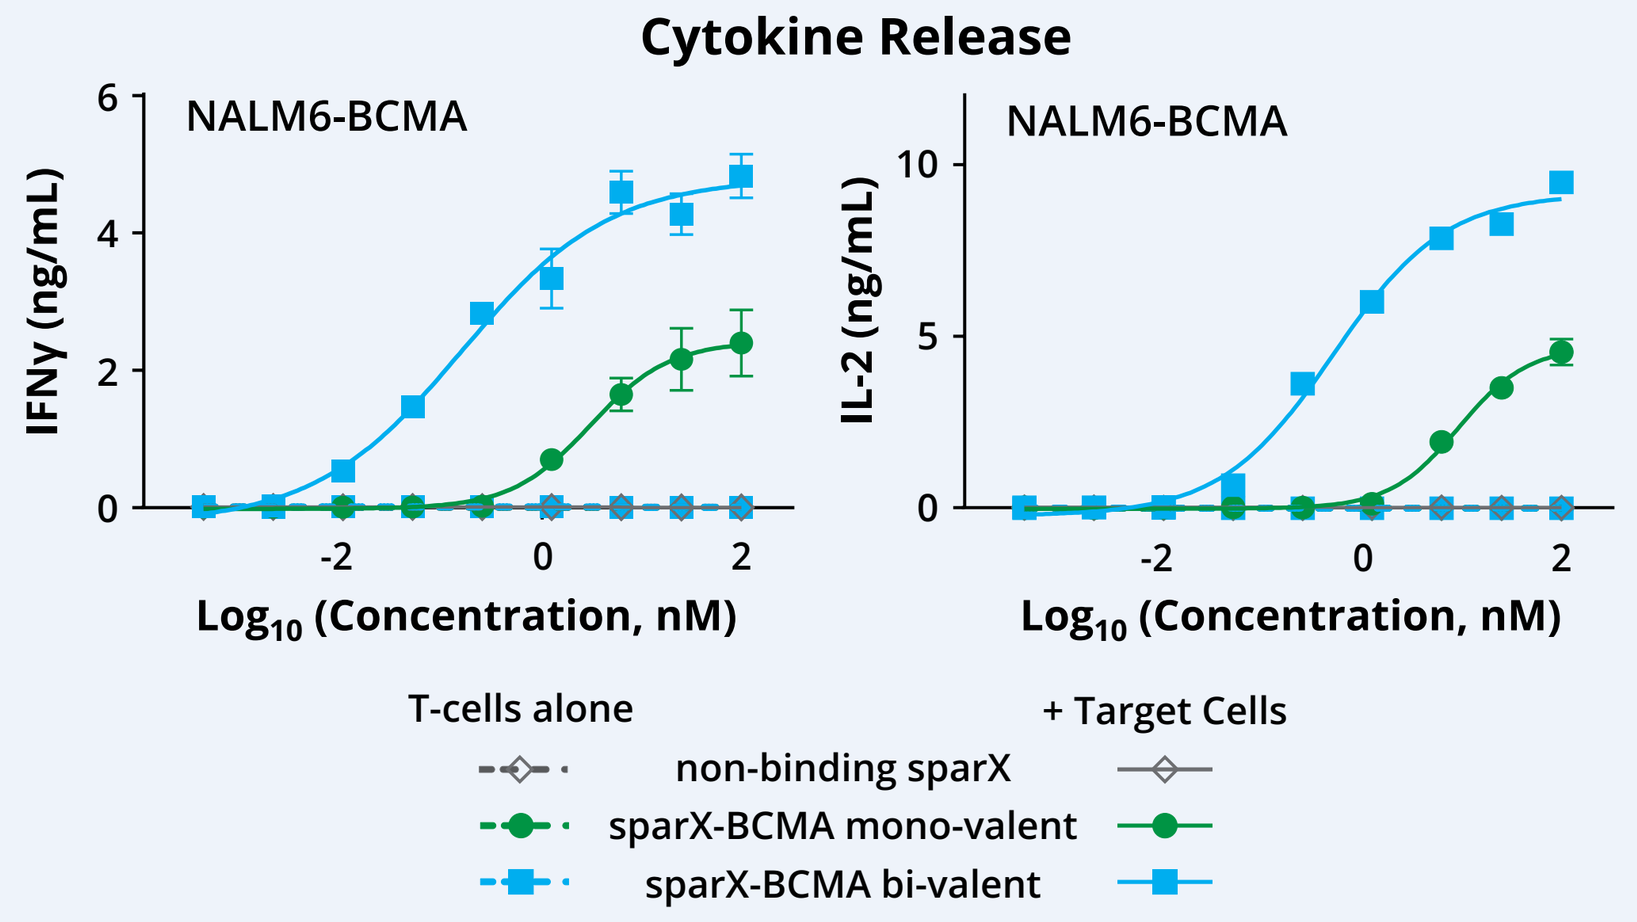

NALM6-BCMA
IFNγ (ng/mL)
IL-2 (ng/mL)
Log10 (Concentration, nM)
T-cells alone
+ Target Cells
non-binding sparX
sparX-BCMA mono-valent
sparX-BCMA bi-valent

- Affinity measurements by surface plasmon resonance (SPR) using Biacore, shows 500-fold improvement in K_D with bi-valent vs mono-valent BCMA binding **(table above)**
- EC_{50}s for ARC-T cell induced lysis of GFP/Luciferase expressing target cells **(top)** and cytokine release **(bottom)** are significantly enhanced in bi-valent format. The maxima for IFNγ and IL-2 release are also increased

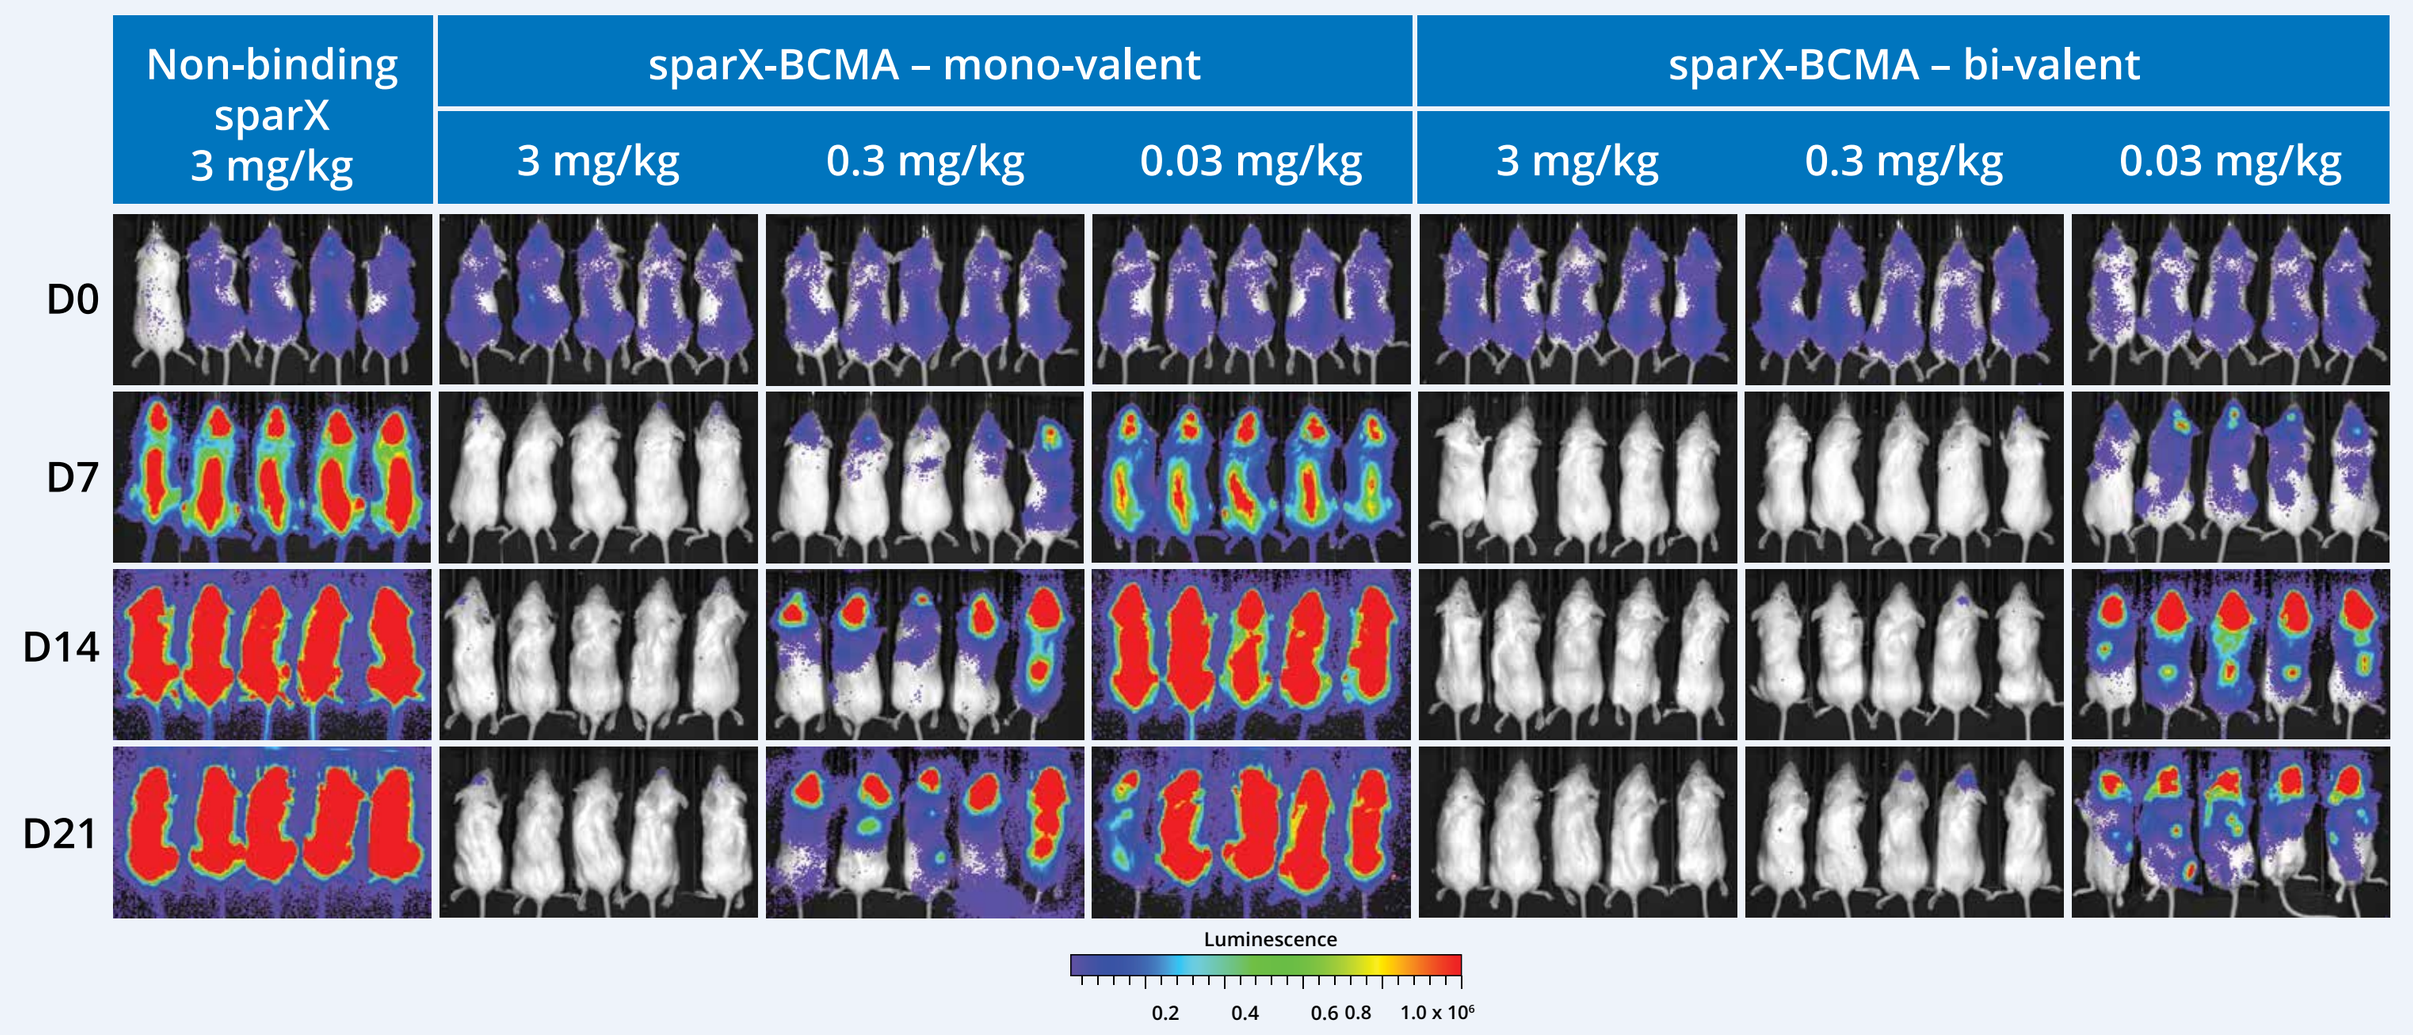

Non-binding sparX 3 mg/kg
sparX-BCMA – mono-valent
sparX-BCMA – bi-valent
3 mg/kg
0.3 mg/kg
0.03 mg/kg
D0
D7
D14
D21

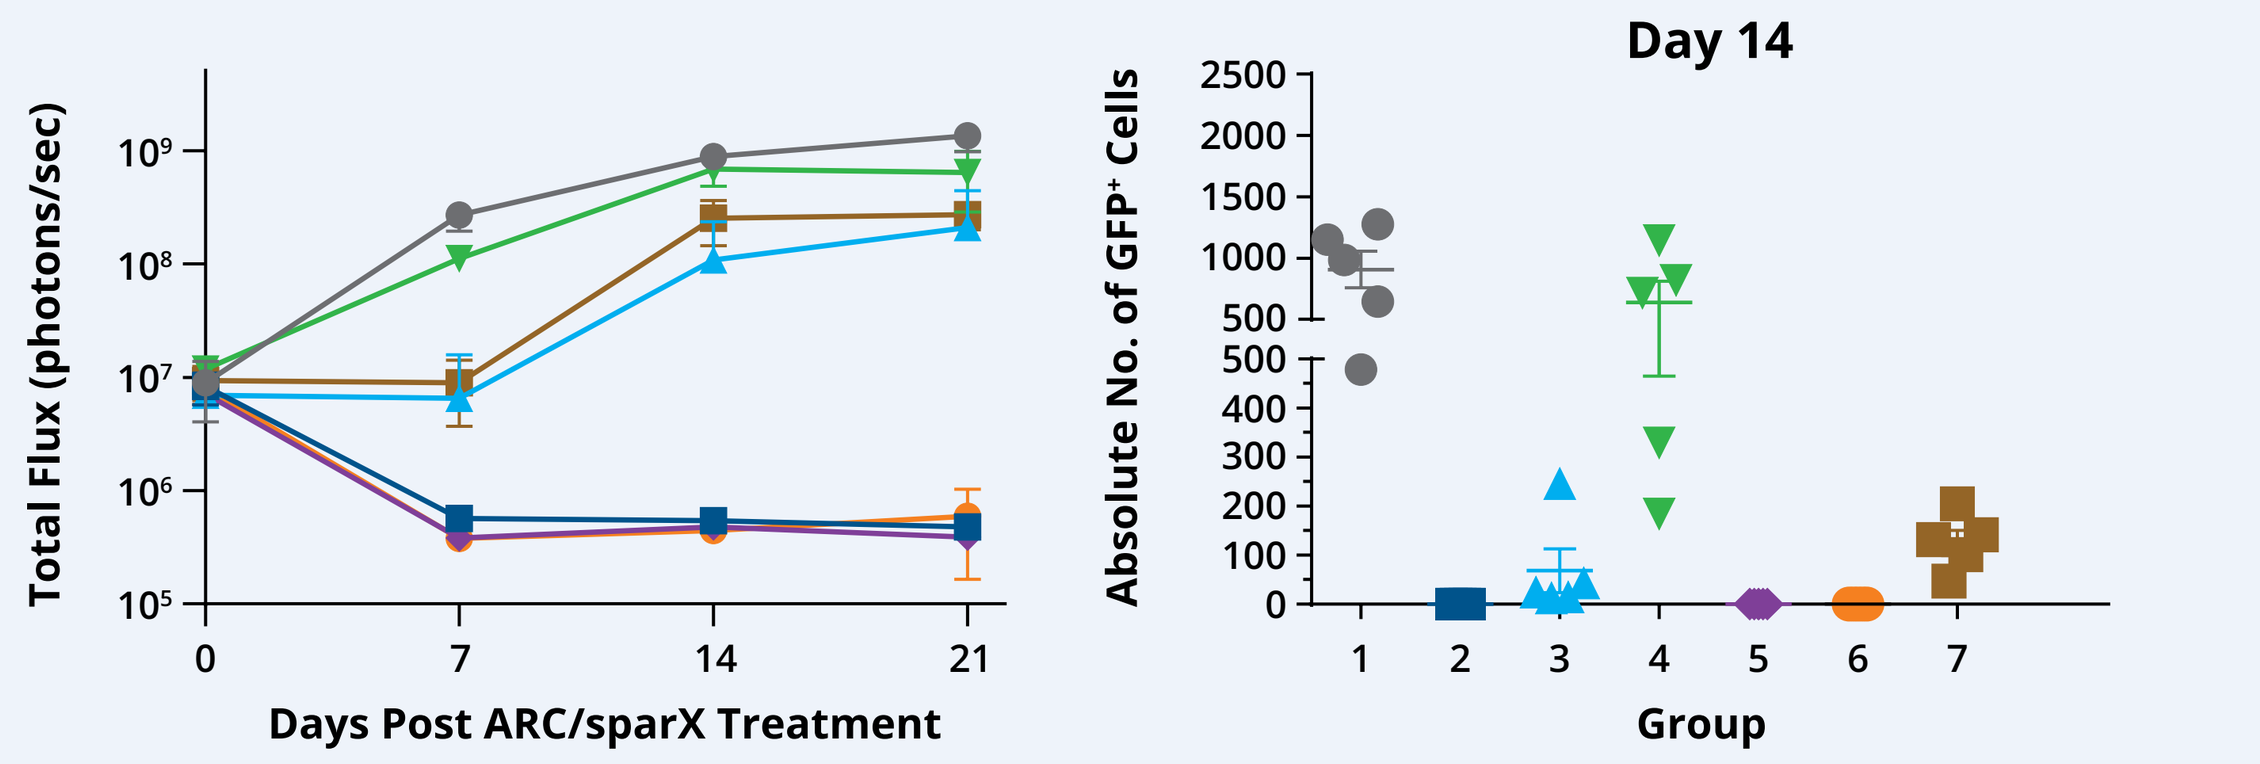

Total Flux (photons/sec)
Days Post ARC/sparX Treatment
Day 14
Absolute No. of GFP+ Cells
Group

Group	T cells	sparX	sparX Dose (mg/kg)
1	ARC-T	non-binding sparX	3
2	ARC-T	sparX-BCMA mono-valent	3
3	ARC-T	sparX-BCMA mono-valent	0.3
4	ARC-T	sparX-BCMA mono-valent	0.03
5	ARC-T	sparX-BCMA bi-valent	3
6	ARC-T	sparX-BCMA bi-valent	0.3
7	ARC-T	sparX-BCMA bi-valent	0.03

- Tumors generated with NALM6-BCMA cells were grown for 10 days prior to administration of CAR-T cells and subsequent first dose of sparX (Day 0). Daily sparX continued for 21 days
- Both mono- and bi-valent sparX-BCMA demonstrated dose-dependent elimination of NALM6-BCMA tumors, with increased potency for the bi-valent format
- Enumeration of GFP$^+$ NALM6-BCMA cells from blood drawn on Day 14 confirms dose-dependent elimination of circulating tumor cells **(right)**

ARC-T Can Be Reprogrammed Using sparX *In Vivo*

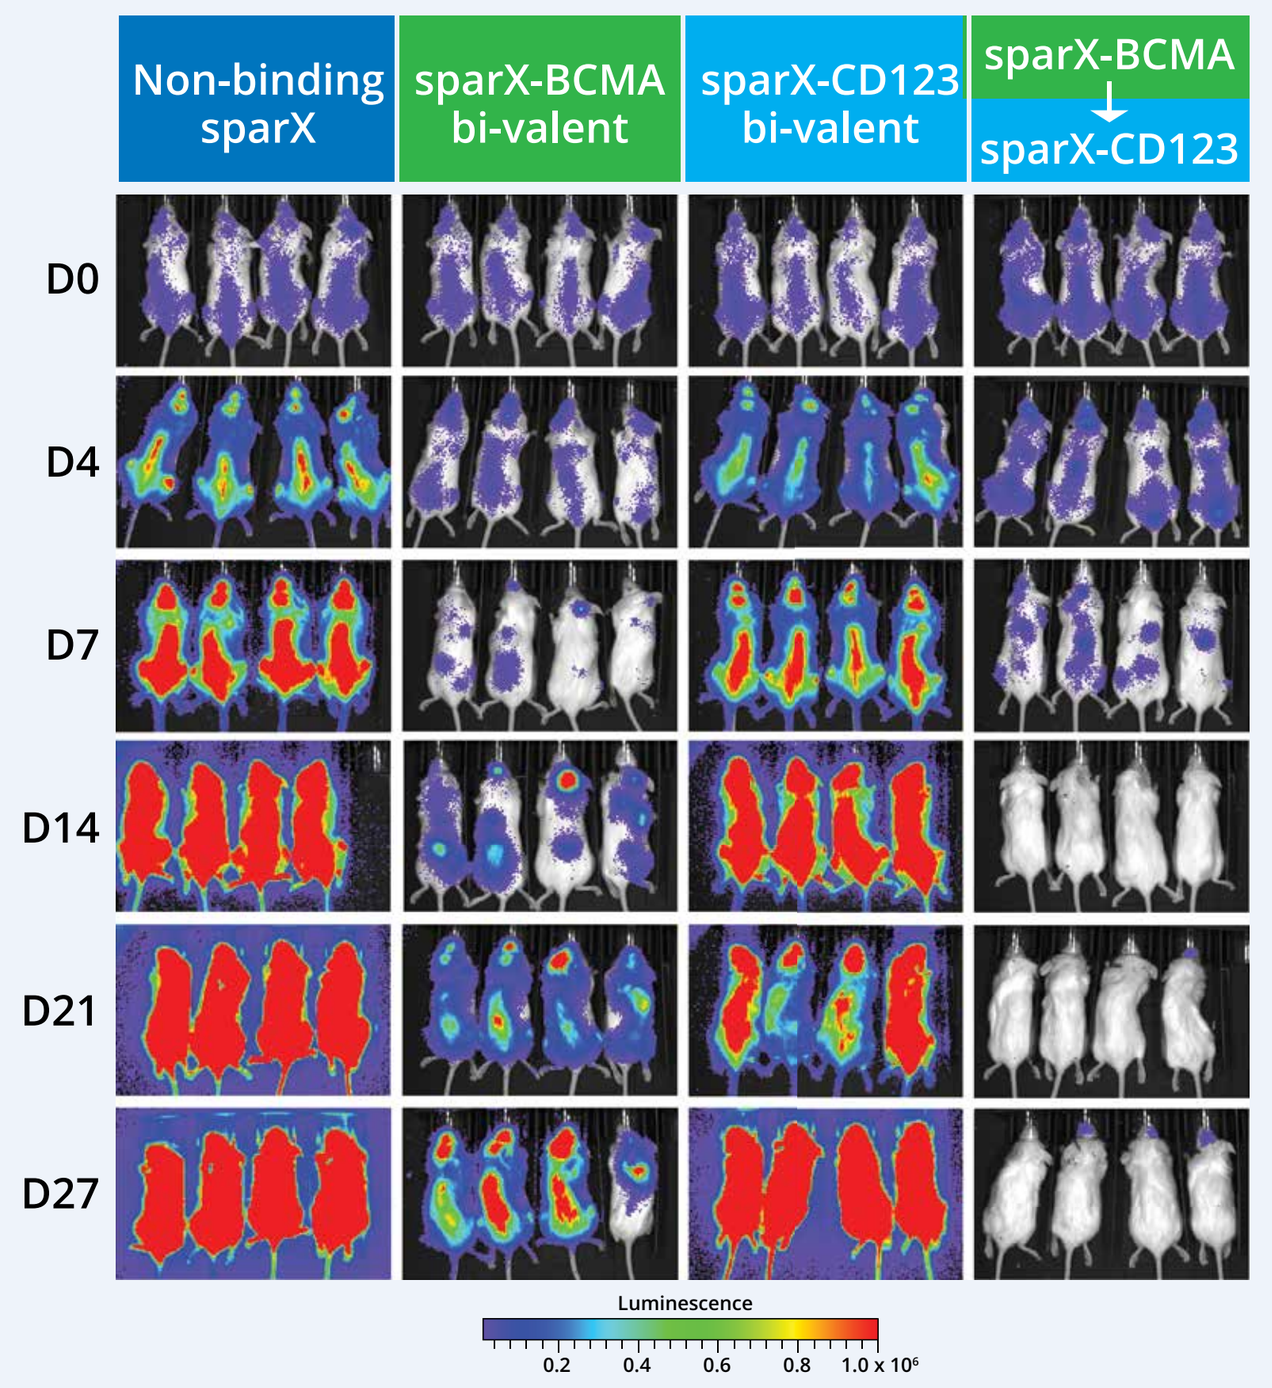

Non-binding sparX
sparX-BCMA bi-valent
sparX-CD123 bi-valent
sparX-BCMA
sparX-CD123
D0
D4
D7
D14
D21
D27
Luminescence

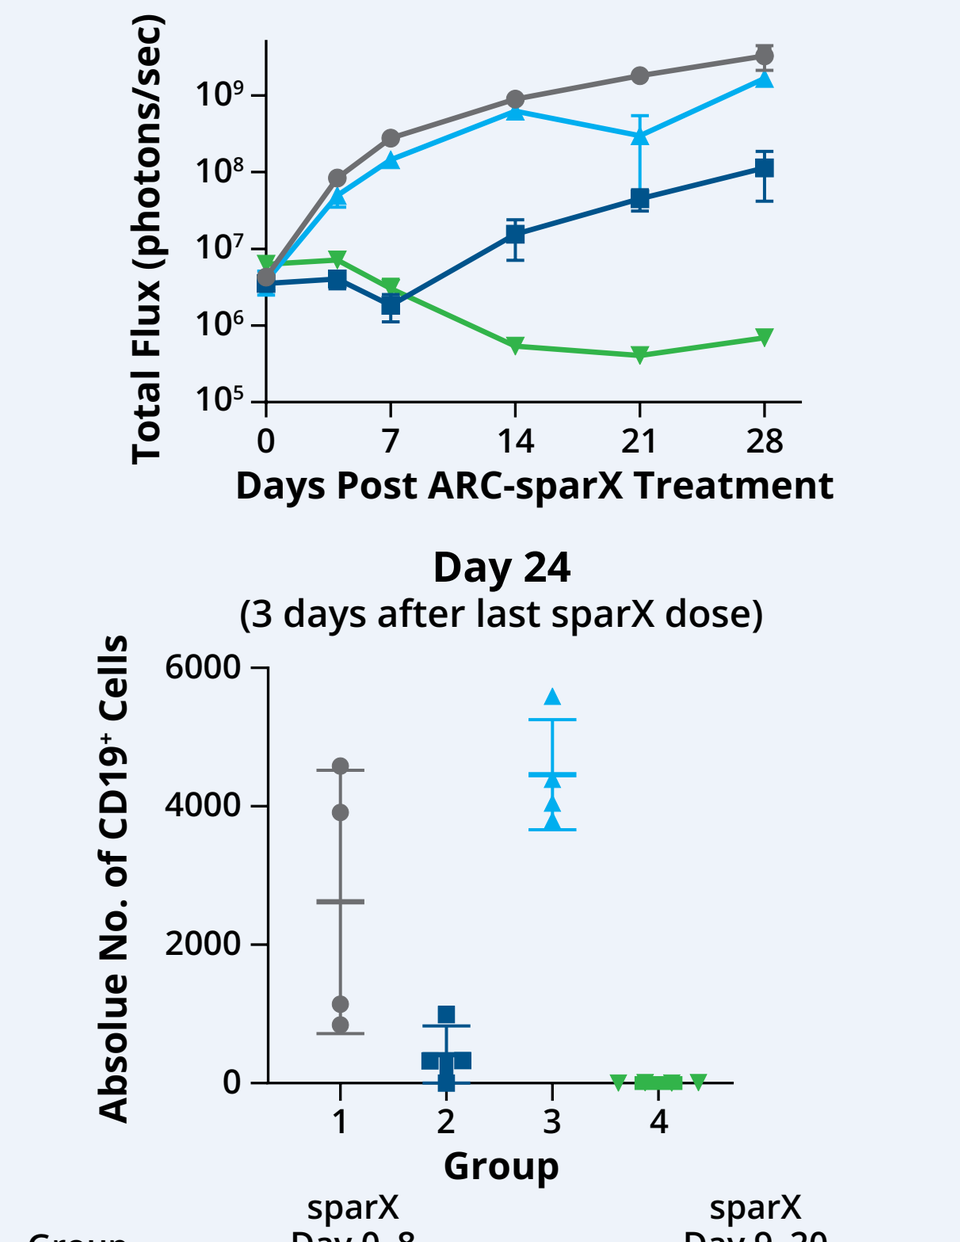

Total Flux (photons/sec)
Days Post ARC-sparX Treatment
Day 24
(3 days after last sparX dose)
Absolue No. of CD19+ Cells
Group

Group	sparX Day 0–8	sparX Day 9–20
1	non-binding sparX	non-binding sparX
2	sparX-BCMA bi-valent	sparX-BCMA bi-valent
3	sparX-CD123 bi-valent	sparX-CD123 bi-valent
4	sparX-BCMA bi-valent	sparX-CD123 bi-valent

- NALM6 cells were transduced with BCMA or CD123 and fluorescent markers GFP or Amcyan, respectively. Tumors were seeded with mixed cell population containing 96% NALM6-BCMA and 4% NALM6-CD123 cells
- Mixed population tumors were grown for 10 days prior to administration of ARC-T cells and subsequent first dose of bi-valent sparX recognizing BCMA or CD123. After 9 days of daily sparX administration, Group 4 was switched from sparX-BCMA to sparX-CD123 for an additional 11 doses
- CD19$^+$ enumeration was used to capture both NALM6-BCMA/GFP and NALM6-CD123/Amcyan cells
- sparX-BCMA without CD123-targeting leads to emergence of NALM6-CD123 tumor rebound
- Reprogramming with sparX-CD123 after sparX-BCMA drove deeper response to heterogenous tumor

Conclusions

- ARC-sparX is a novel, adaptive, and controllable cell therapy platform
 - sparX proteins can be engineered to target different antigens
 - Universal ARC-T cell product can be paired with any sparX
- ARC-sparX activity can be tuned through engineering sparX valency and titrating dose
- sparX targeting BCMA or CD123 potently induce ARC-T activity to eliminate tumors
- ARC-sparX performs on-par with traditional scFv-based CAR-T with the following differences:
 - ARC-T cells are not constitutively active
 - sparX dosing controls ARC-T cell killing in a dose-dependent manner
 - ARC-T cells can be reprogrammed *in vivo* by switching sparX

Future Plans

- Use current collection of sparX to treat hematologic malignancies, solid tumors, and autoimmune diseases
- Ongoing Phase I clinical trial of CART-ddBCMA for the treatment of multiple myeloma (MM) is designed to validate the functional properties of our novel non-scFv binding domain
- Planned Clinical Trials
 - Phase I trial for ARC-T + sparX-BCMA for r/r MM
 - Phase I trial for ARC-T + sparX-CD123 for AML/MDS
- Continue to expand collection of sparX proteins to bind different antigens, including novel targets
- Pursue allogeneic ARC-T program

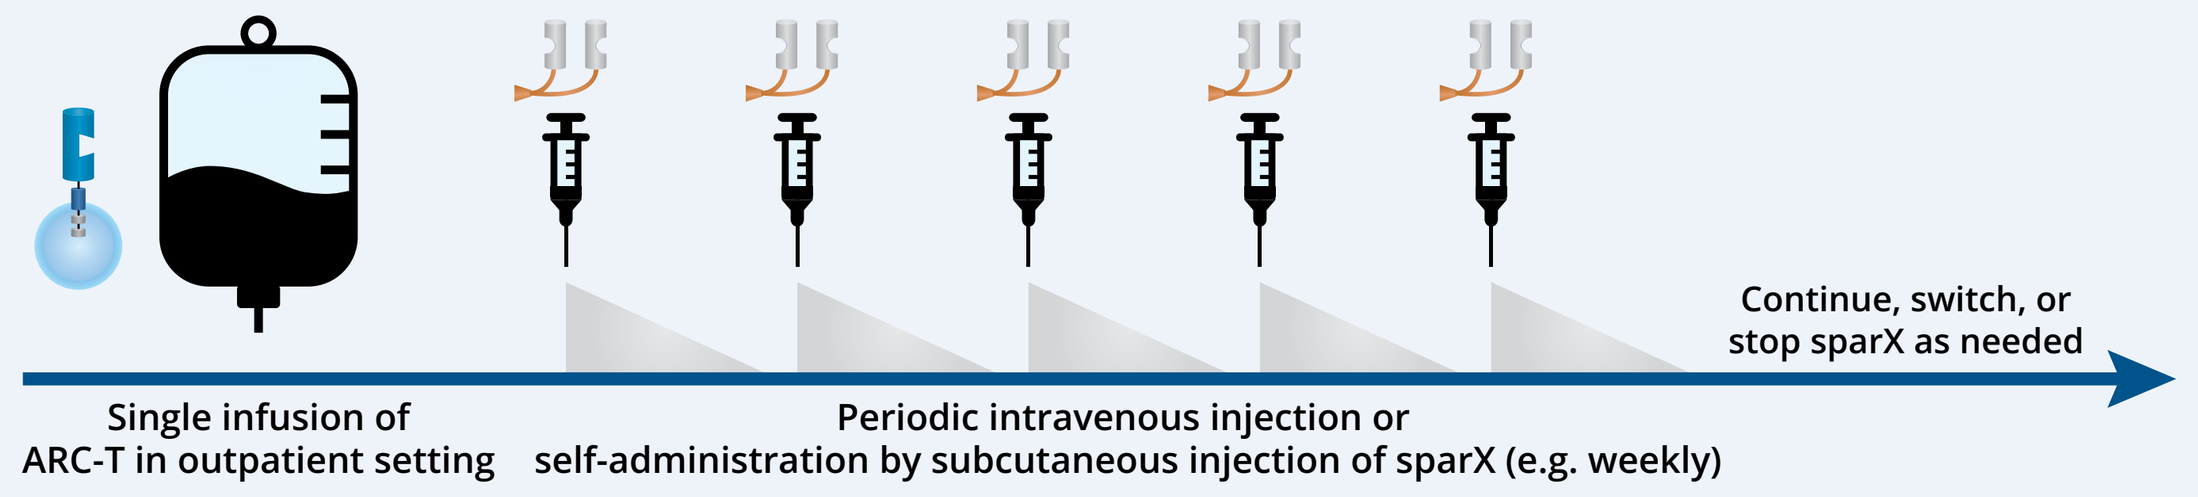

Continue, switch, or stop sparX as needed
Single infusion of ARC-T in outpatient setting
Periodic intravenous injection or self-administration by subcutaneous injection of sparX (e.g. weekly)

References

1. Kymriah Package Insert; Novartis Pharmaceuticals Corp. East Hanover, New Jersey 07936. 2018; https://www.fda.gov/downloads/BiologicsBloodVaccines/CellularGeneTherapyProducts/ApprovedProducts/UCM573941.pdf. **2.** Yescarta Package Insert; Kite Pharma, Inc. Santa Monica, CA 90404. 2017; https://www.fda.gov/downloads/BiologicsBloodVaccines/CellularGeneTherapyProducts/ApprovedProducts/UCM581226.pdf. **3.** Morgan R, Yang J, Kitano M, *et al.* Case Report of a Serious Adverse Event Following the Administration of T Cells Transduced With a Chimeric Antigen Receptor Recognizing ERBB2. *Molecular Therapy.* 2010 Apr;18(4):843–51. **4.** Viaud S, Ma JSY, Hardy IR et al. Switchable control over in vivo CAR T expansion, B cell depletion, and induction of memory. *Proc Natl Acad Sci USA.* 2018 Nov;115(46):E10898–E10906. **5.** Lamers HJ, Willemsen R, van Elzakker P, *et al.* Immune responses to transgene and retroviral vector in patients treated with *ex vivo*–engineered T cells. *Blood.* 2011;117(1):72-82. **6.** Walsh S, Cheng H, Bryson J, *et al.* Solution Structure and Dynamics of a de novo designed three-helix bundle protein. *Proc Natl Acad Sci USA.* 1999 May;96(10):5486-91. **7.** Qin H, Edwards JP, Zaritskaya L, *et al.* Chimeric antigen receptors incorporating D domains targeting CD123 direct potent mono-and bi-specific antitumor activity of T cells. *Mol Ther.* 2019 Jul;27(7):1262-74.

Presented at the American Society of Gene & Cell Therapy Virtual Annual Meeting, May 12–15, 2020

©2020 Arcellx, Inc. All rights reserved.